Peptide vaccination against PD-L1 reduces tumor growth in preclinical models through stimulation of PD-L1-targeting T cells in the tumor microenvironment.

Marion Chapellier[1], Marco Carretta[1], Erika Sutanto-Ward[2], James DuHadaway[2], Souvik Dey[2], Dema Ghaban[2,3], Preeyam Patel[1], Marcos Iglesias[1], Qin Tang[1], Megan Biller[1], Inés Lecoq[1], Lea Svendsen[1], Evelina Martinenaite[1], Alexander J Muller[2], Ayako Pedersen[1]

[1] IO Biotech, Copenhagen, Denmark, [2] Lankenau Institute for Medical Research, Wynnewood, Pennsylvania, USA, [3] Drexel University College of Medicine, Philadelphia, Pennsylvania, USA

IO BIOTECH

Background

Programmed Death Ligand 1 (PD-L1) is a type 1 transmembrane protein encoded by CD274 gene, its interaction with PD-1 inhibits T cell proliferation and activation to control immune response and contributes to cancer progression and immune escape. The PD-1/PD-L1 blocking antibodies have shown clinical efficacy in many cancers, however drug resistance mechanisms in patients remain a key challenge.

As an alternative and supplemental approach to PD-1/PD-L1 blockade, vaccination approach to promote T-cell immunity against PD-L1$^+$ target cells has been proposed after the observation that effector/cytotoxic T cells reactive against PD-L1 are found in the peripheral blood of cancer patients and healthy individuals [1,2]. Our current study aims to evaluate the efficacy and mechanism of PD-L1 peptide-vaccine using preclinical models.

PD-L1 expression in tumor models

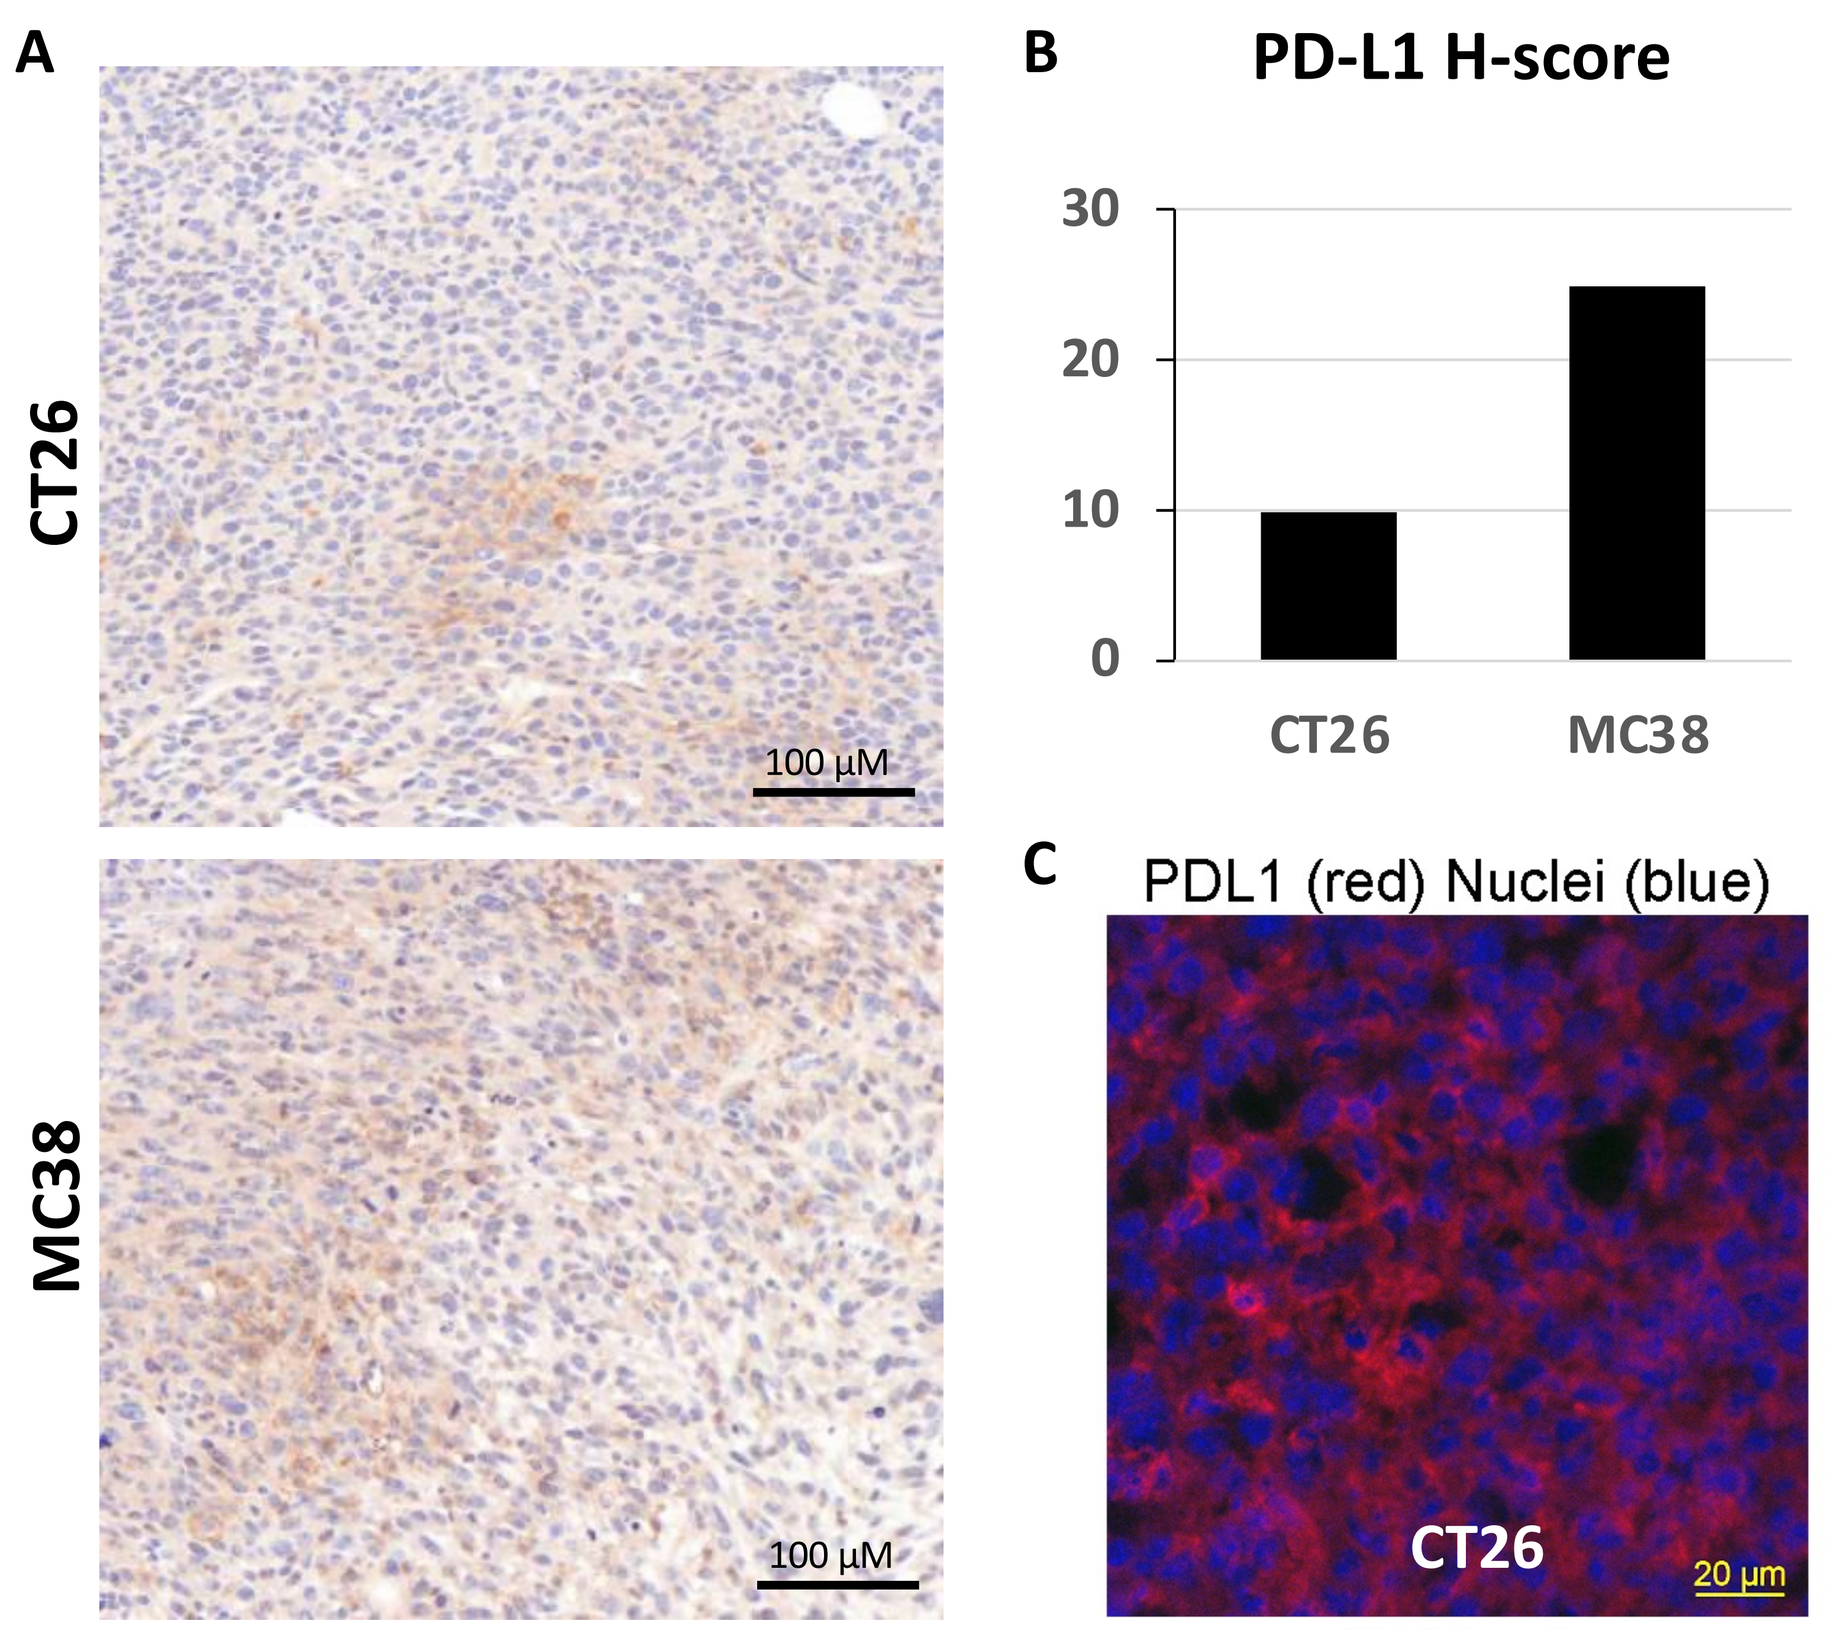
A
CT26
MC38
100 µM
B
PD-L1 H-score
30
20
10
0
CT26
MC38
C
PDL1 (red) Nuclei (blue)
CT26
20 µm

