T Cell Receptor Diversity Analysis of *in vitro*-Expanded T Cells Against IDO1 and PD-L1-Derived Peptides

Preeyam Patel[1], Marcos Iglesias[1], Qin Tang[1], Megan Biller[1], Alireza Alavi, Ayako Wakatsuki Pedersen[1]
[1]IO Biotech, Copenhagen, Denmark

Abstract #1029

Abstract:

Background:
Recognition of cognate peptides results in expansion of antigen-specific T cells. Since humans have ~10^{15} TCRs, identification of responding TCRs has remained difficult. The variable (V) region gene segment, which comprises one of the components of the CDR3 loop of the TCRβ chain, determines TCR uniqueness. IO Biotech is pioneering development of a novel immunomodulatory vaccine, with our lead candidate IO102-IO103, that is designed to activate and expand T cells against IDO1+ and PD-L1+ cells, respectively. In this study we explore TCR diversity analysis as a potential platform to characterize vaccine-expanded T cells.

Methods:
Human PBMCs from healthy subjects were cultured with IO102 (IDO1 peptide) or IO103 (PD-L1 peptide). Samples were collected 6 days following culture, and 24 hours following restimulation with peptide at day 8. Control samples included PBMCs prior to culture, samples cultured without peptide, and restimulation without peptide. ELISPOT assay was conducted in tandem. Following extraction of RNA, samples were analyzed on the Nanostring nCounter Analysis System using the TCR diversity panel kit. Using the nSolver Analysis Software, data were quality controlled and normalized prior to analysis of TCR variable regions. Rosalind Software was used to calculate TCR Score to assess diversity. Changes detected in T cell phenotype by NanoString were validated using flow cytometry.

Results:
Exposure to and restimulation with IO102 or IO103 peptides in short-term *in vitro* cultures resulted in expansion of a specific and narrow set of TCR variable regions. Expansion of TCRV genes were mostly represented by TRAV and TRBV, with limited expansion of TRGV and TRDV. Six-day culture with peptide resulted in expansion of certain TCRVs that were distinctly and further expanded following restimulation with the same peptide. Uniqueness of TCRV usage was determined between peptides and donors. Expansion of peptide-responsive T cells further impacted TCR clonality and diversity. Expanded TCRV regions were correlated to T cell phenotypes and associated with markers of activation and memory.

Conclusions:
Our results demonstrate that exposure to IO102 or IO103 in short-term *in vitro* cultures results in a discernable impact to the TCR repertoire. Further, we were able to identify TCRV segments that are responsive to either IO102 or IO103 peptides in a donor-specific manner. These studies will allow us to further characterize peptide-specific T cell responses that arise following vaccination. We are working to pair TCRV expansion with functional cytokine assays and spatial transcriptomics in tumor microenvironments for personalizing monitoring of peptide vaccine responsiveness.

Background:

T Cell Receptor V(D)J Arrangement of the Alpha and Beta Chains

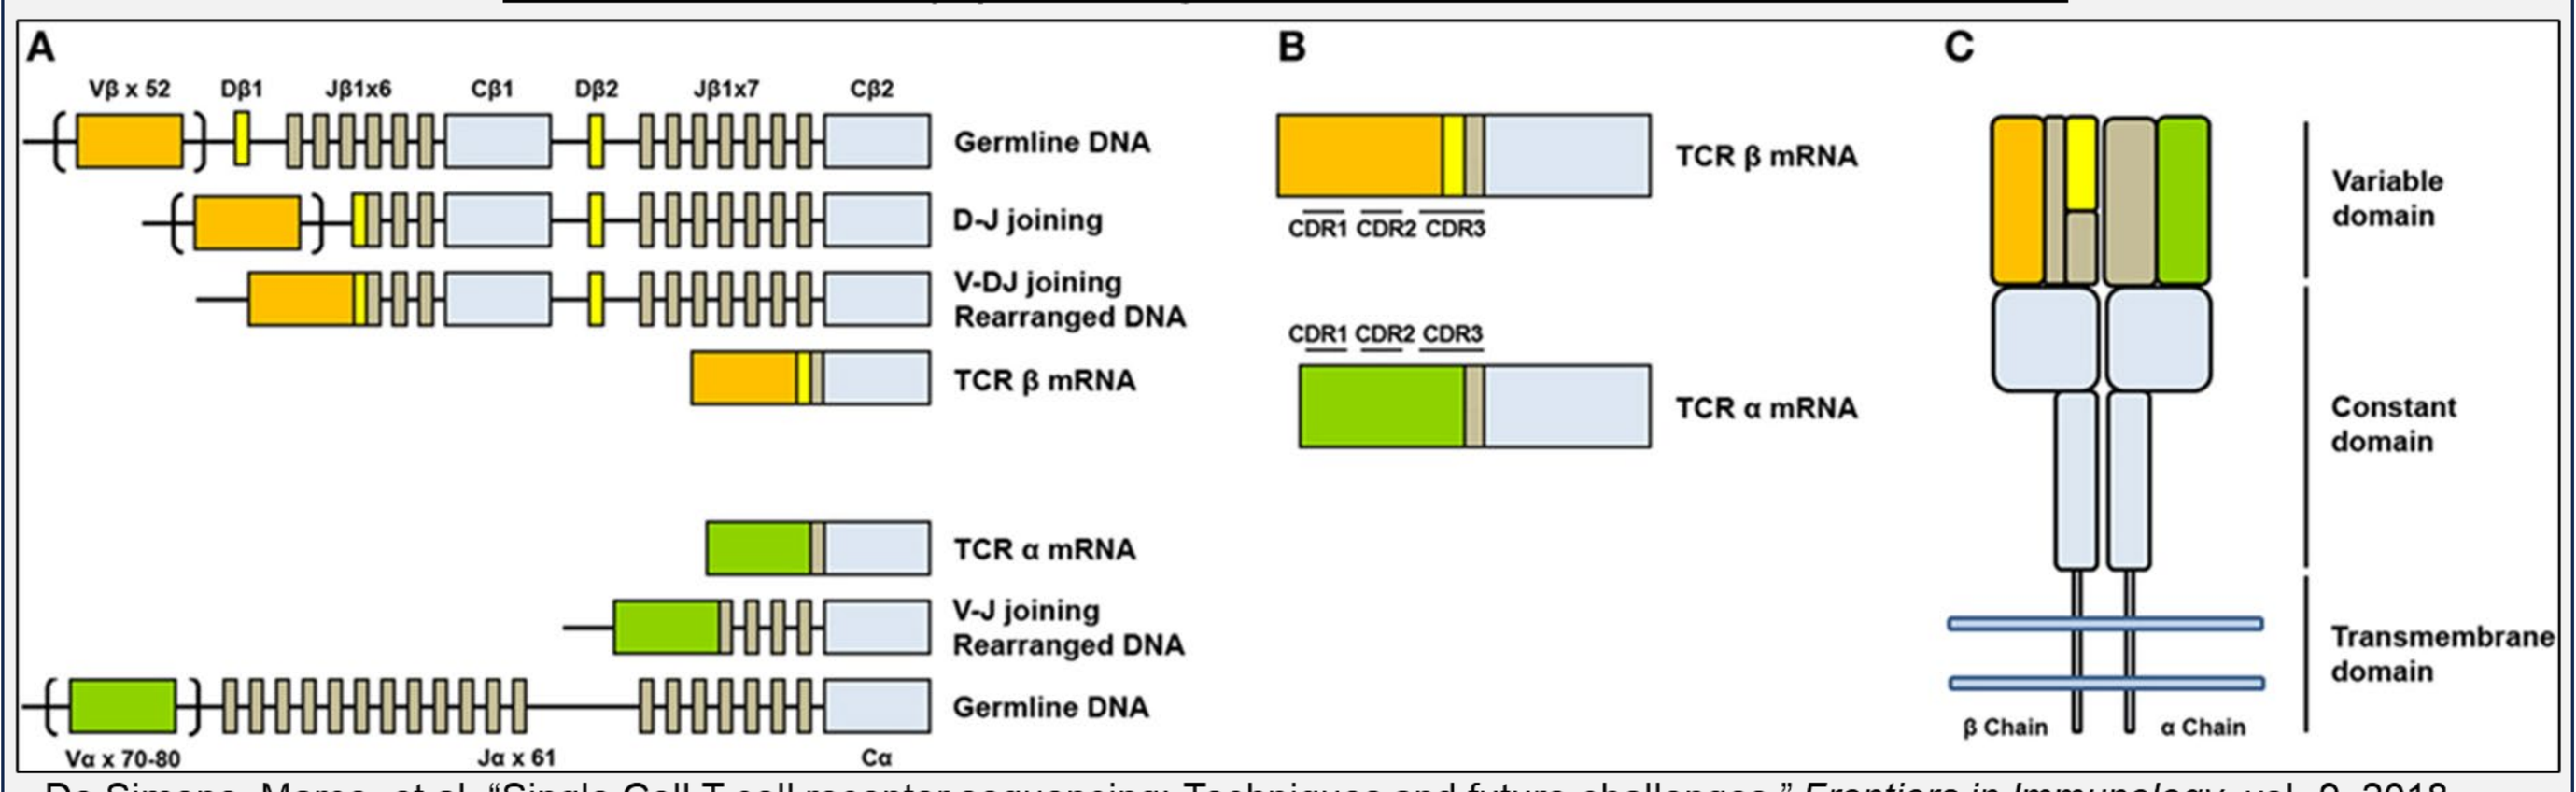
A
Germline DNA
D-J joining
V-DJ joining Rearranged DNA
TCR β mRNA
TCR α mRNA
V-J joining Rearranged DNA
Germline DNA
B
TCR β mRNA
TCR α mRNA
CDR1 CDR2 CDR3
C
Variable domain
Constant domain
Transmembrane domain
β Chain
α Chain

De Simone, Marco, et al. "Single Cell T cell receptor sequencing: Techniques and future challenges." *Frontiers in Immunology*, vol. 9, 2018, https://doi.org/10.3389/fimmu.2018.01638.