(A) PD-L1 expression was detected on resected tumors from CT26 and MC38 syngeneic tumor models and (B) PD-L1$^+$ cells were scored following intensity levels, H-score was calculated according to standard formula. (C) PD-L1 expression in CT26 tumors was further confirmed per immunofluorescence on frozen tissues sections.

PD-L1 peptide vaccination induces the expansion of PD-L1 specific T cells in mice

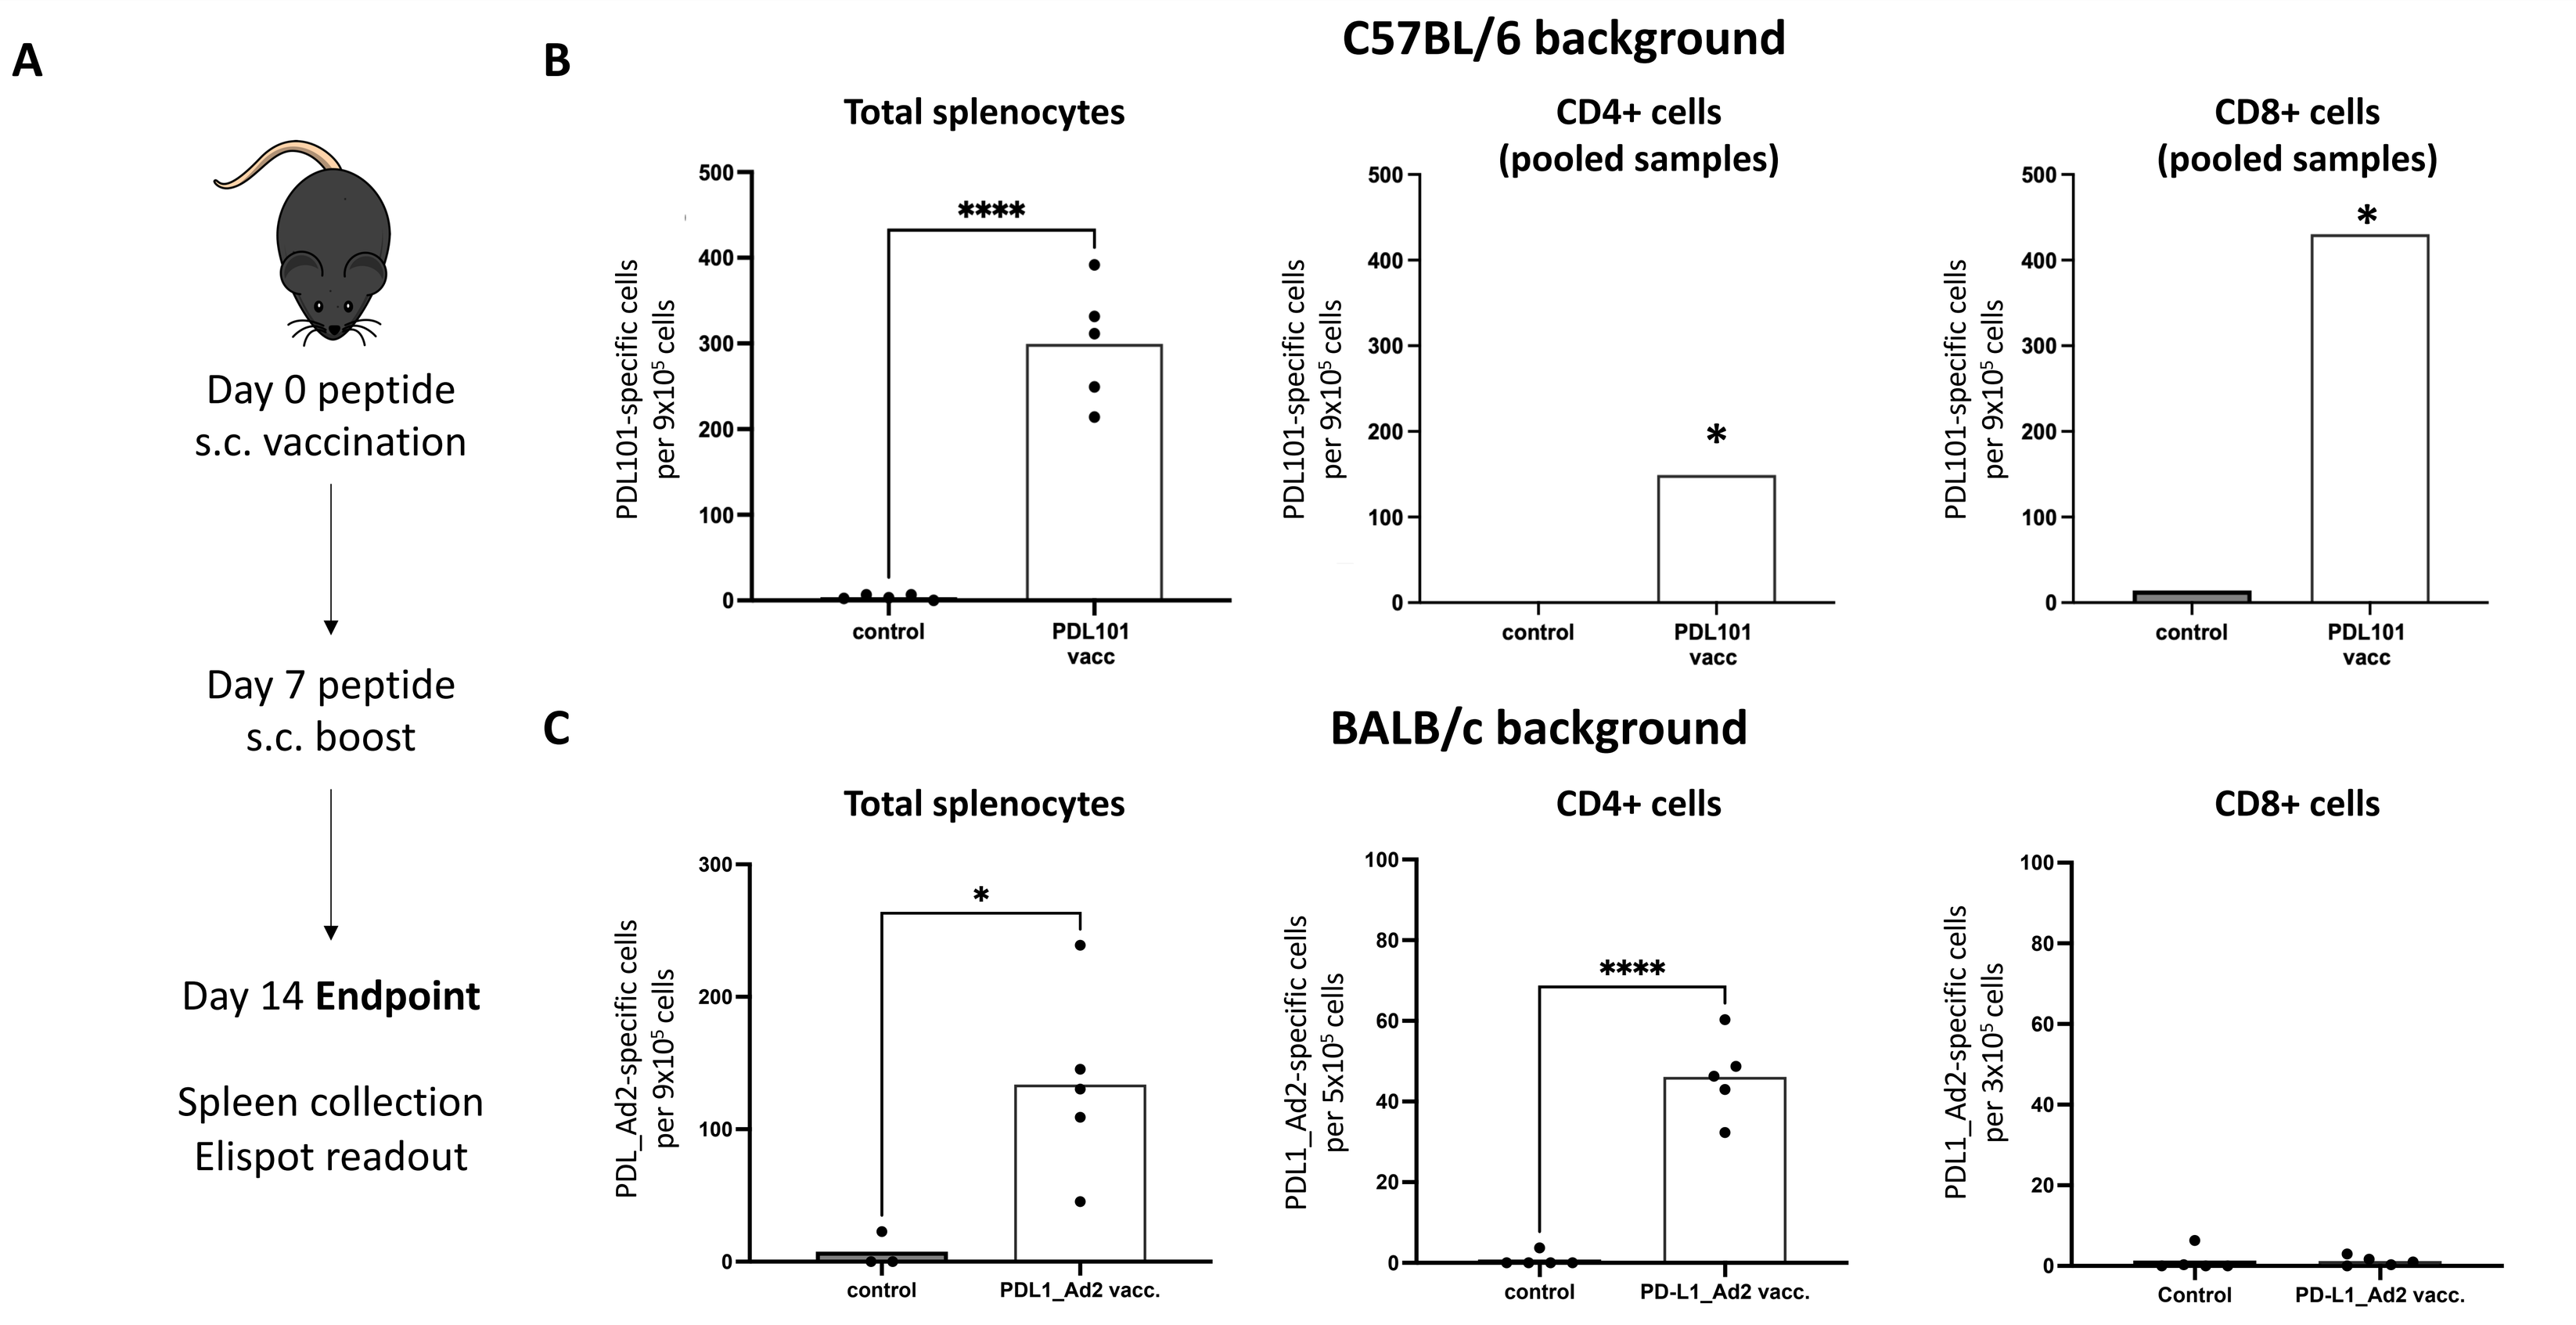
A
Day 0 peptide
s.c. vaccination
Day 7 peptide
s.c. boost
Day 14 Endpoint
Spleen collection
Elispot readout
B
C57BL/6 background
Total splenocytes
CD4+ cells
(pooled samples)
CD8+ cells
(pooled samples)
PDL101-specific cells per 9x10⁵ cells
control
PDL101 vacc
C
BALB/c background
Total splenocytes
CD4+ cells
CD8+ cells
PDL_Ad2-specific cells per 9x10⁵ cells
PDL1_Ad2-specific cells per 5x10⁵ cells
PDL1_Ad2-specific cells per 3x10⁵ cells
control
PDL1_Ad2 vacc.
PD-L1_Ad2 vacc.

(A) C57BL/6 animals were vaccinated with an MHC-I directed peptide PDL101 and BALB/c animals were vaccinated with an MHC-II directed peptide PD-L1_Ad2. Animals received two treatments on days 0 and 7 and splenocytes were collected 7 days after the last vaccination. The presence of PD-L1 specific T cells was evaluated in an IFNγ Elispot assay after overnight stimulation of total or CD4/CD8 sorted cells. Scatter plots show number of PD-L1 specific T cells detected in cell populations from (B) C57BL6 strain and (C) BALB/c strain treated with background relevant peptides. Data shown as mean +/- SEM.

PD-L1 peptide vaccination reduces tumor growth in murine models

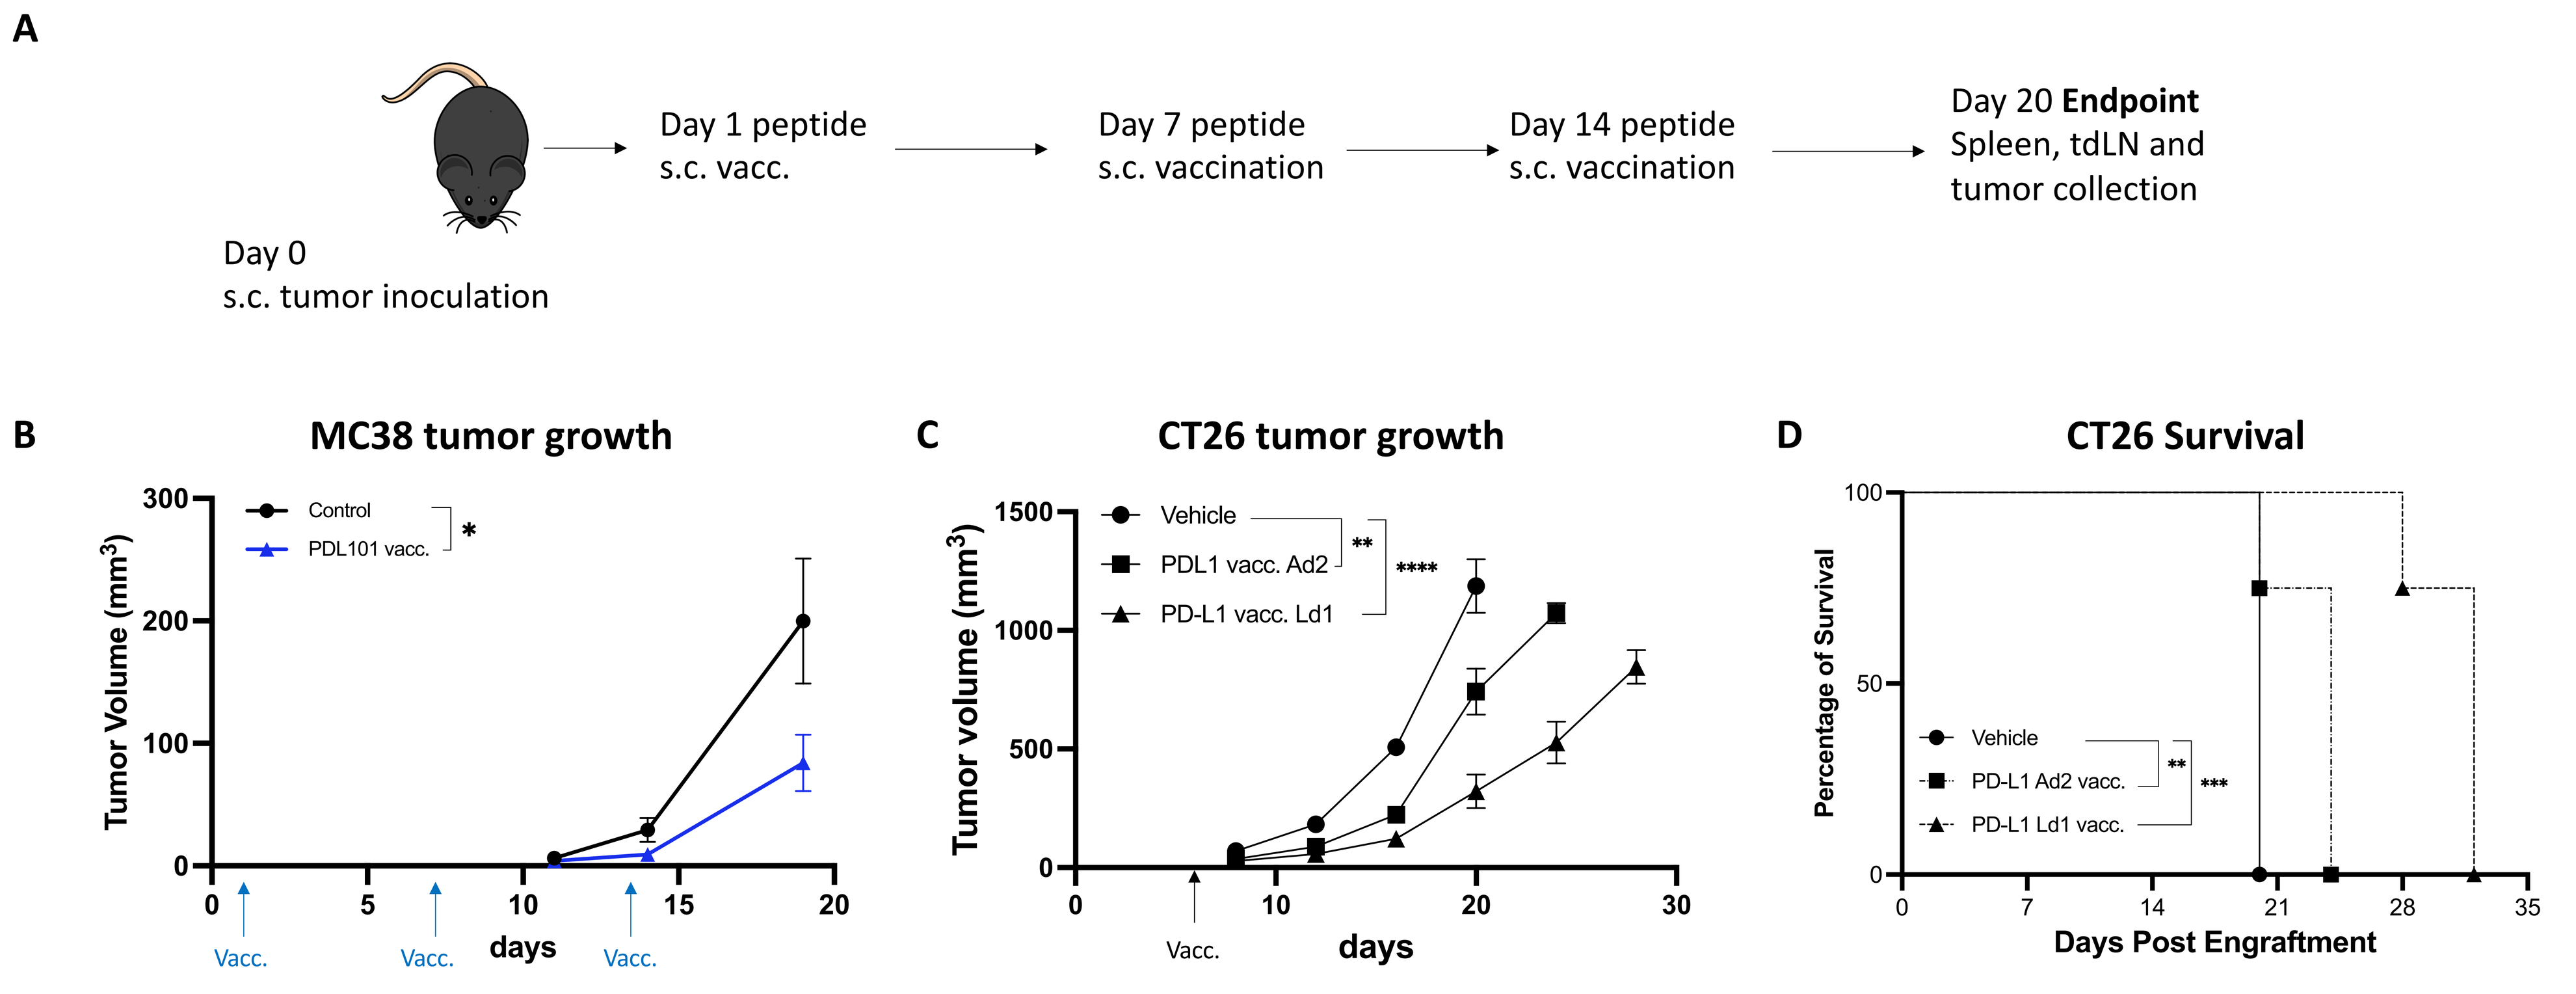
A
Day 0
s.c. tumor inoculation
Day 1 peptide
s.c. vacc.
Day 7 peptide
s.c. vaccination
Day 14 peptide
s.c. vaccination
Day 20 Endpoint
Spleen, tdLN and
tumor collection
B
MC38 tumor growth
Control
PDL101 vacc.
Tumor Volume (mm³)
days
Vacc.
C
CT26 tumor growth
Vehicle
PDL1 vacc. Ad2
PD-L1 vacc. Ld1
Tumor volume (mm³)
days
Vacc.
D
CT26 Survival
Vehicle
PD-L1 Ad2 vacc.
PD-L1 Ld1 vacc.
Percentage of Survival
Days Post Engraftment