nCounter Workflow for Custom Gene Expression Assays

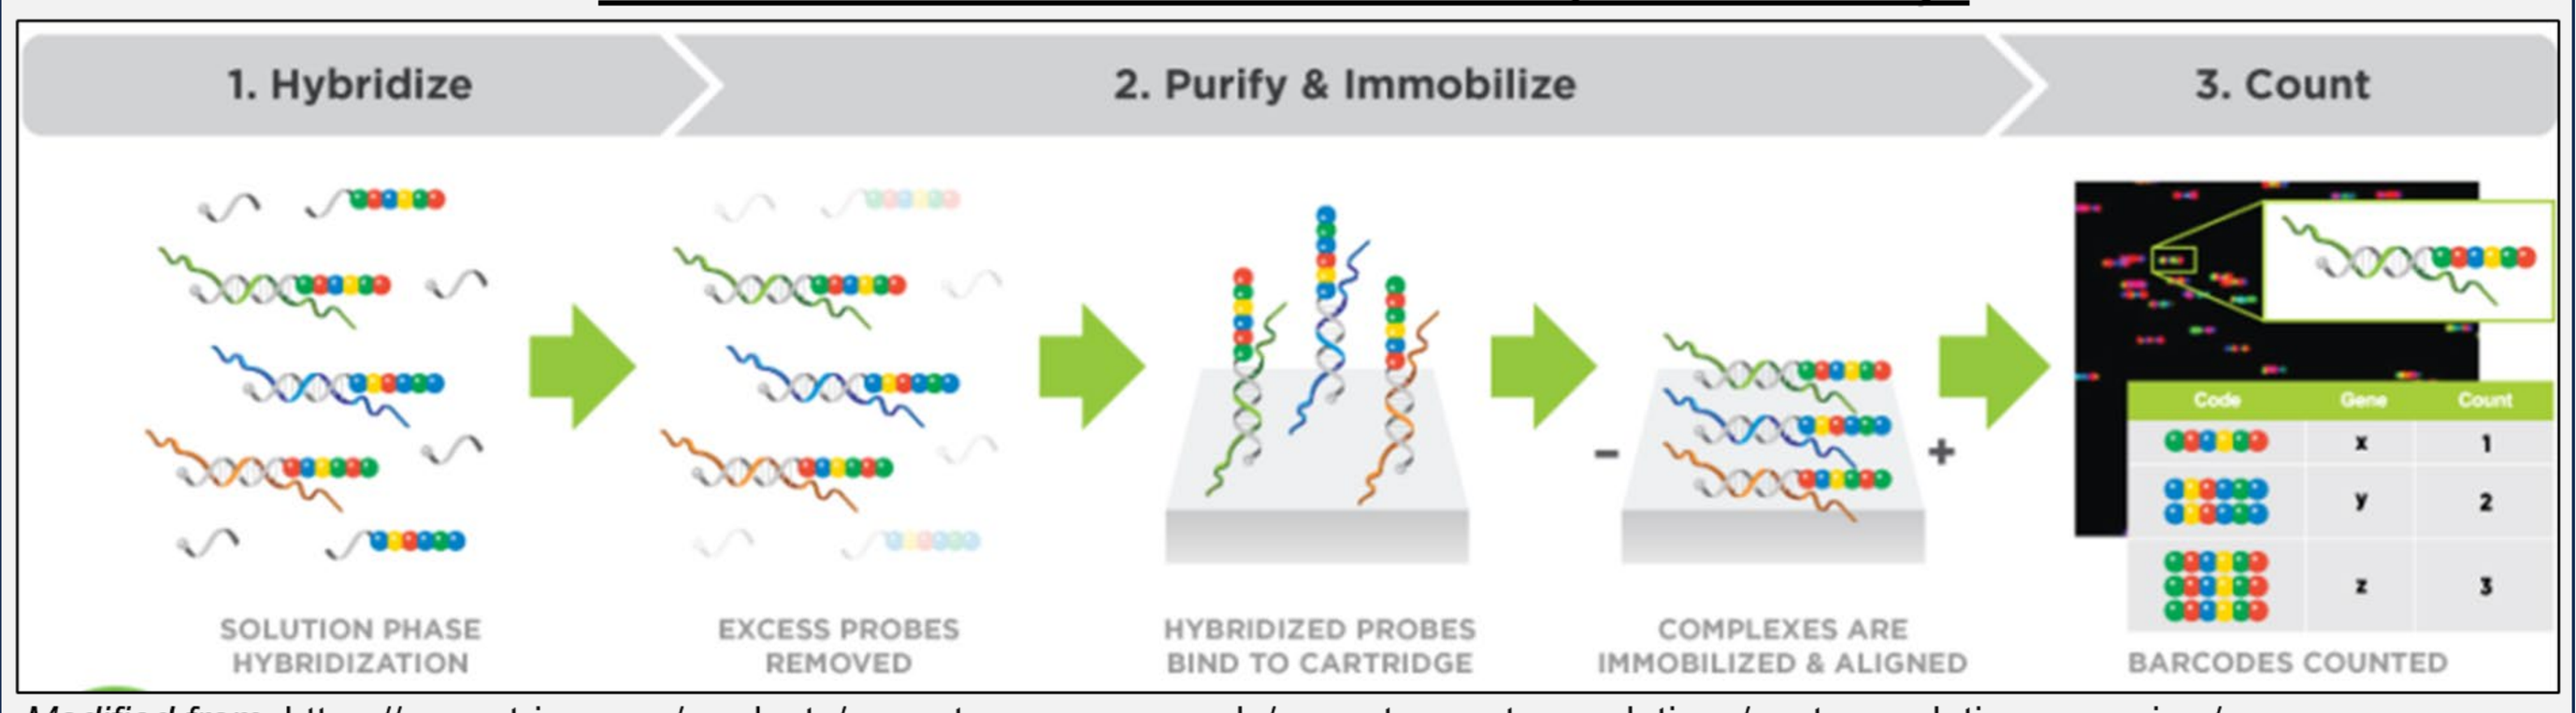
1. Hybridize
2. Purify & Immobilize
3. Count
SOLUTION PHASE HYBRIDIZATION
EXCESS PROBES REMOVED
HYBRIDIZED PROBES BIND TO CARTRIDGE
COMPLEXES ARE IMMOBILIZED & ALIGNED
BARCODES COUNTED

Modified from: https://nanostring.com/products/ncounter-assays-panels/ncounter-custom-solutions/custom-solutions-overview/

Methods:

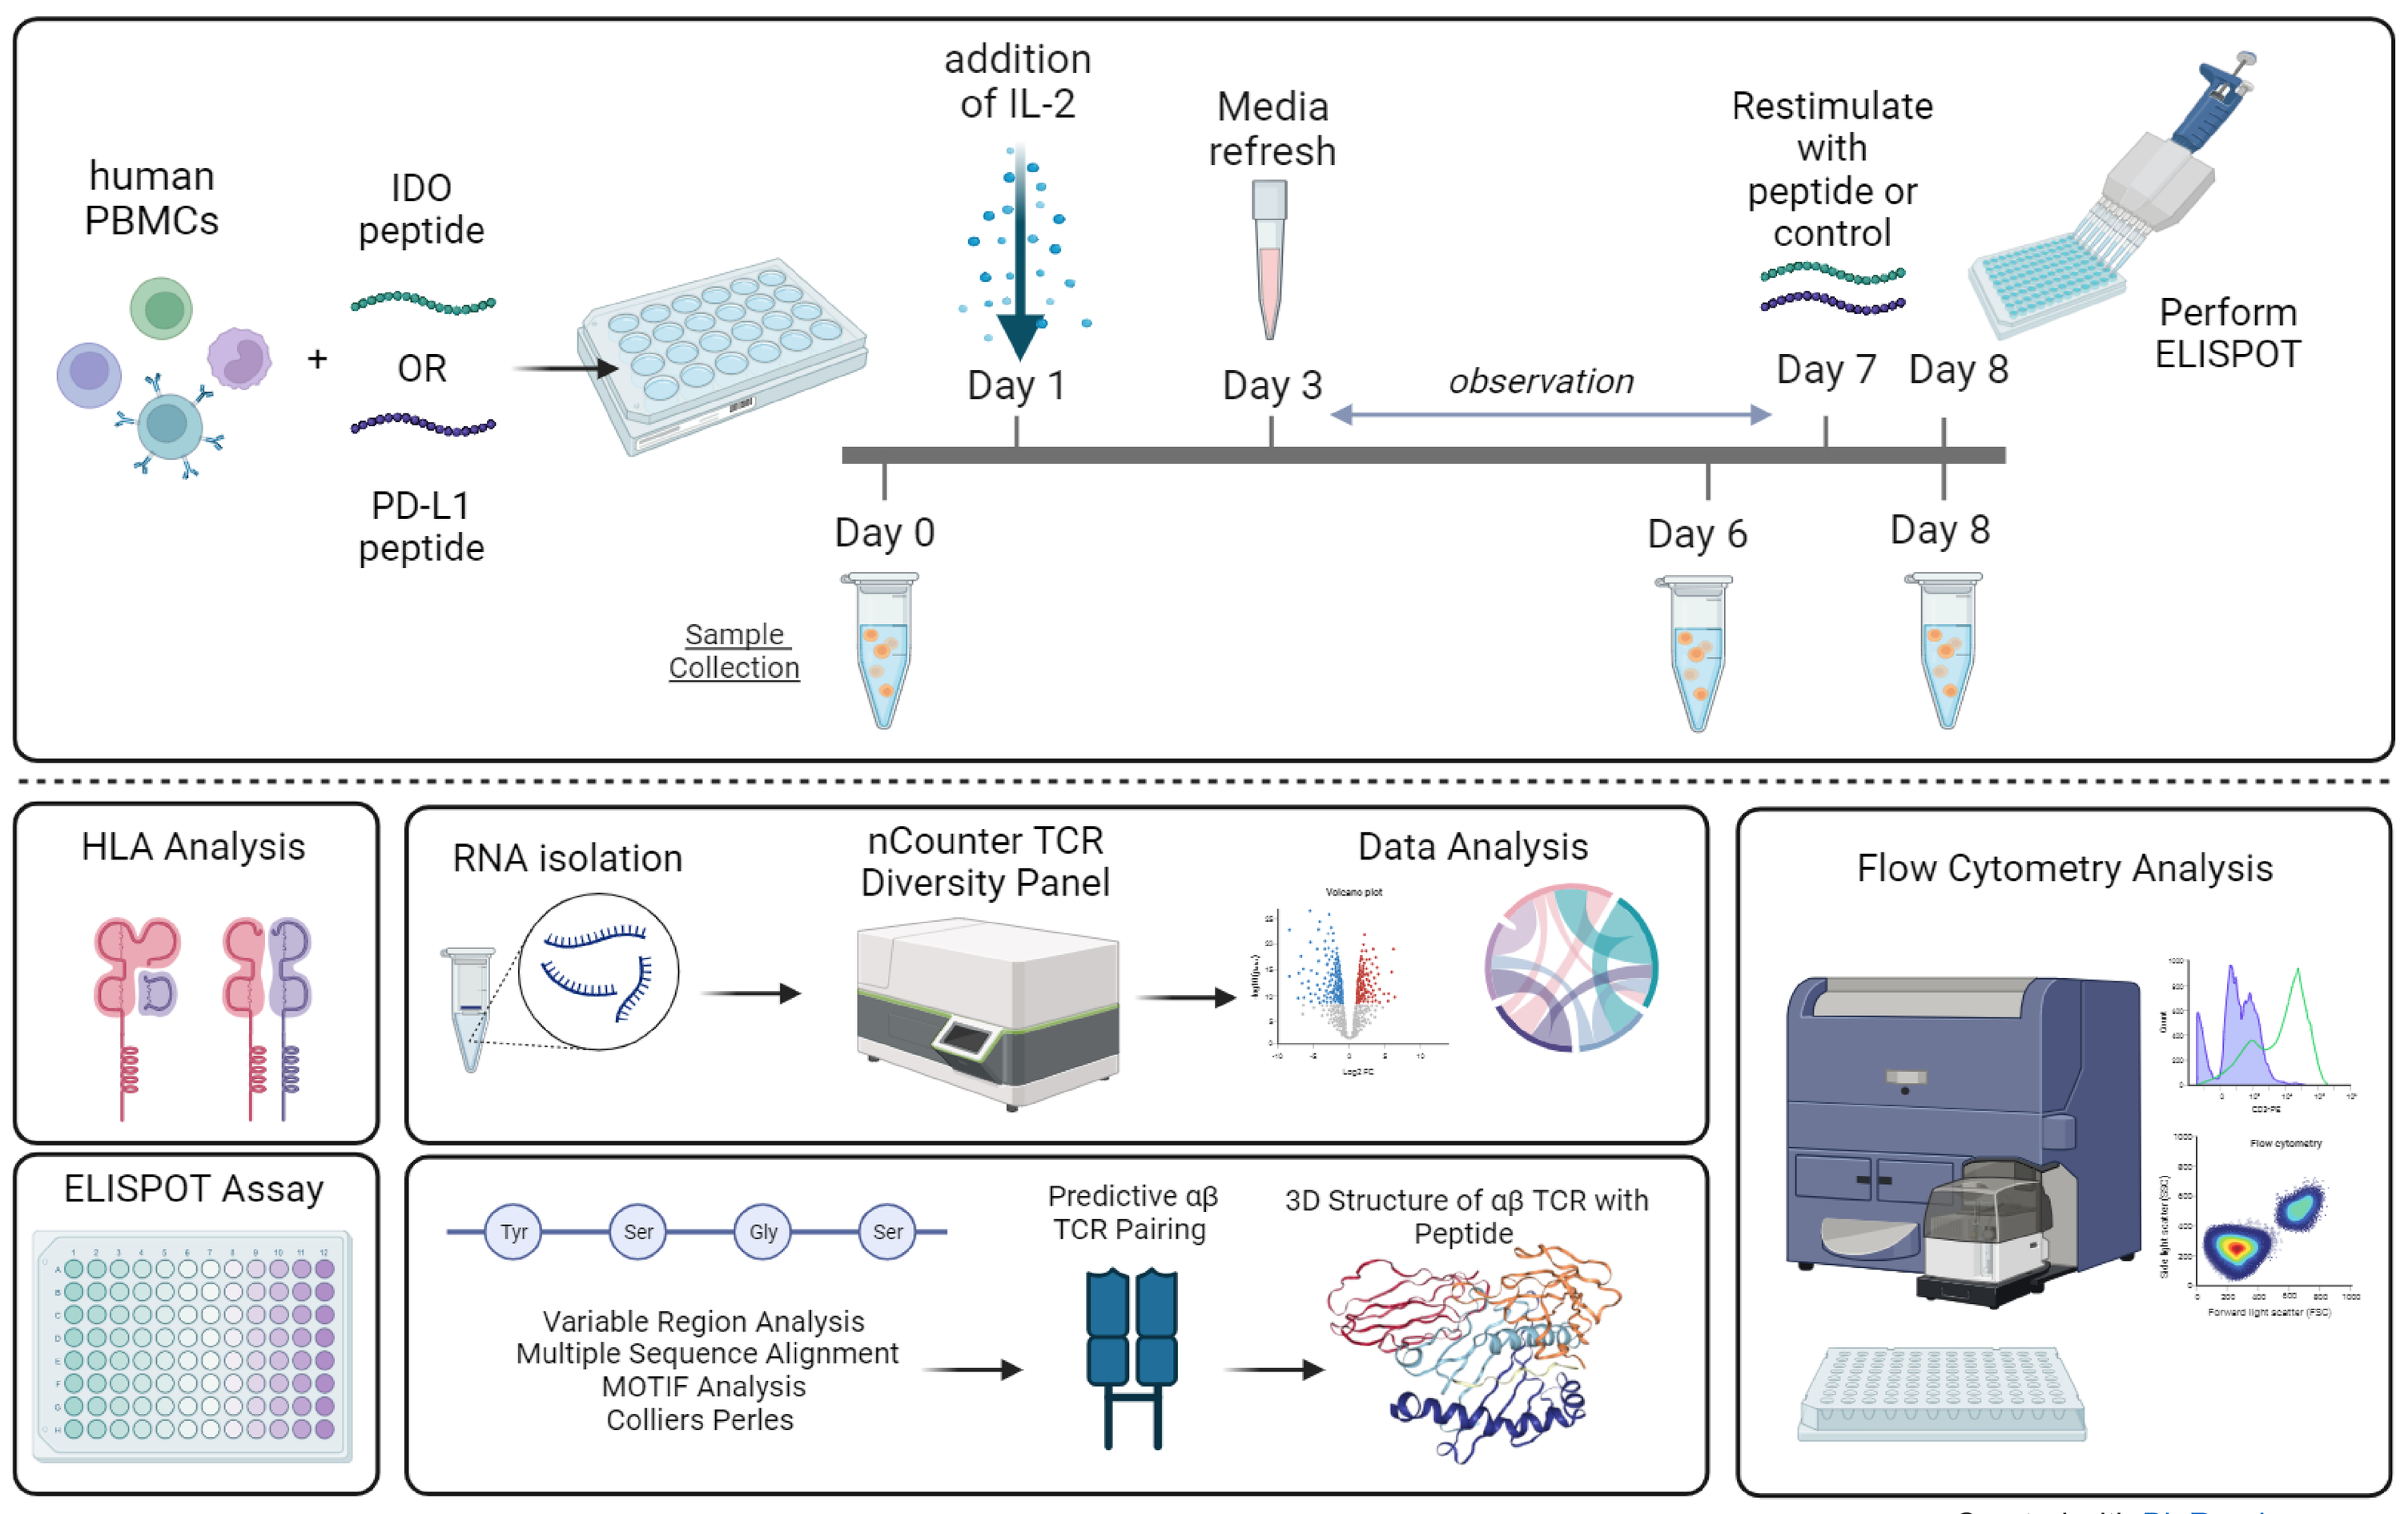
human PBMCs
IDO peptide
OR
PD-L1 peptide
addition of IL-2
Media refresh
Restimulate with peptide or control
Perform ELISPOT
Day 1
Day 3
observation
Day 7
Day 8
Day 0
Day 6
Day 8
Sample Collection
HLA Analysis
RNA isolation
nCounter TCR Diversity Panel
Data Analysis
Flow Cytometry Analysis
ELISPOT Assay
Variable Region Analysis
Multiple Sequence Alignment
MOTIF Analysis
Colliers Perles
Predictive αβ TCR Pairing
3D Structure of αβ TCR with Peptide

Created with BioRender.com

Data:

Human Leukocyte Antigen (HLA) Genes for Study PBMC Donors

	HLA-A Allele 1	HLA-A Allele 2	HLA-B Allele 1	HLA-B Allele 2	HLA-C Allele 1	HLA-C Allele 2	HLA-DPA1 Allele 1	HLA-DPA1 Allele 2	HLA-DPB1 Allele 1	HLA-DPB1 Allele 2	HLA-DQA1 Allele 1	HLA-DQA1 Allele 2	HLA-DQB1 Allele 1	HLA-DQB1 Allele 2	HLA-DRB1 Allele 1	HLA-DRB1 Allele 2	HLA-DRB345 Allele 1	HLA-DRB345 Allele 2
LKP	A*68:02:01	A*74:01:01	B*15:03:01	B*53:01:01	C*02:10:01	C*04:01:01	DPA1*02:01:08	DPA1*02:01:08	DPB1*01:01:01	DPB1*01:01:01	DQA1*01:02:01	DQA1*05:05:01	DQB1*03:01:01	DQB1*05:01:01	DRB1*13:02:01	DRB1*13:03:01	DRB3*01:01:02	DRB3*03:01:01
415	A*11:01:01	A*24:02:01	B*40:01:02	B*40:01:02	C*03:04:01	C*03:04:01	DPA1*01:03:01	DPA1*02:06	DPB1*03:01:01	DPB1*05:01:01	DQA1*01:02:01	DQA1*04:01:01	DQB1*04:02:01	DQB1*06:04:01	DRB1*08:01:01	DRB1*13:02:01	DRB3*03:01:01	DRB345*Not_Present