(A) Animals were inoculated with MC38 or CT26 tumor cells and vaccinated with relevant PD-L1 peptide on days 1, 7 and 14 (MC38) or on day 7 (CT26). (B-C) Tumor growth curves of MC38 and CT26 following treatment with PD-L1 peptides were established using mean tumor volume at each time point +/- SEM. (D) Kaplan-Meier survival analysis was performed in CT26 model.

PD-L1 vaccine induces PD-L1 specific T cells traffic into the tumor site

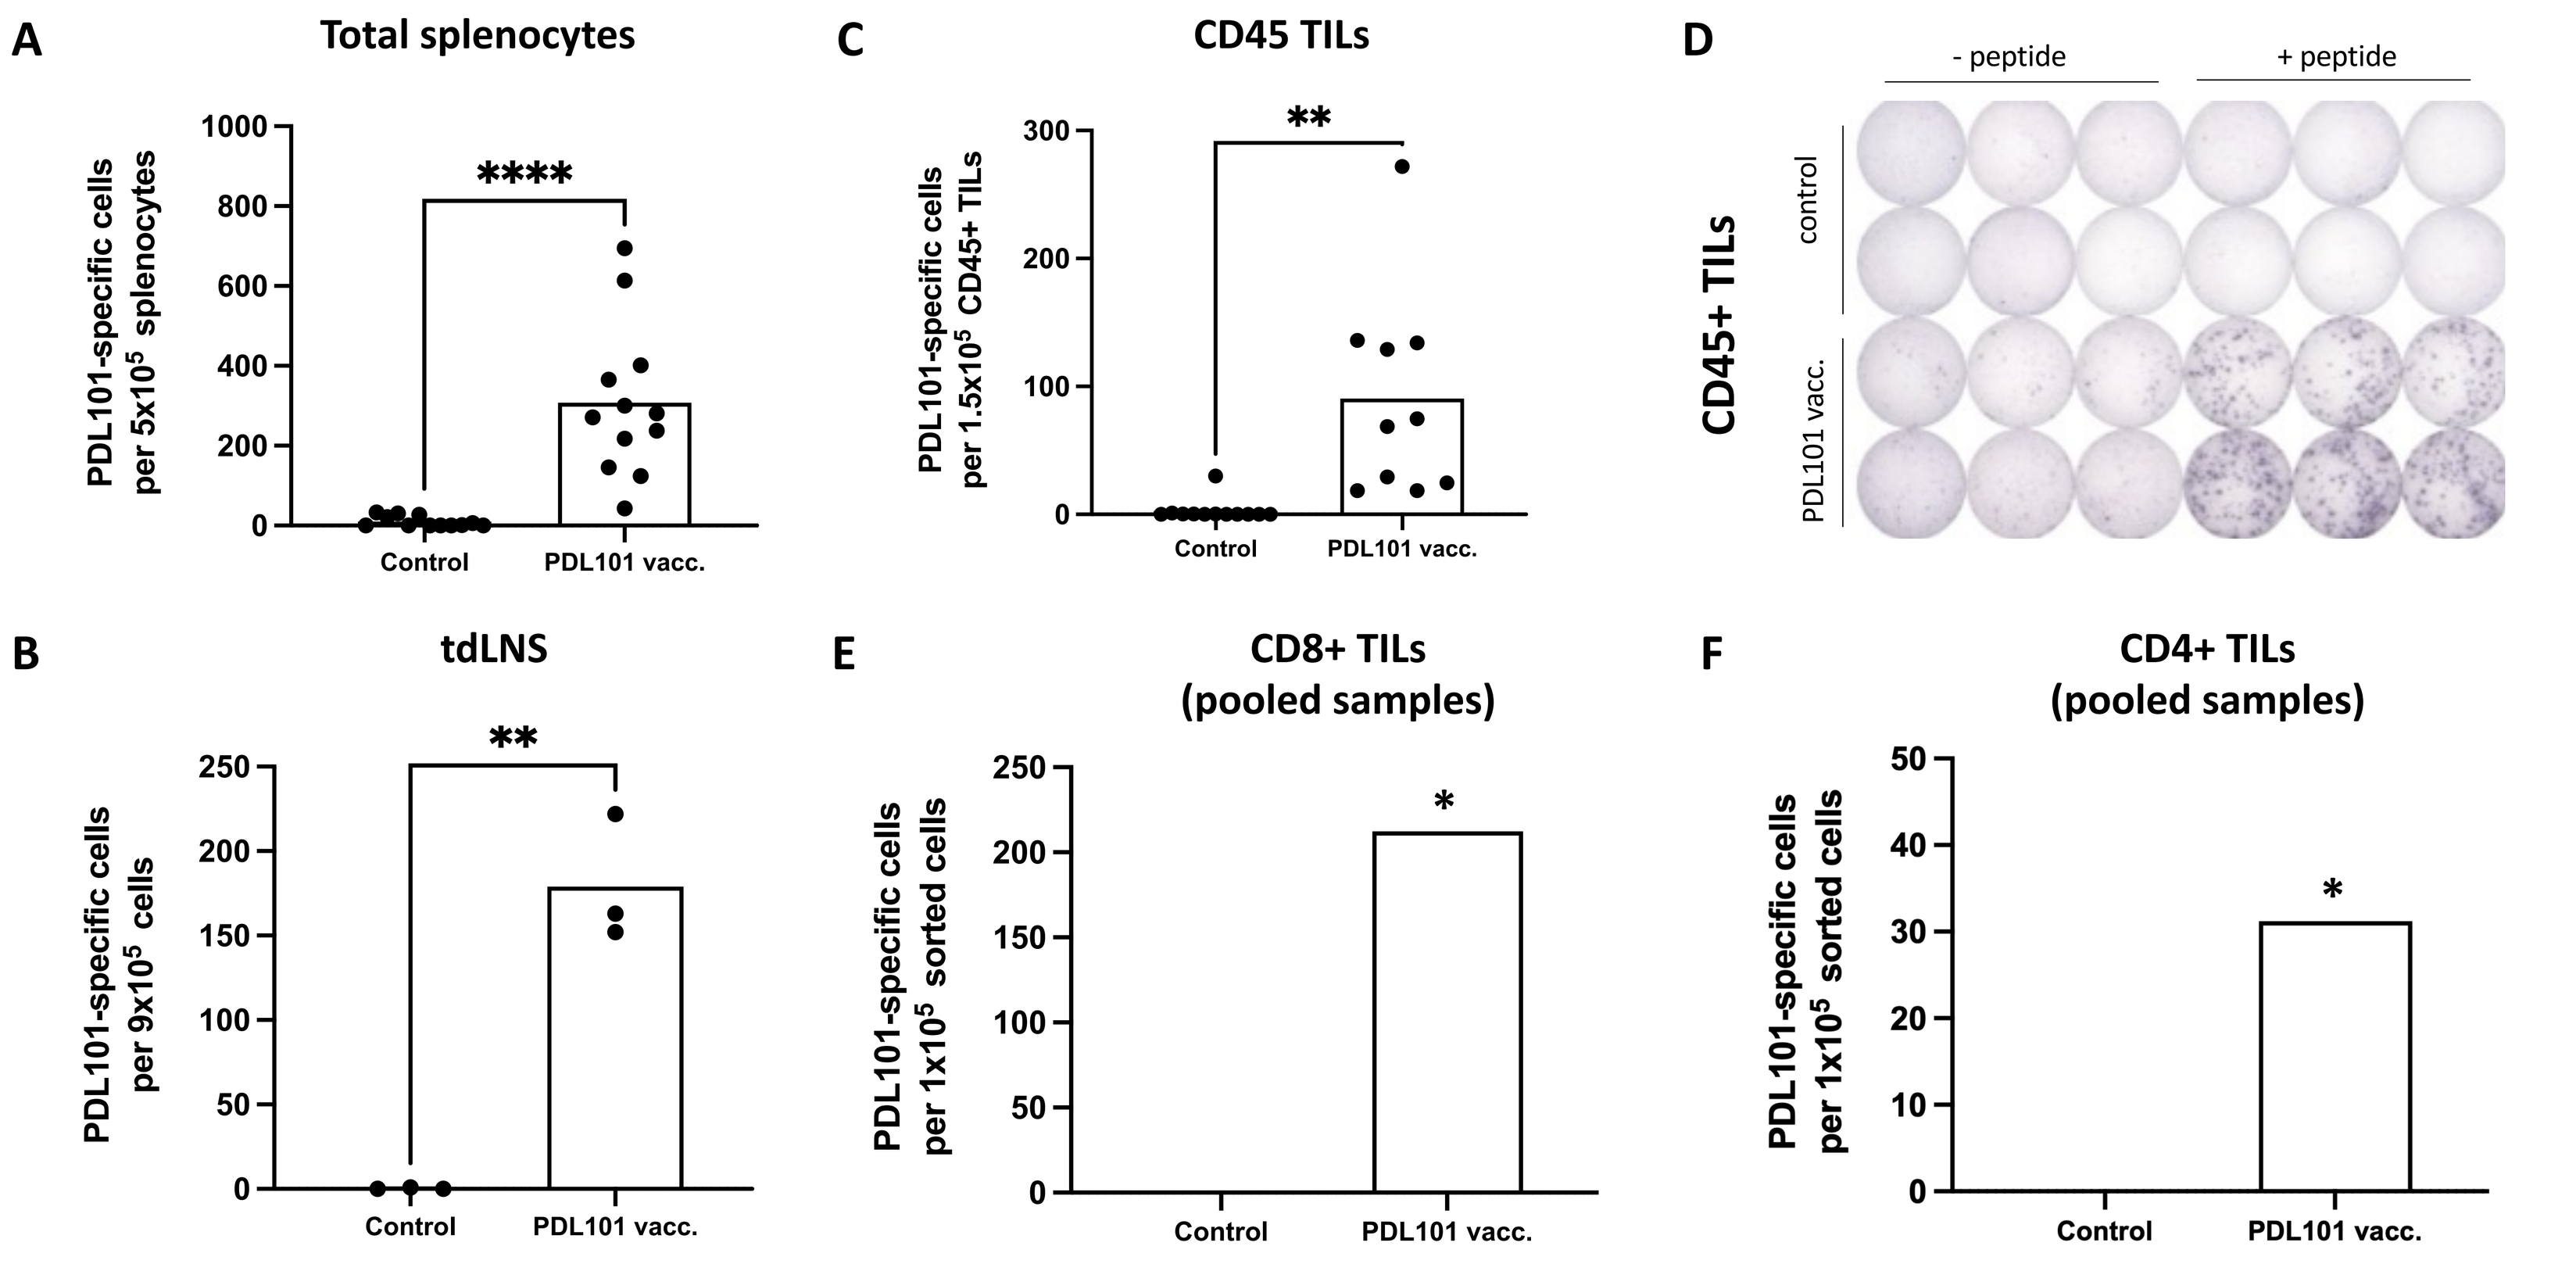
A
Total splenocytes
PDL101-specific cells per 5x10⁵ splenocytes
B
tdLNS
PDL101-specific cells per 9x10⁵ cells
C
CD45 TILs
PDL101-specific cells per 1.5x10⁵ CD45+ TILs
D
- peptide
+ peptide
control
PDL101 vacc.
CD45+ TILs
E
CD8+ TILs
(pooled samples)
F
CD4+ TILs
(pooled samples)
PDL101-specific cells per 1x10⁵ sorted cells
Control
PDL101 vacc.

The presence of PD-L1 specific cells was verified by IFNγ Elispot assays on (A) total splenocytes, (B) tumor draining lymph node cells (tdLNs), (C-D) CD45+ TILs, (E) CD4+ TILs and (F) CD8+ TILs collected from MC38-bearing animals untreated or treated with PD-L1 peptide. Data shown as mean +/- SEM.

T cells from PD-L1 vaccinated animals target PD-L1+ cells

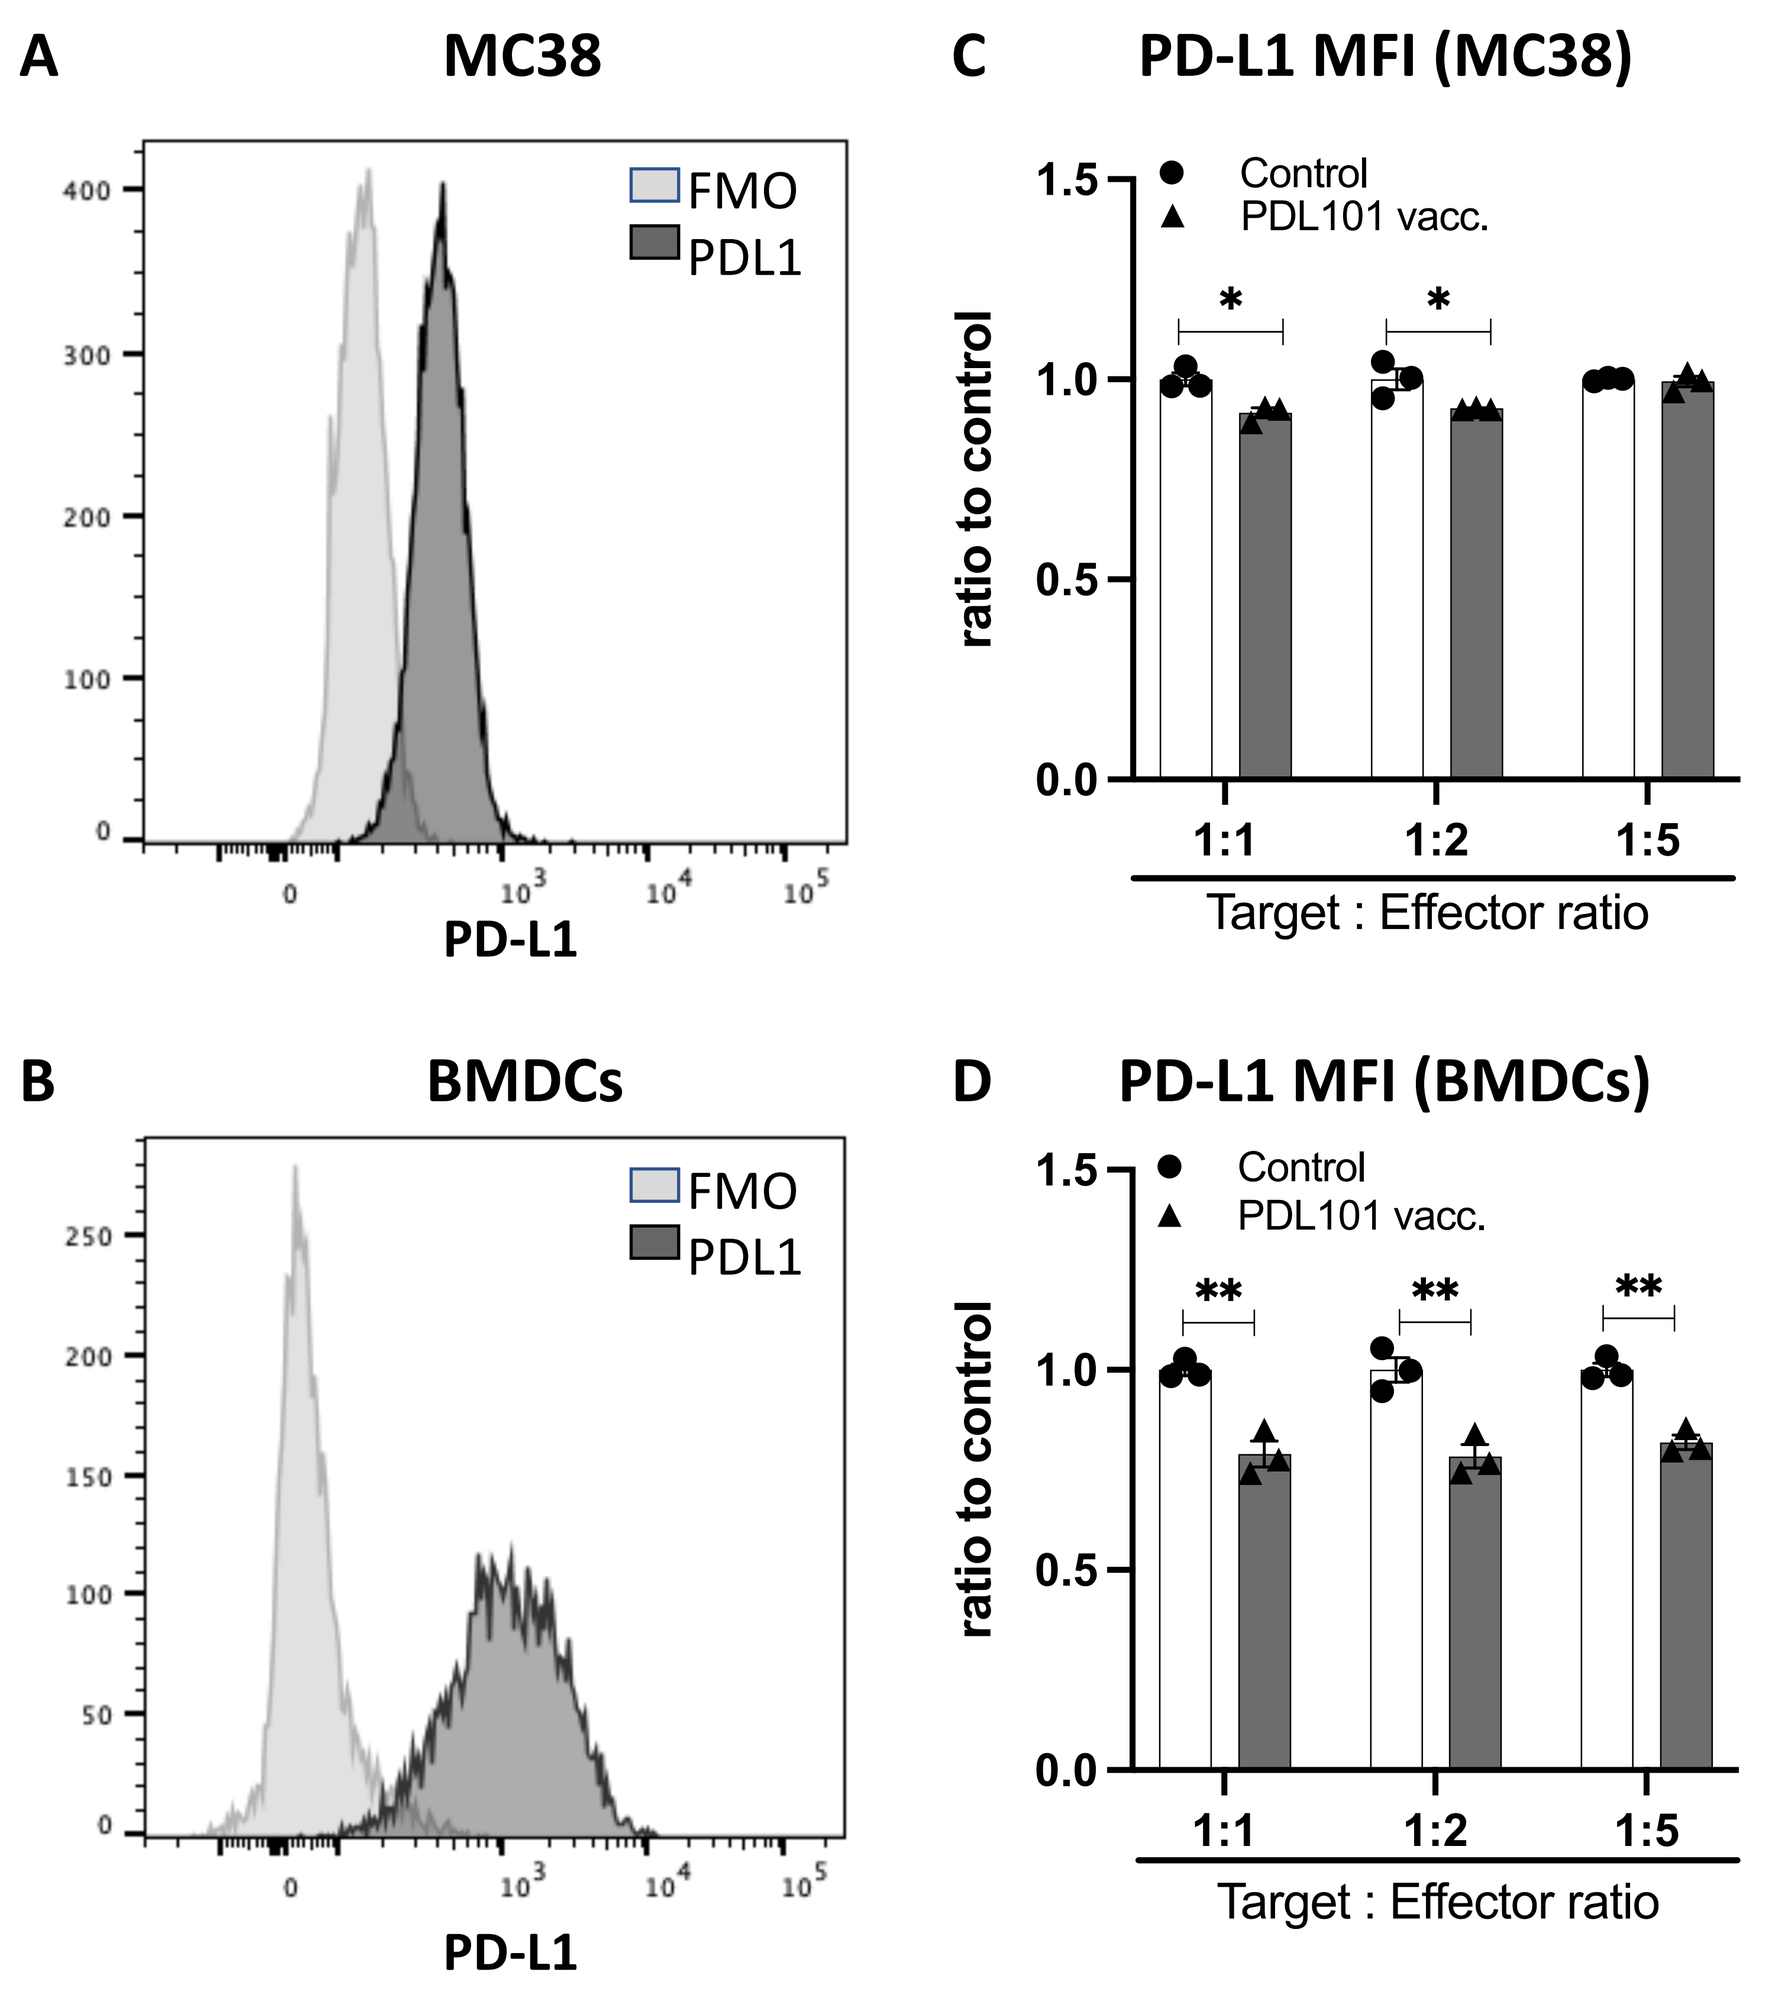
A
MC38
FMO
PDL1
PD-L1
B
BMDCs
C
PD-L1 MFI (MC38)
D
PD-L1 MFI (BMDCs)
Control
PDL101 vacc.
ratio to control
1:1
1:2
1:5
Target : Effector ratio

(A) PD-L1 expressing cells (MC38 cells or murine BMDCs) were labelled with cell trace (Targets) and co-cultured overnight with splenocytes from control or PD-L1 vaccinated animals (Effectors). After overnight culture, PD-L1 expression was assessed within Cell trace$^+$ target population. (C-D) % of PD-L1 positive within Cell trace$^+$ populations normalized to average control value as mean +/- SEM.