Results of ELISPOT Assay Following Culture with IO102 or IO103 Peptide

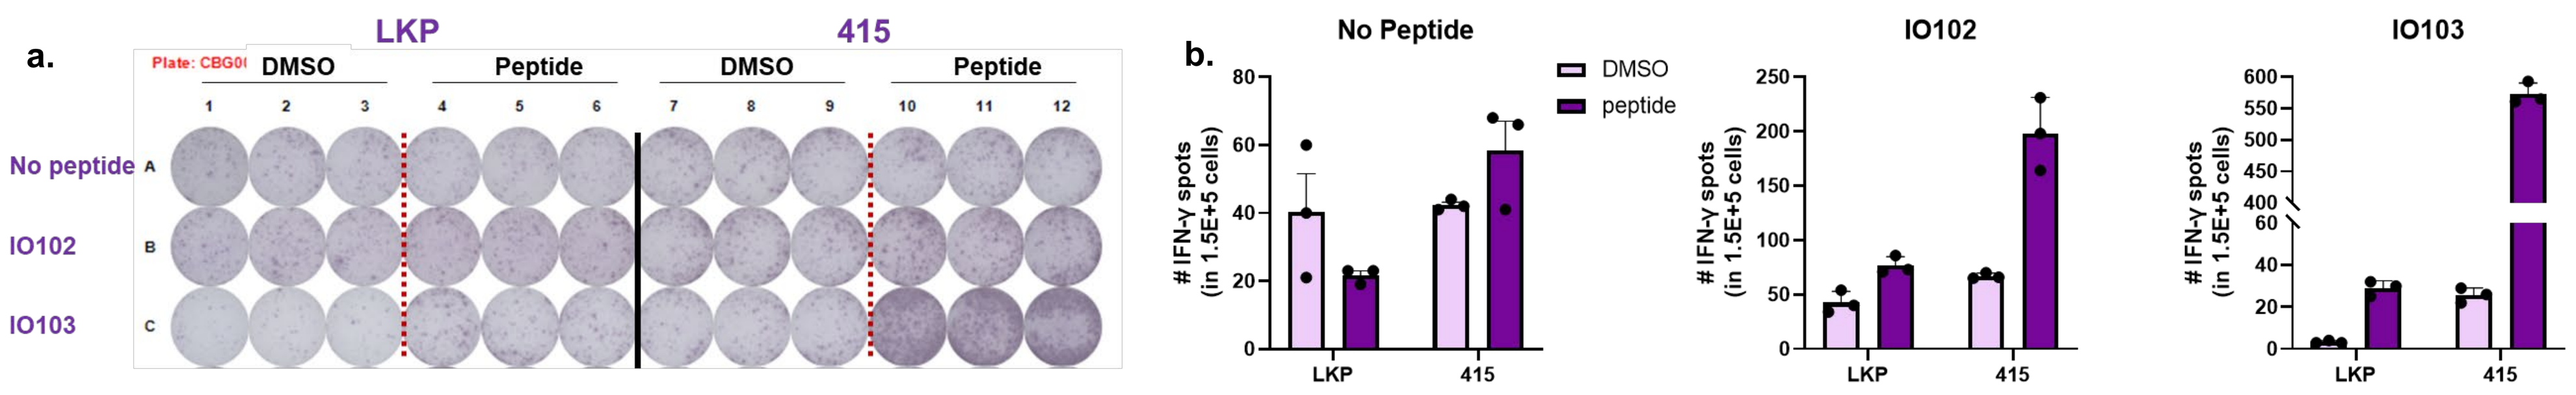
a.
LKP
415
DMSO
Peptide
DMSO
Peptide
No peptide
IO102
IO103
b.
No Peptide
IO102
IO103
DMSO
peptide
IFN-γ spots (in 1.5E+5 cells)
LKP
415

Stimulation and culture conditions along with scientific procedure for ELISPOT assay are detailed under "Methods." **a.** IFN-γ producing spots generated by PBMCs from 2 donors (LKP or 415) cultured with no peptide, IO102, or IO103 at day 8, 24 hours following restimulation **b.** Counts of IFN-γ producing spots corresponding to culture conditions, peptide, and donor

Baseline Expression of TRAV and TRBV Sequences Among Both Donors

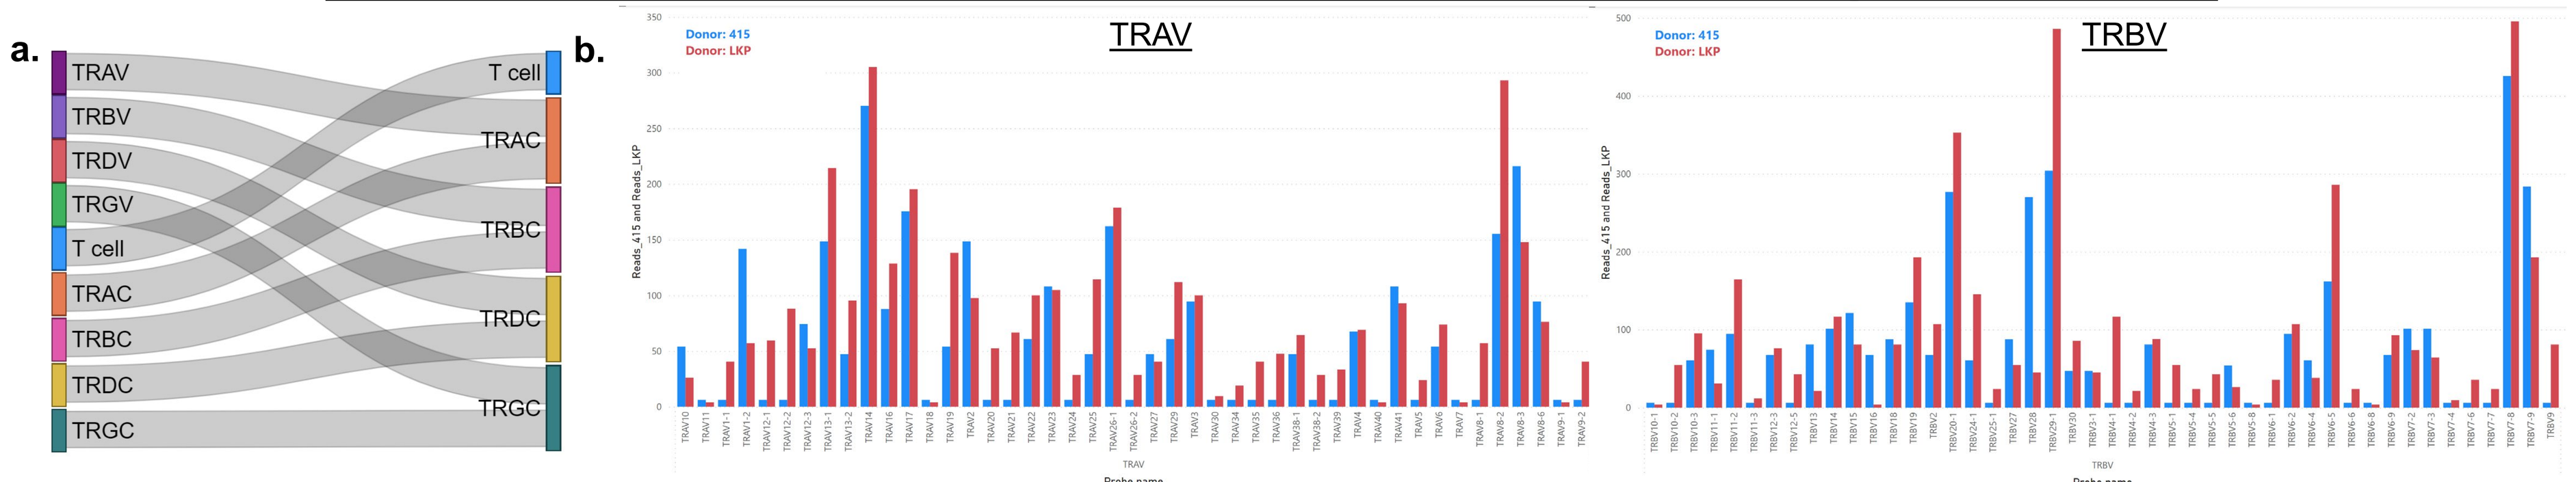
a.
TRAV
TRBV
TRDV
TRGV
T cell
TRAC
TRBC
TRDC
TRGC
b.
TRAV
TRBV
Donor: 415
Donor: LKP

a. Sankey plot demonstrating relationship of variable to constant regions using Nanostring TCRD probes. **b.** Baseline (day 0) expression of TRAV and TRBV usage for both donors (415 & LKP)

TCR Constant Region Usage, TCR Diversity, and TCR Variable Gene Expression Among LKP and 415 Stimulated with IO102 or IO103