Combined PD-L1 and IDO1 vaccination enhances anti-tumor effect

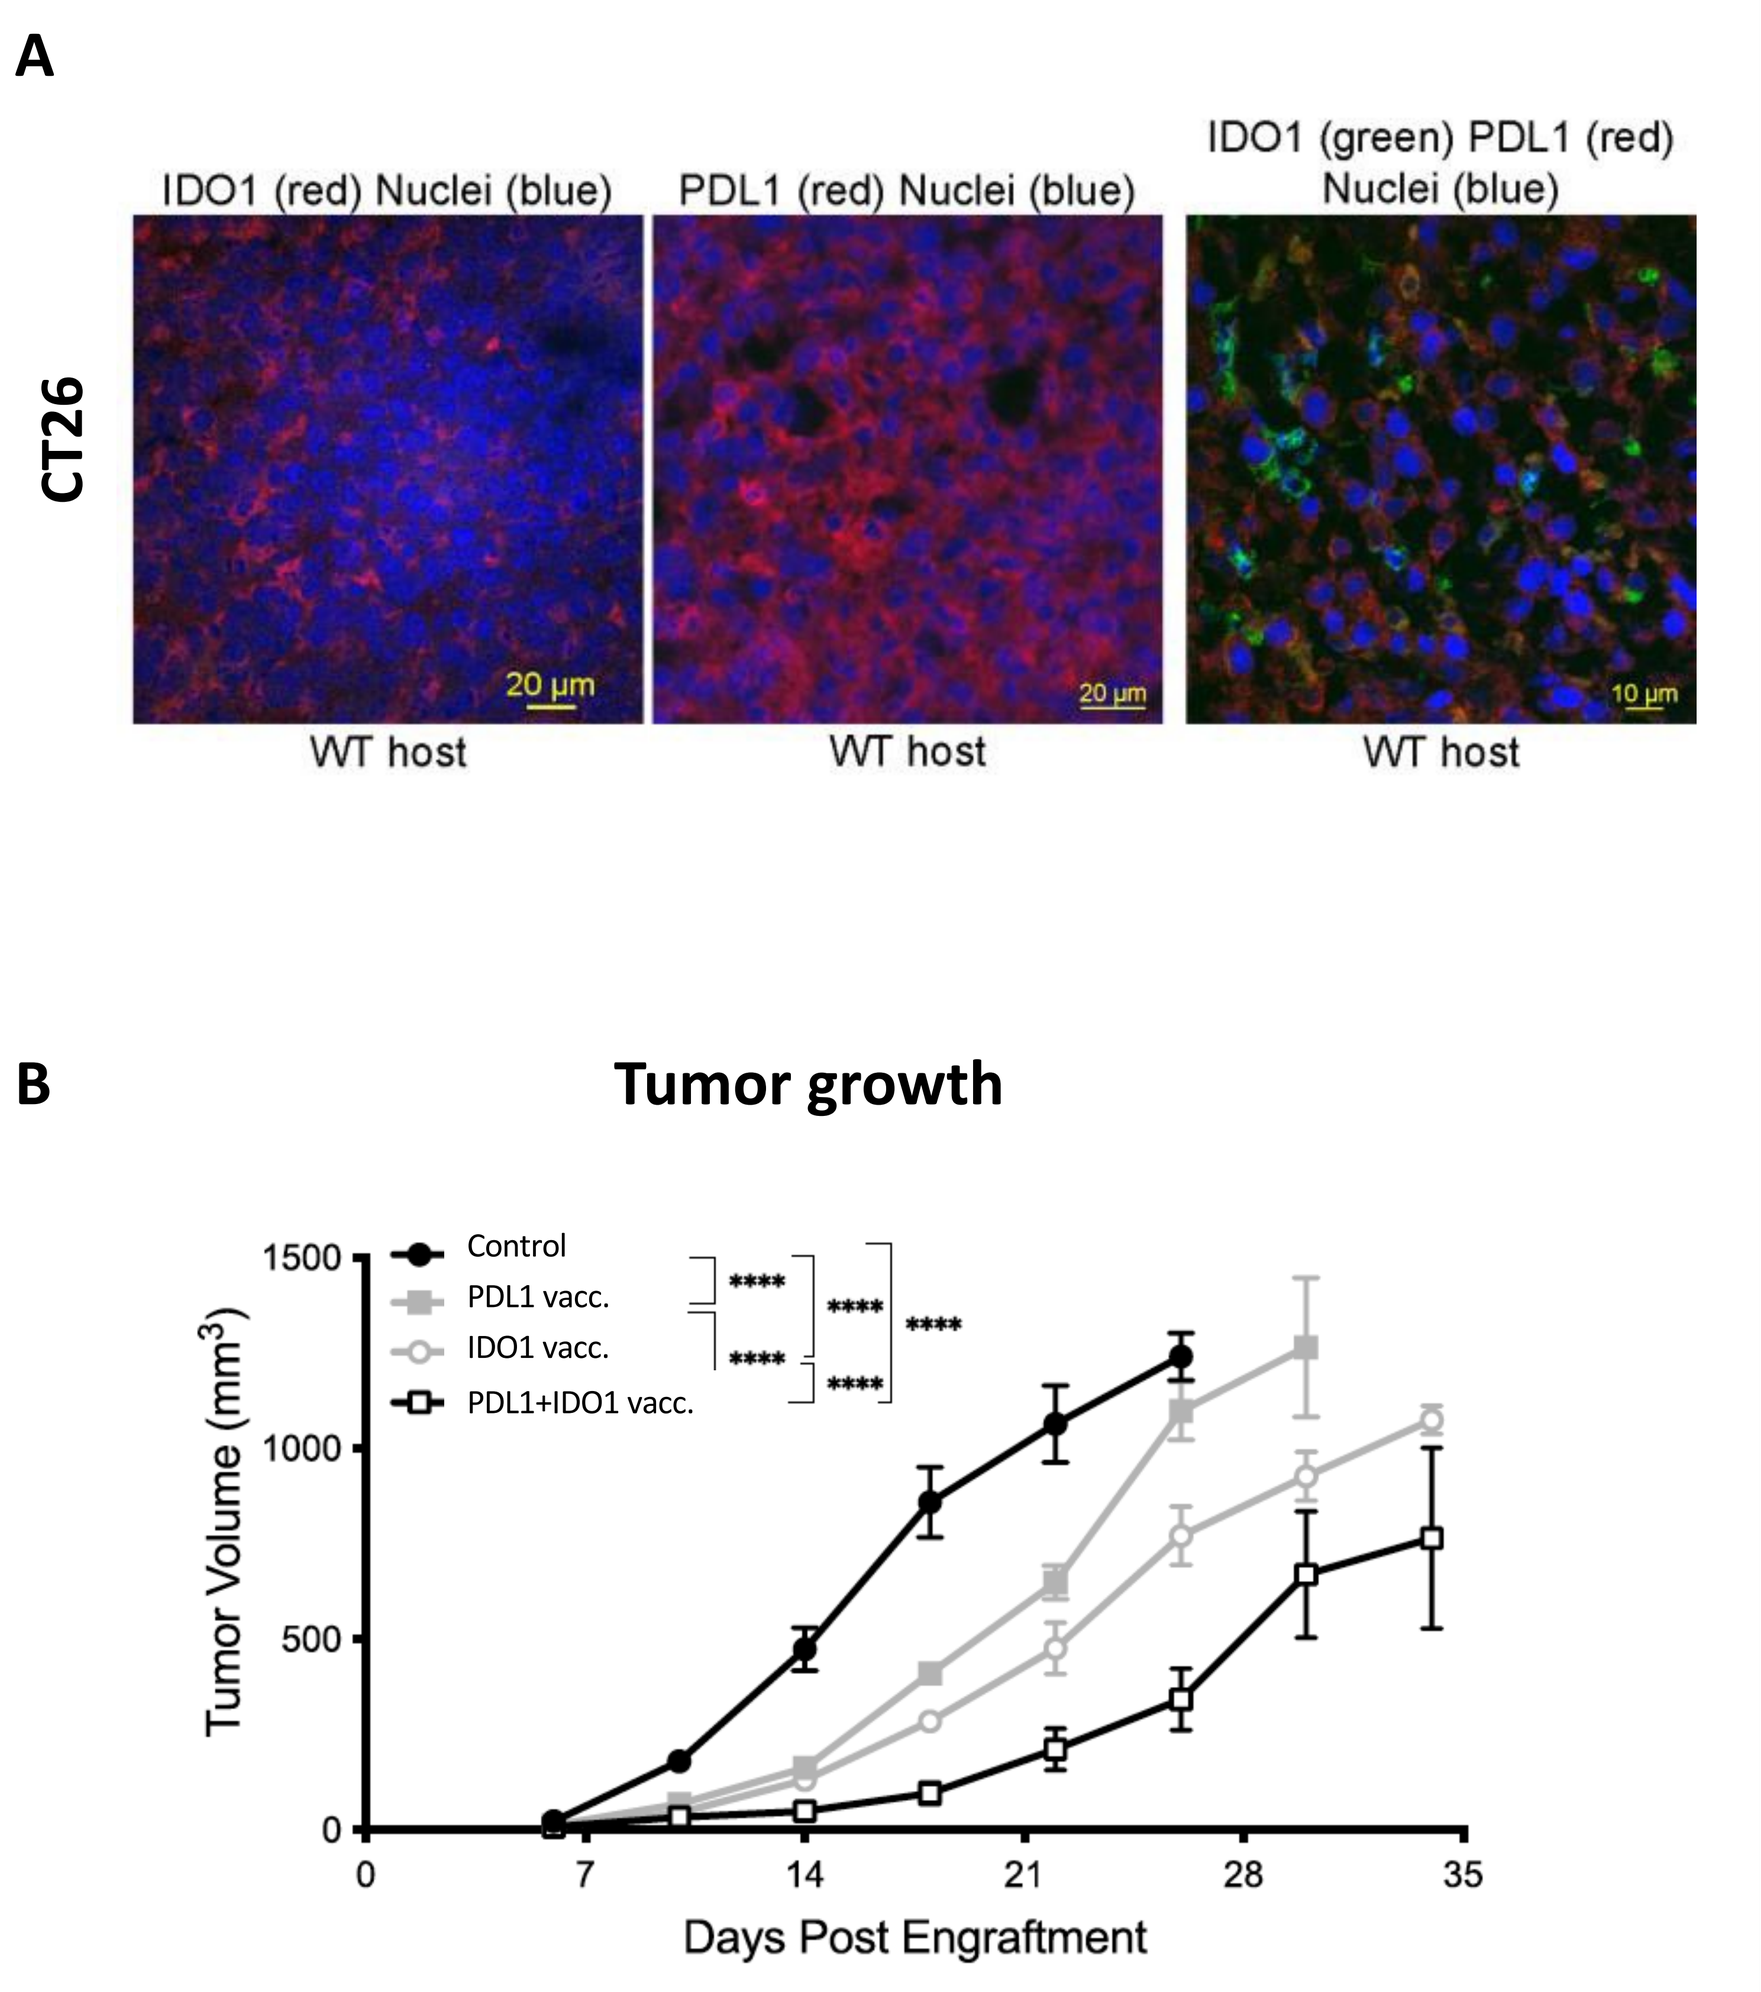
A
CT26
IDO1 (red) Nuclei (blue)
PDL1 (red) Nuclei (blue)
IDO1 (green) PDL1 (red) Nuclei (blue)
WT host
B
Tumor growth
Control
PDL1 vacc.
IDO1 vacc.
PDL1+IDO1 vacc.
Tumor Volume (mm³)
Days Post Engraftment

(A) IDO1 and PD-L1 were found to be expressed by different cells in within CT26 tumors analyzed per immunofluorescence. (B) BALB/c animals were inoculated with CT26 tumor cells and treated with PD-L1 peptide alone or combined to IDO1 peptide and tumor growth was analyzed. Data shown as mean value +/- SEM.