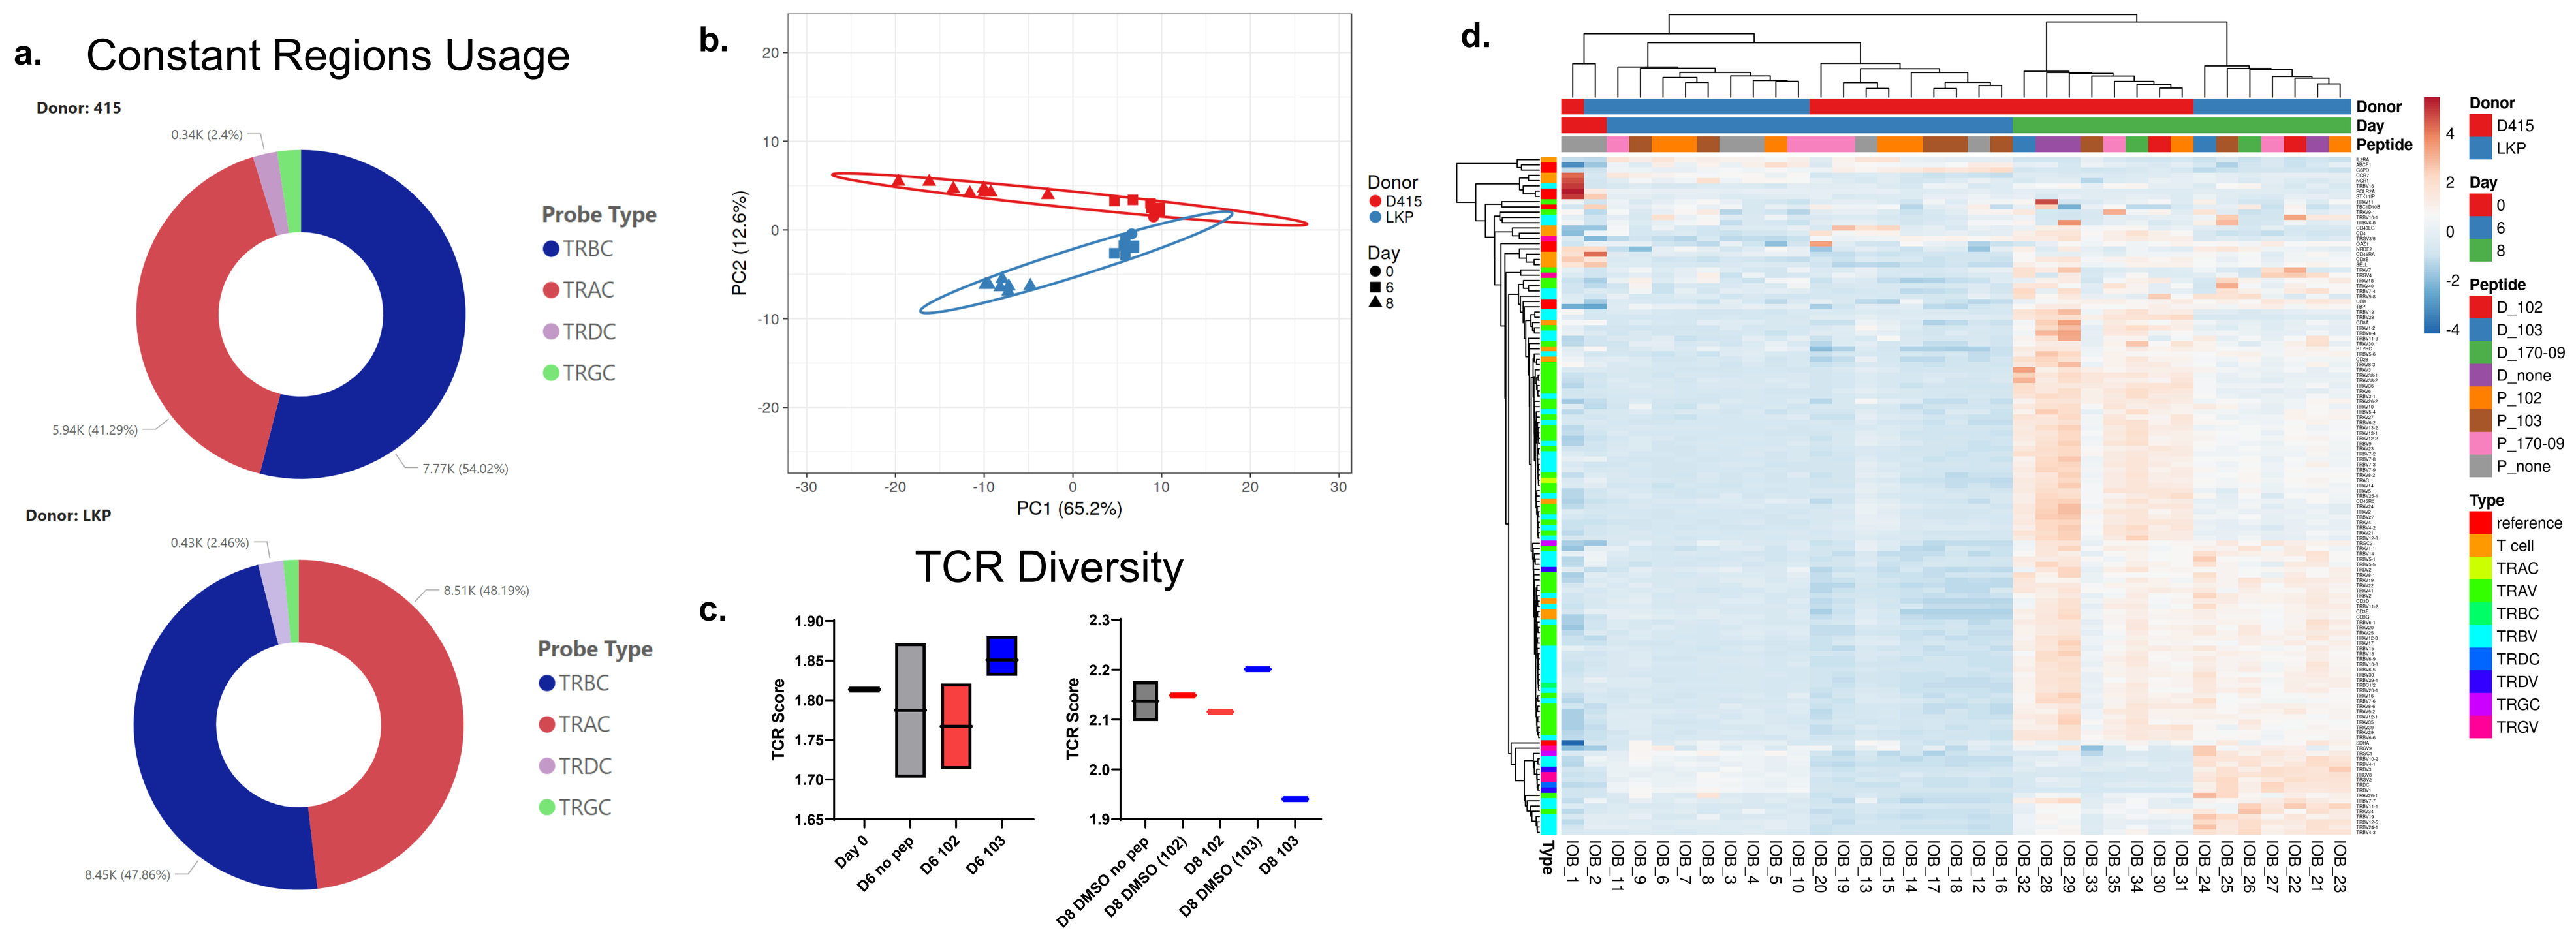
a. Constant Regions Usage
Donor: 415
Donor: LKP
Probe Type
TRBC
TRAC
TRDC
TRGC
b.
PC1 (65.2%)
PC2 (12.6%)
Donor
D415
LKP
Day
c.
TCR Diversity
TCR Score
Day 0
D6 no pep
D6 102
D6 103
D8 DMSO no pep
D8 DMSO (102)
D8 102
D8 DMSO (103)
D8 103
d.
Donor
Day
Peptide
Type

a. Baseline (day 0) constant region usage for both donors (415 and LKP) **b.** Timepoints for sample collection & PBMC culture conditions are detailed under "Methods." PCA plot of all samples for both donors **c.** TCR Score demonstrating TCR Diversity of TCRV usage at day 6 (left) and day 8 (right). **d.** Unsupervised heatmap of T cell phenotype, constant and variable gene usage across time

Case Study of Donor LKP + IO102 Peptide for TCRα and TCR-β Pairing

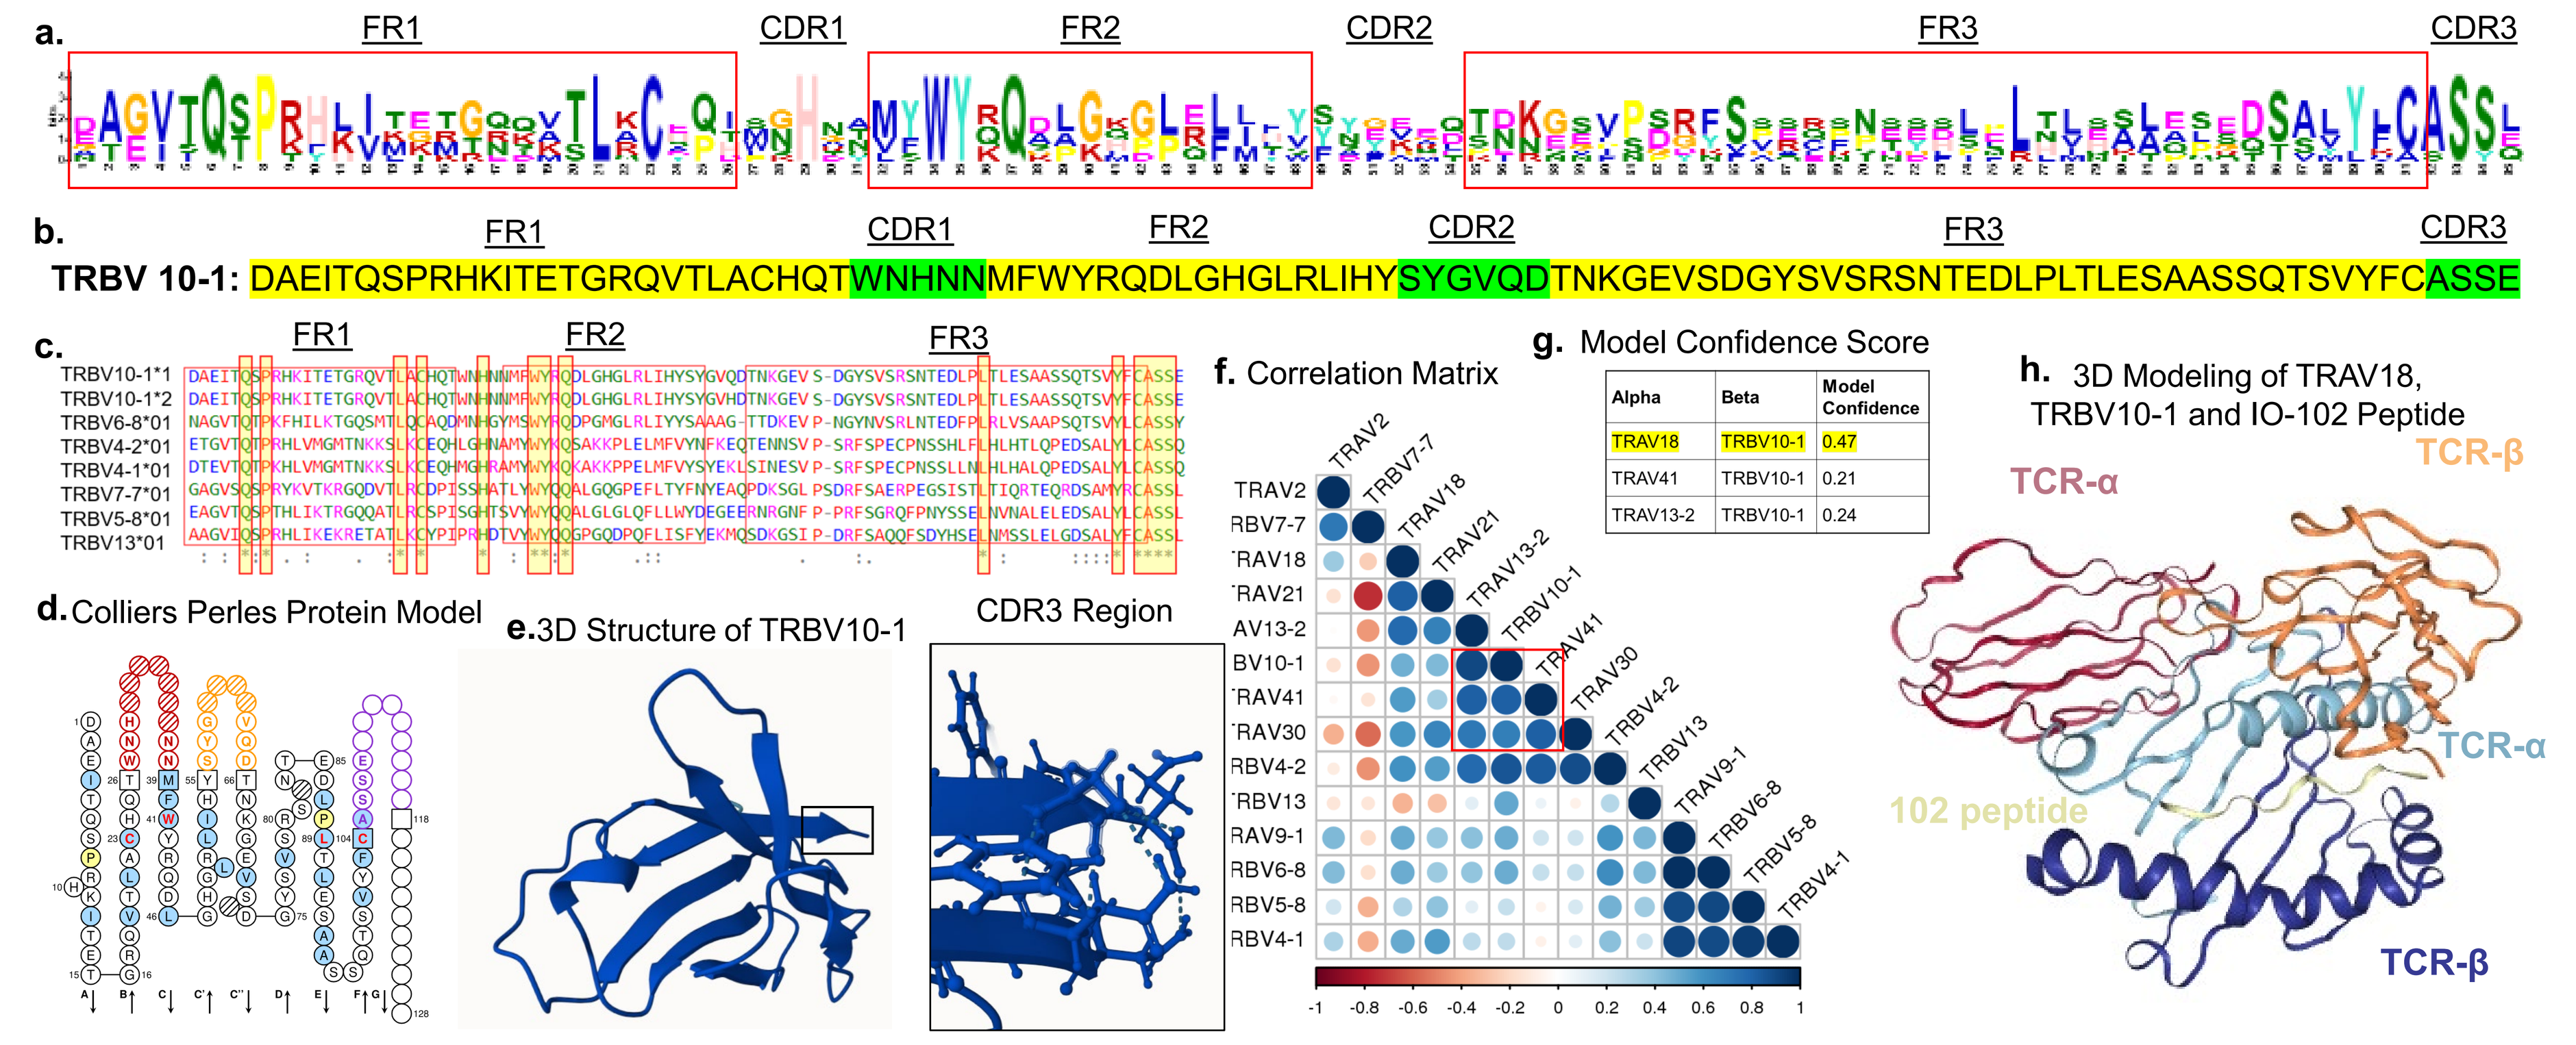
a.
FR1
CDR1
FR2
CDR2
FR3
CDR3
b.
TRBV 10-1: DAEITQSPRHKITETGRQVTLACHQTWNHNNMFWYRQDLGHGLRLIHYSYGVQDTNKGEVSDGYSVSRSNTEDLPLTLESAASSQTSVYFCASSE
c.
d. Colliers Perles Protein Model
e. 3D Structure of TRBV10-1
CDR3 Region
f. Correlation Matrix
g. Model Confidence Score
Alpha
Beta
Model Confidence
TRAV18
TRBV10-1
0.47
TRAV41
TRBV10-1
0.21
TRAV13-2
TRBV10-1
0.24
h. 3D Modeling of TRAV18, TRBV10-1 and IO-102 Peptide
TCR-α
TCR-β
102 peptide

a. MAST (Motif Alignment and Search Tool) and **c.** Multiple sequence alignment (MSA) for top 7 TCRBV regions enriched among LKP donor stimulated with IO102. Framework region (FR) and complementarity determining region (CDR) are annotated and conserved regions are highlighted **b.** Variable region sequence and annotation for TRBV 10-1 **d.** Colliers Perles Protein Model of TRBV 10-1 **e.** 3D structure of TRBV10-1 and detail of the CDR3 region **f.** Correlation matrix of the top 6 TRAV and top 6 TRBV regions enriched among LKP donor stimulated with IO102. Highlight (red box) of predictive pairing of TRBV 10-1 with correlated TRAV regions **g.** Model Confidence Score of top TRAV binding to TRBV 10-1. **h.** 3D structure of TRAV18, TRBV10-1 and IO-102 peptide accounting for donor HLA

Differential Expression of TCRV Regions 6 Days Following IO102 or IO103 Stimulation Among 2 Donors

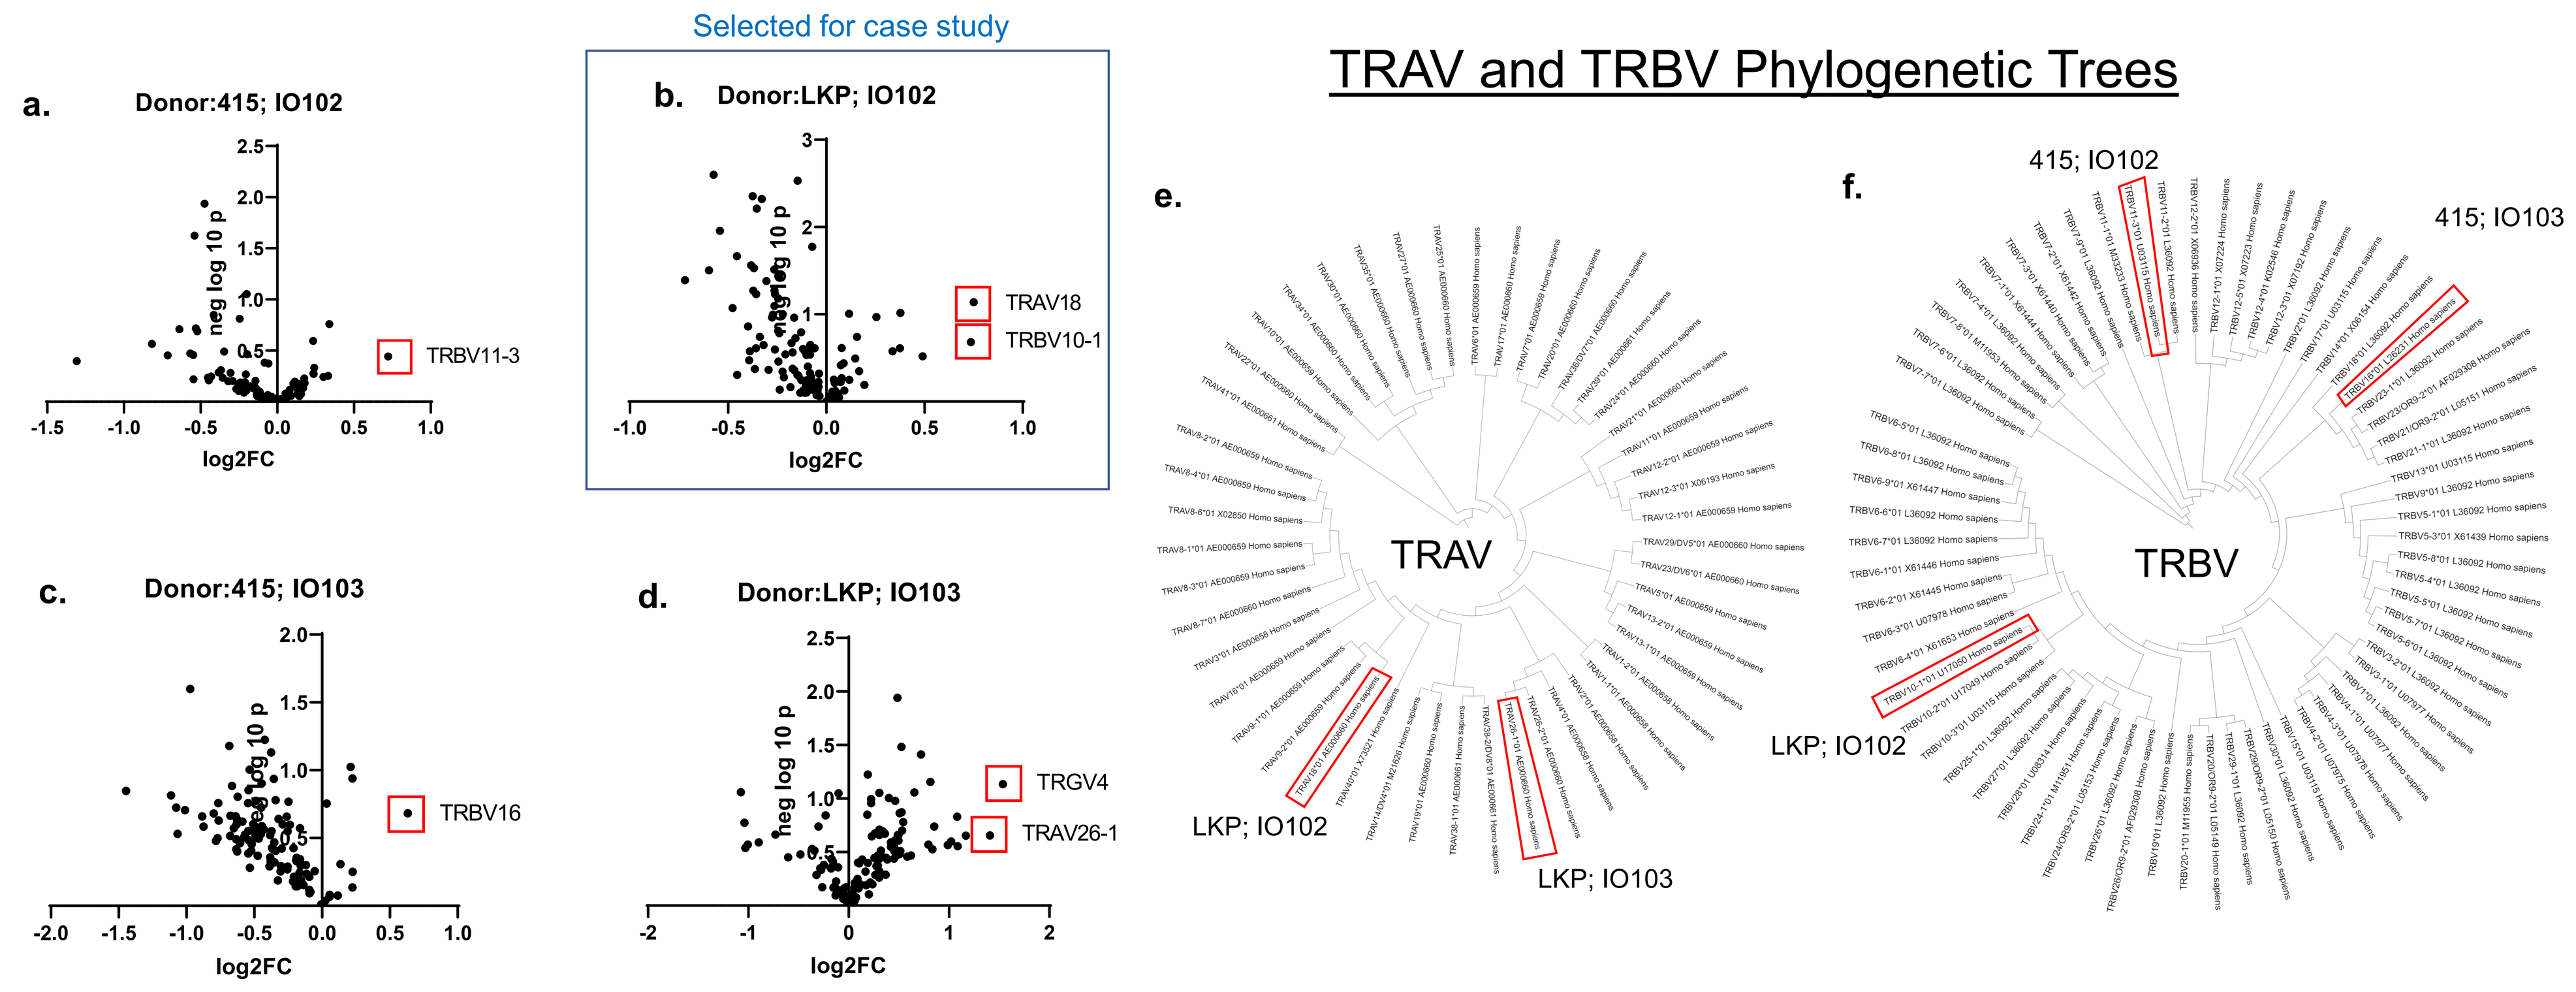
Selected for case study
TRAV and TRBV Phylogenetic Trees
a. Donor:415; IO102
TRBV11-3
b. Donor:LKP; IO102
TRAV18
TRBV10-1
c. Donor:415; IO103
TRBV16
d. Donor:LKP; IO103
TRGV4
TRAV26-1
neg log 10 p
log2FC
e.
TRAV
LKP; IO102
LKP; IO103
f.
TRBV
415; IO102
415; IO103
LKP; IO102