PD-L1 and IDO1 vaccine induce distinct molecular changes at tumor site

Nanostring gene expression analysis in MC38

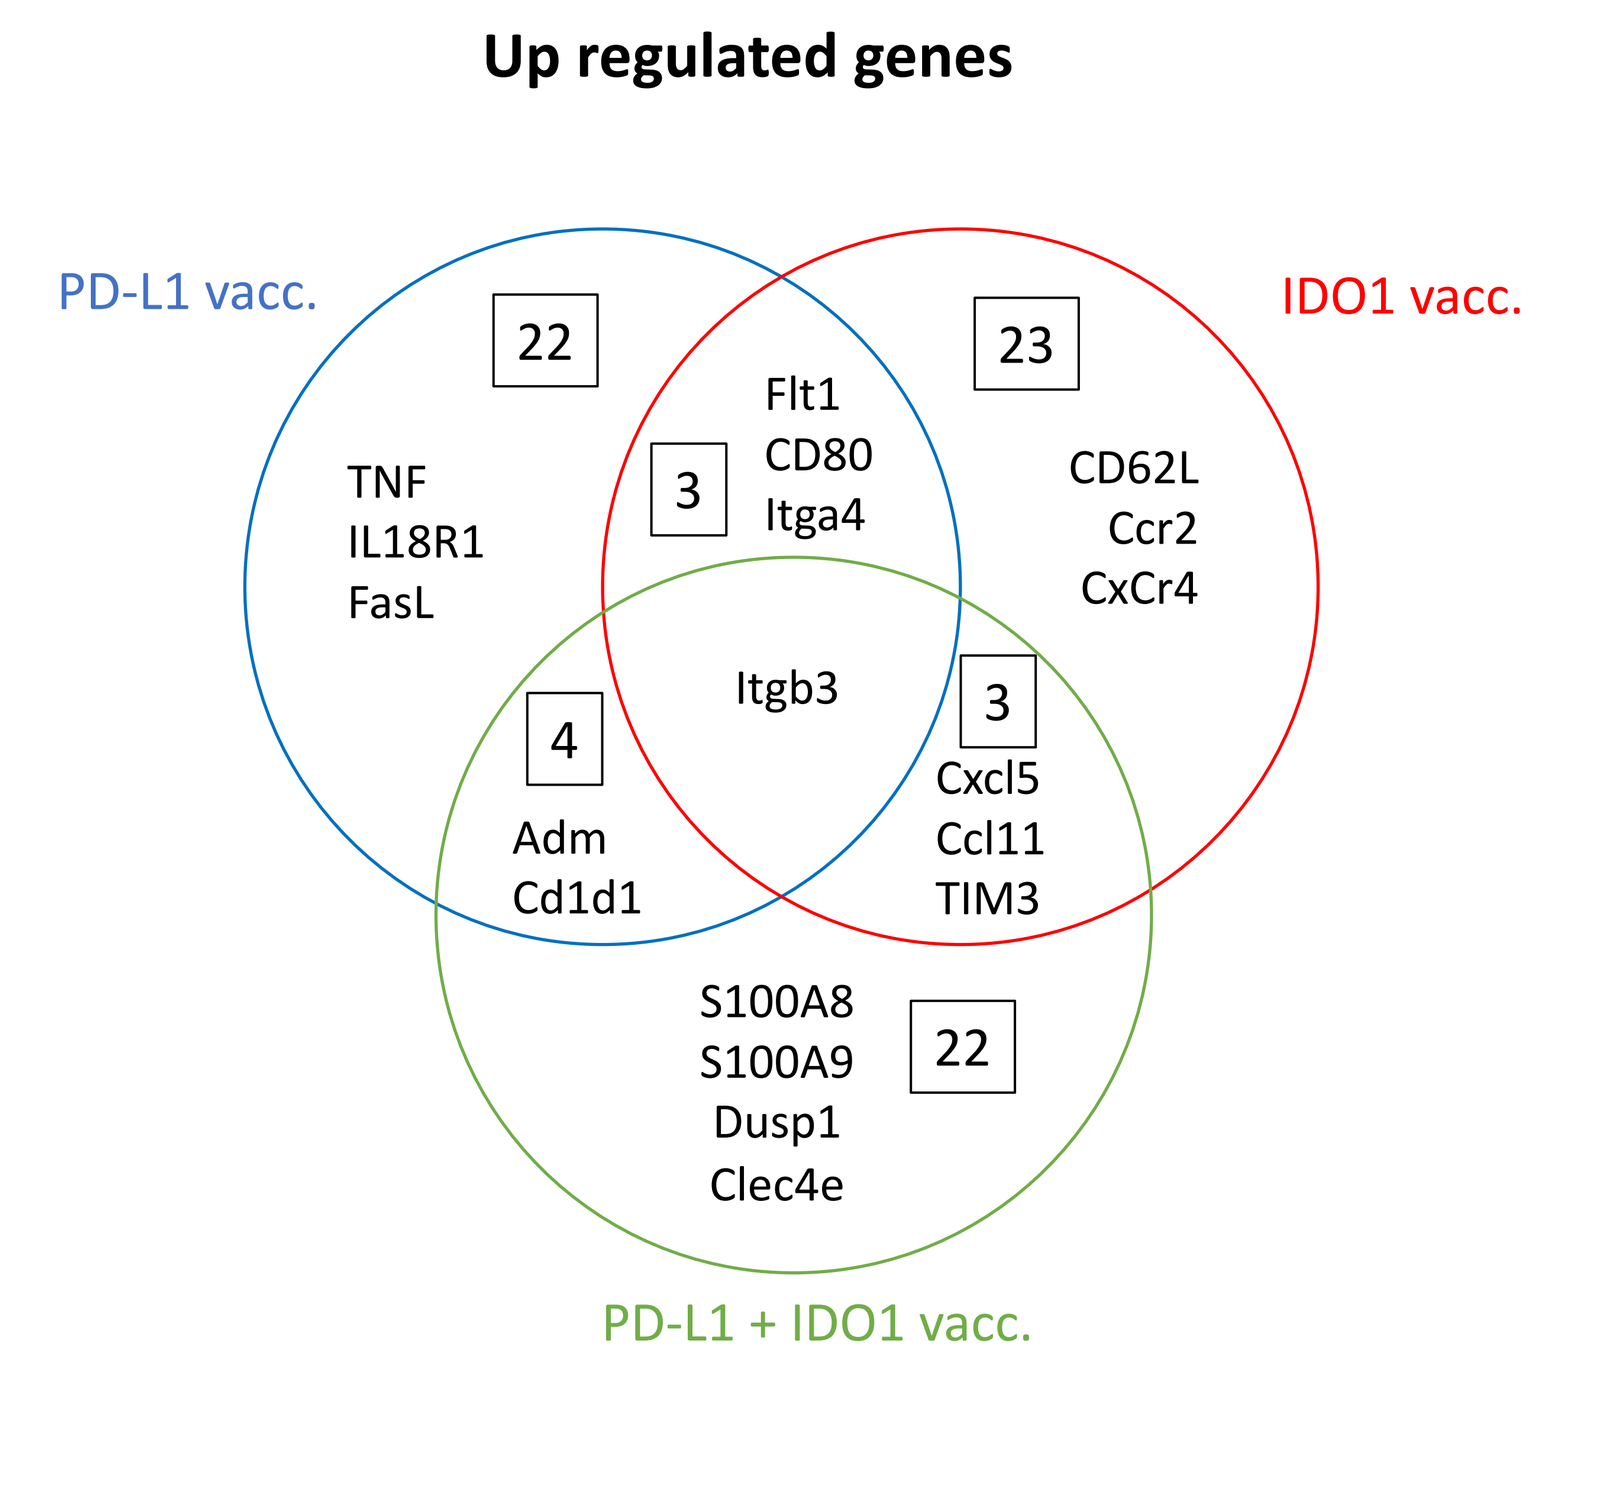
Up regulated genes
PD-L1 vacc.
IDO1 vacc.
PD-L1 + IDO1 vacc.
22
TNF
IL18R1
FasL
3
Flt1
CD80
Itga4
23
CD62L
Ccr2
CxCr4
Itgb3
4
Adm
Cd1d1
3
Cxcl5
Ccl11
TIM3
22
S100A8
S100A9
Dusp1
Clec4e

Gene expression analysis was performed using Nanostring nCounter PanCancer IO360 Panel on MC38 tumors from animals treated with PD-L1, IDO1 or combined vaccine. Differentially upregulated genes were identified in the different treatment groups compared to untreated control samples. Lists of top30 differentially upregulated genes from each treatment group were then compared to identify specific and shared molecular changes.

Conclusion

The effect of a peptide-based treatment against PD-L1 was evaluated in two PD-L1+ murine tumor models. A delay in tumor growth was observed in both MC38 and CT26 tumor models upon treatment with PD-L1 peptides. We here highlight that PD-L1 vaccine-induced T cells localize to the TME where they target PD-L1 expressing cells thus reducing the immunosuppression at tumor site and leading to decreased tumor growth as observed in two different models.

A similar peptide-based vaccine against IDO1 previously showed anti-tumor activity and changes in the tumor microenvironment[3]. Combination of PD-L1 and IDO1 vaccines showed enhanced effect *in vivo* in CT26 model where IDO1 and PD-L1 were found expressed by different target populations. Molecular analysis showed distinct gene expression profiles for each monotherapy alone or combined.

Our data support further development of peptide-based vaccines as therapeutic strategies against immuno-suppressive targets in the TME for the treatment of a wide range of tumors.

Contact

Marion Chapellier, PhD
Senior Scientist
IO Biotech, Denmark
mc@iobiotech.com

References

1. Munir S, Andersen GH, Met Ö, Donia M, Frøsig TM, Larsen SK, Klausen TW, Svane IM, Andersen MH. HLA-restricted CTL that are specific for the immune checkpoint ligand PD-L1 occur with high frequency in cancer patients. Cancer Res. 2013 Mar 15;73(6):1764-76. doi: 10.1158/0008-5472.CAN-12-3507. Epub 2013 Jan 17. PMID: 23328583.
2. Munir S, Andersen GH, Svane IM, Andersen MH. The immune checkpoint regulator PD-L1 is a specific target for naturally occurring CD4 T cells. Oncoimmunology. 2013 Apr 1;2(4):e23991. doi: 10.4161/onci.23991. PMID: 23734334; PMCID: PMC3654604.
3. Dey S, Sutanto-Ward E, Kopp KL, DuHadaway J, Mondal A, Ghaban D, Lecoq I, Zocca MB, Merlo LMF, Mandik-Nayak L, Andersen MH, Pedersen AW, Muller AJ. Peptide vaccination directed against IDO1-expressing immune cells elicits CD8$^+$ and CD4$^+$ T-cell-mediated antitumor immunity and enhanced anti-PD1 responses. J Immunother Cancer. 2020 Jul;8(2):e000605. doi: 10.1136/jitc-2020-000605. PMID: 32690770; PMCID: PMC7373332.