a-d. Enrichment of TCRV gene usage among donors 415 or LKP stimulated with IO102 or IO103 for 6 days. Peptide stimulation was compared to no-peptide control with same culture conditions and timing **b.** LKP donor PBMCs cultured with IO102 was selected for case study (blue box) for TCRα and TCRβ pairing and CDR3 analysis **e, f.** Phylogenetic trees of TRAV or TRBV regions with highlighted TRAV and TRBV regions enriched following either IO102 or IO103 peptide stimulation between donors for assessment of relatedness

Profile of Public Human TCR Clones Expressing TRBV10-1:

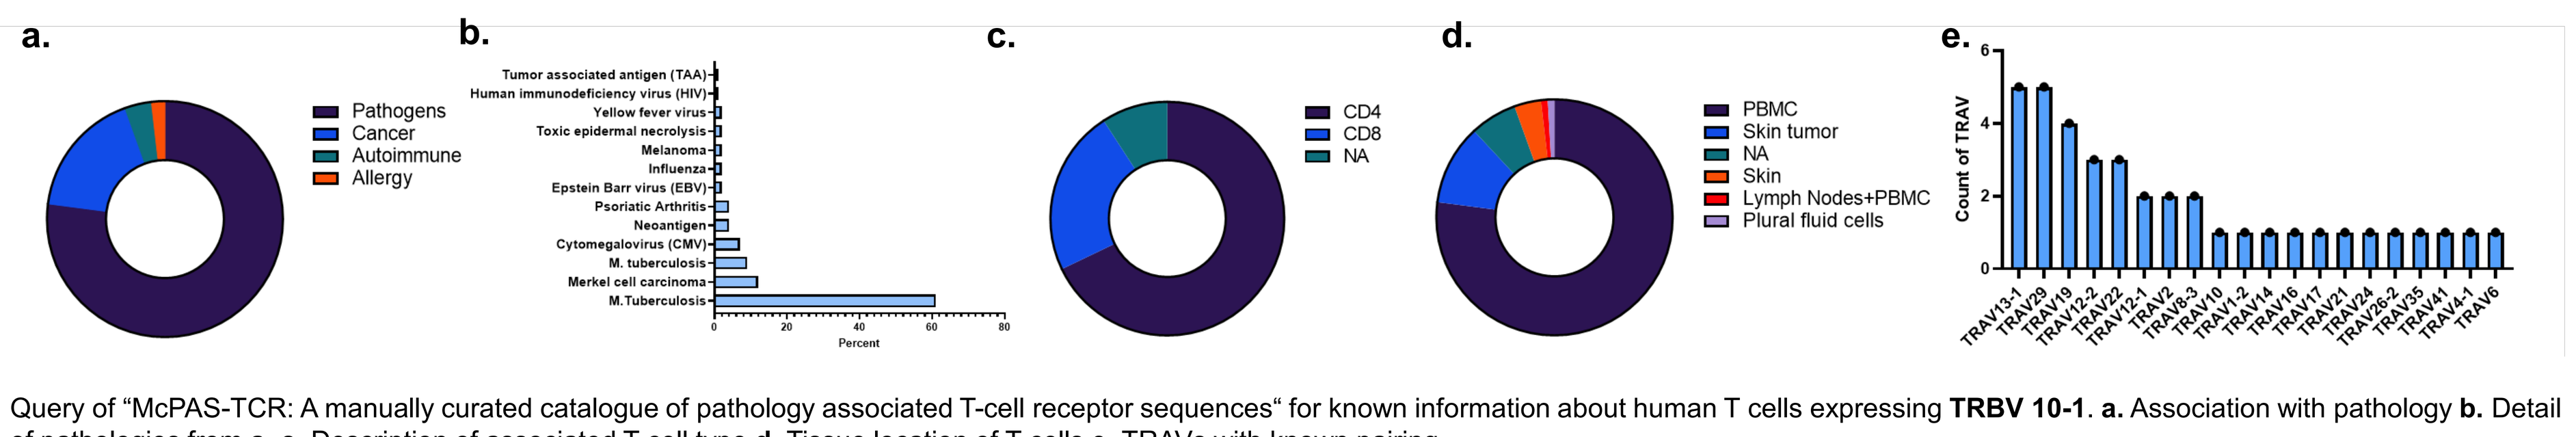
a.
Pathogens
Cancer
Autoimmune
Allergy
b.
Percent
c.
CD4
CD8
NA
d.
PBMC
Skin tumor
NA
Skin
Lymph Nodes+PBMC
Plural fluid cells
e.
Count of TRAV

Query of "McPAS-TCR: A manually curated catalogue of pathology associated T-cell receptor sequences" for known information about human T cells expressing **TRBV 10-1**. **a.** Association with pathology **b.** Detail of pathologies from a. **c.** Description of associated T cell type **d.** Tissue location of T cells **e.** TRAVs with known pairing

Assessment of T Cell Phenotype from Nanostring TCRD, including Validation of Memory Markers

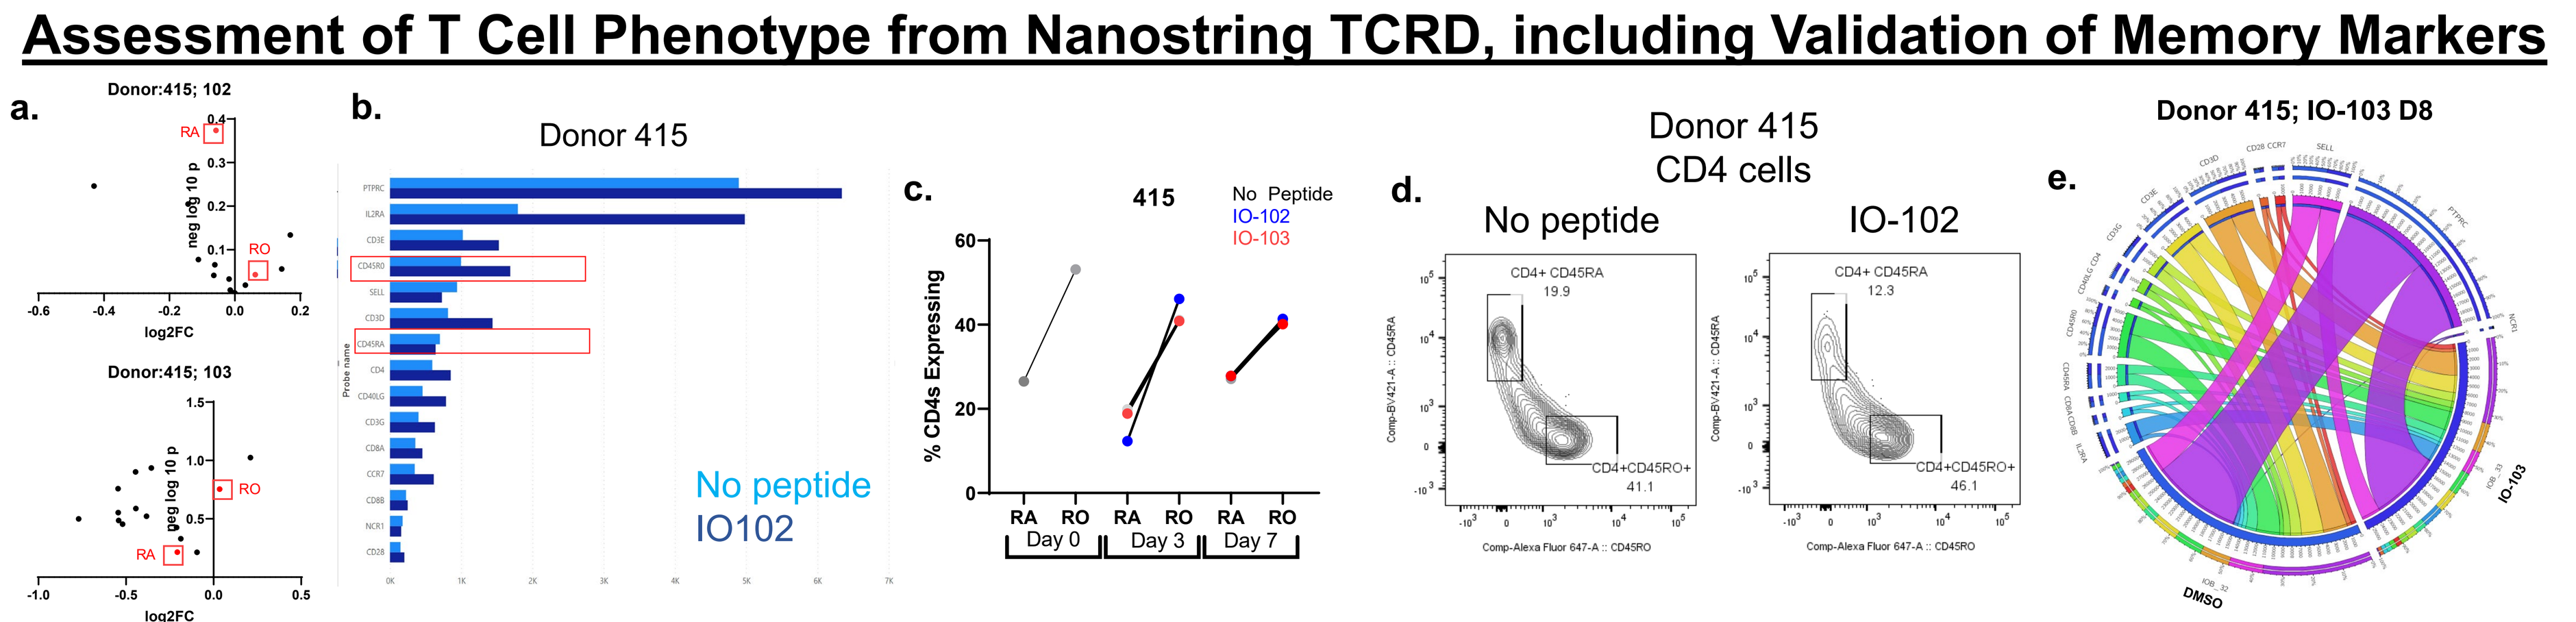
a.
Donor:415; 102
Donor:415; 103
RA
RO
b.
Donor 415
No peptide
IO102
c.
415
No Peptide
IO-102
IO-103
% CD4s Expressing
Day 0
Day 3
Day 7
d.
Donor 415
CD4 cells
No peptide
IO-102
e.
Donor 415; IO-103 D8

a. Volcano plot and **b.** Histogram of T cell phenotype probes from Nanostring TCRD panel. CD45RA (RA) and CD45RO (RO) highlighted in red. **c.** Expression of CD45RA and CD45RO on CD4 T cells following 3 or 7 day culture with no peptide, IO102, or IO103. **d.** Representative flow cytometry at day 3 from c. **e.** Circos plot of T cell phenotype probes at day 8, 24 hours following restimulation

Conclusions:

- The nCounter TCR Diversity panel from NanoString provides wide coverage of T cell variable gene usage in healthy human PBMCs
- Among the 2 donors analyzed, exposure to IO102 or IO103 in short-term *in vitro* cultures resulted in discernable changes to the TCR repertoire, as per assessment of variable gene region usage and TCR diversity
- Using control cultures, we are able to identify TCRV segments that are responsive to either IO102 or IO103 peptides in a donor-specific manner
- Relatedness of TRAV and TRBV regions between peptides and donors can be assessed at a phylogenetic level
- Top TRBVs, including TRBV10-1, responding to IO102 peptide have regions of epitopes within their protein sequence that are conserved
- Correlation matrixes of top TRAV and TRBV usage along with enrichment in TCRV gene usage provides scenarios for potential TCRα and TCRβ pairing
- Using 3D modeling, we can predict which TCRα chain potentially pairs with TRBV10-1 given IO102 peptide sequence and donor HLA
- TRBV10-1 appears in public profiles of T cells and its association with pathogenicity, T cell phenotype, tissue, and potential TCRα pairing is noted
- Exposure to antigen shifts T cell phenotype slightly towards a memory phenotype
- Future studies: We are further working on validation of TCRV gene usage among IO102 or IO103 responsive T cells using enriched cultures, qPCR primer probes specific for variable regions, and antibodies targeting specific TRAV and TRBV regions

References:

- Bailey, T. L., and M. Gribskov. "Methods and Statistics for Combining Motif Match Scores." *Journal of Computational Biology: A Journal of Computational Molecular Cell Biology* 5, no. 2 (1998): 211–21. https://doi.org/10.1089/cmb.1998.5.211. "Score Distributions for Simultaneous Matching to Multiple Motifs." *Journal of Computational Biology: A Journal of Computational Molecular Cell Biology* 4, no. 1 (1997): 45–59. https://doi.org/10.1089/cmb.1997.4.45.
- BAILEY, TIMOTHY L., and MICHAEL GRIBSKOV. "Methods and Statistics for Combining Motif Match Scores." *Journal of Computational Biology* 5, no. 2 (January 1, 1998): 211–21. https://doi.org/10.1089/cmb.1998.5.211.
- Callaway, Ewen. "What's next for AlphaFold and the AI Protein-Folding Revolution." *Nature* 604, no. 7905 (April 2022): 234–38. https://doi.org/10.1038/d41586-022-00997-5.
- Davis, M. M., and P. J. Bjorkman. "T-Cell Antigen Receptor Genes and T-Cell Recognition." *Nature* 334, no. 6181 (August 4, 1988): 395–402. https://doi.org/10.1038/334395a0.
- Kjeldsen, Julie Westerlin, Cathrine Lund Lorentzen, Evelina Martinenaite, Eva Ellebaek, Marco Donia, Rikke Boedker Holmstroem, Tobias Wirenfeldt Klausen, et al. "A Phase 1/2 Trial of an Immune-Modulatory Vaccine against IDO/PD-L1 in Combination with Nivolumab in Metastatic Melanoma." *Nature Medicine* 27, no. 12 (December 1, 2021): 2212–23. https://doi.org/10.1038/s41591-021-01544-x.
- Krzywinski, Martin, Jacqueline Schein, Inanç Birol, Joseph Connors, Randy Gascoyne, Doug Horsman, Steven J. Jones, and Marco A. Marra. "Circos: An Information Aesthetic for Comparative Genomics." *Genome Research* 19, no. 9 (September 2009): 1639–45. https://doi.org/10.1101/gr.092759.109.
- Lythe, Grant, Robin E. Callard, Rollo L. Hoare, and Carmen Molina-París. "How Many TCR Clonotypes Does a Body Maintain?" *Journal of Theoretical Biology* 389 (January 21, 2016): 214–24. https://doi.org/10.1016/j.jtbi.2015.10.016.
- Madeira, Fábio, Matt Pearce, Adrian R. N. Tivey, Prasad Basutkar, Joon Lee, Ossama Edbali, Nandana Madhusoodanan, Anton Kolesnikov, and Rodrigo Lopez. "Search and Sequence Analysis Tools Services from EMBL-EBI in 2022." *Nucleic Acids Research* 50, no. W1 (July 5, 2022): W276–79. https://doi.org/10.1093/nar/gkac240.
- Manso, Taciana, Géraldine Folch, Véronique Giudicelli, Joumana Jabado-Michaloud, Anjana Kushwaha, Viviane Nguefack Ngoune, Maria Georga, et al. "IMGT® Databases, Related Tools and Web Resources through Three Main Axes of Research and Development." *Nucleic Acids Research* 50, no. D1 (January 7, 2022): D1262–72. https://doi.org/10.1093/nar/gkab1136.
- Ruiz, Manuel, and Marie-Paule Lefranc. "IMGT Gene Identification and Colliers de Perles of Human Immunoglobulins with Known 3D Structures." *Immunogenetics* 53, no. 10–11 (February 2002): 857–83. https://doi.org/10.1007/s00251-001-0408-6.
- Tickotsky, Nili, Tal Sagiv, Jaime Prilusky, Eric Shifrut, and Nir Friedman. "McPAS-TCR: A Manually Curated Catalogue of Pathology-Associated T Cell Receptor Sequences." *Bioinformatics (Oxford, England)* 33, no. 18 (September 15, 2017): 2924–29. https://doi.org/10.1093/bioinformatics/btx286.
- Yin, Rui, Helder V. Ribeiro-Filho, Valerie Lin, Ragul Gowthaman, Melyssa Cheung, and Brian G. Pierce. "TCRmodel2: High-Resolution Modeling of T Cell Receptor Recognition Using Deep Learning." *Nucleic Acids Research* 51, no. W1 (July 5, 2023): W569–76. https://doi.org/10.1093/nar/gkad356.