Modulating the tumor microenvironment by targeting TGFB1 with vaccine-induced immune responses

Contact information
Brian Weinert
Principal Scientist – IO Biotech
btw@iobiotech.com

Brian Weinert[1], Tine Hannibal[1], Preeyam Patel[2], Evelina Martinenaite[1], Marion Chapellier[1], Marco Carretta[1], Alireza Alavi[2], Ayako Wakatsuki Pedersen[1], Muhammad Al-Hajj[2]

[1] IO Biotech, Ole Maaloes Vej 3, DK - 2200 Copenhagen N, Denmark [2] IO Biotech, 5640 Fishers Lane, Suite C, Rockville, MD 20852, USA

Background and Aims

A key challenge of the immunotherapy space of solid tumors is the suppressive tumor microenvironment. Immunotherapy can drive highly durable responses; however, this is the case for a minority of patients as the majority do not respond to these therapies. Recent clinical trial results [1] brought an exciting novel way to tackle the immune suppressive TME by vaccinating against immunosuppressive antigens [2]. Transforming growth factor beta 1 (TGFB1) is a key element of the TGFB pathway that plays a major role in immune suppression in various cancer indications. We hypothesize that activating T cells against TGFB1 presenting cells may allow targeting of cytotoxic and pro-inflammatory immune responses to TGFB1-expressing tumors. TGFB1-reactive T cells are frequently detected in humans [3,4]. Vaccination with a TGFB1 peptide ameliorates fibrosis in a model of chronic colitis [5] and enhances the anti-tumor activity of an HPV16 E7-specific vaccine [6], providing evidence for the therapeutic potential of a TGFB1 vaccine.

Here we set out to explore the feasibility and biological rationale to develop a TGFB1 directed vaccine for cancer therapy applications. We assessed TGFB1 expression and the identities of the cells expressing it in the TME of various solid tumor indications; in addition, we determined the amount of overlap with other immune suppressive antigens (e.g. IDO & PDL1). Furthermore, we identified and characterized TGFB1 peptide antigens that elicit strong and frequent immune responses with no cross-reactivity to TGFB2 and TGFB3. We then developed an optimized murine TGFB1 peptide vaccine and assessed its functionality through analysis of its impact on tumor growth and the modulation of the tumor microenvironment in the MC38 colon cancer mouse model.

TGFB1 characterization in the tumor microenvironment of multiple solid cancer indications

BACKGROUND Neogenomics MultiOmyx™ technology was used to evaluate the expression of a panel of 18 phenotypic markers and immunosuppressive antigens: ARG1, CD3, CD4, CD8, CD11b, CD68, CD163, FAP, FoxP3, HLADR, IDO1, LAG-3, PanCK, PD-1, PD-L1, TGFB1, and TIGIT. Proprietary deep learning algorithms were used to classify positive cells for each marker. The data presented here focus on the frequency of positive classified cells and their overlap between various markers. More than 30 regions of interest (ROI) were analyzed for each cancer type.
RESULTS TGFB1 was found to be highly expressed on tumor cells in Esophageal and Urothelial cancers, while TGFB1 was expressed in the TME of most cancer types. TGFB1 expressing cells constitute a comparable fraction of tumors as IDO1 and PD-L1, the antigens targeted by IO Biotech's lead therapeutics IO102 and IO103.

Cancer types analyzed

CRC	Colorectal
ESOPHAGEAL	Esophageal squamous cell carcinoma
GASTRIC	Gastric
HNN	Head and Neck Cancer
NSCLC	Non-small cell lung cancer (Adenocarcinoma)
OVARIAN	Ovarian Cancer (Serous Papillary Adenocarcinoma)
UROTHELIAL	Urothelial Carcinoma (Bladder Cancer)

Cell types and markers used to define them

Label	Description	Markers
Tumor_cell	Tumor cell	PanCK
CAF	Cancer Associated Fibroblast	FAP
CD8	CD8+ T cell	CD3+,CD8+
CD4	CD4+ T cell	CD3+,CD4+
Treg	Regulatory CD4+ T cell	CD4+,FoxP3+
exCD8	Exhausted CD8+ T cell	CD3+, CD8+, PD-1/TIGIT/LAG-3+
M1_TAM	M1 tumor associated macrophage	CD68+HLADR+CD163-
M2_TAM	M2 tumor associated macrophage	CD68+HLADR-CD163+
APCmye	Myeloid Antigen Presenting Cell	CD11b+HLADR+
Other	No marker expression	None

Representative images showing TGFB1 staining

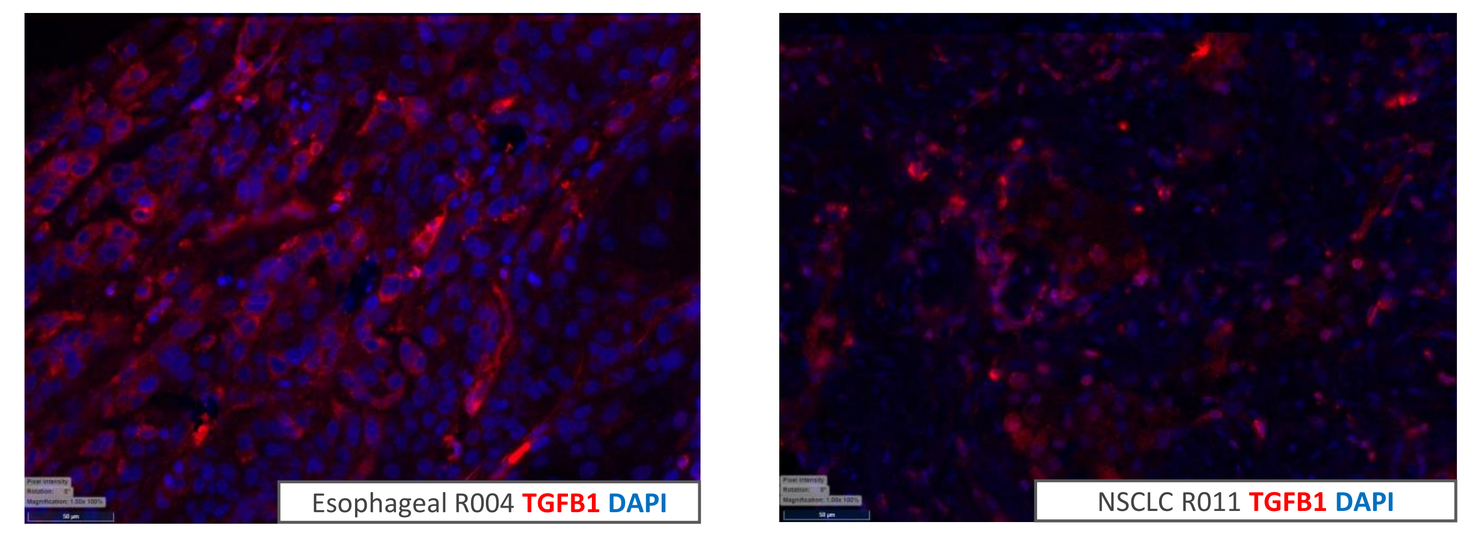

Esophageal R004 TGFB1 DAPI
NSCLC R011 TGFB1 DAPI

TGFB1 is expressed by a substantial fraction of cells in tumors and the tumor microenvironment (TME)

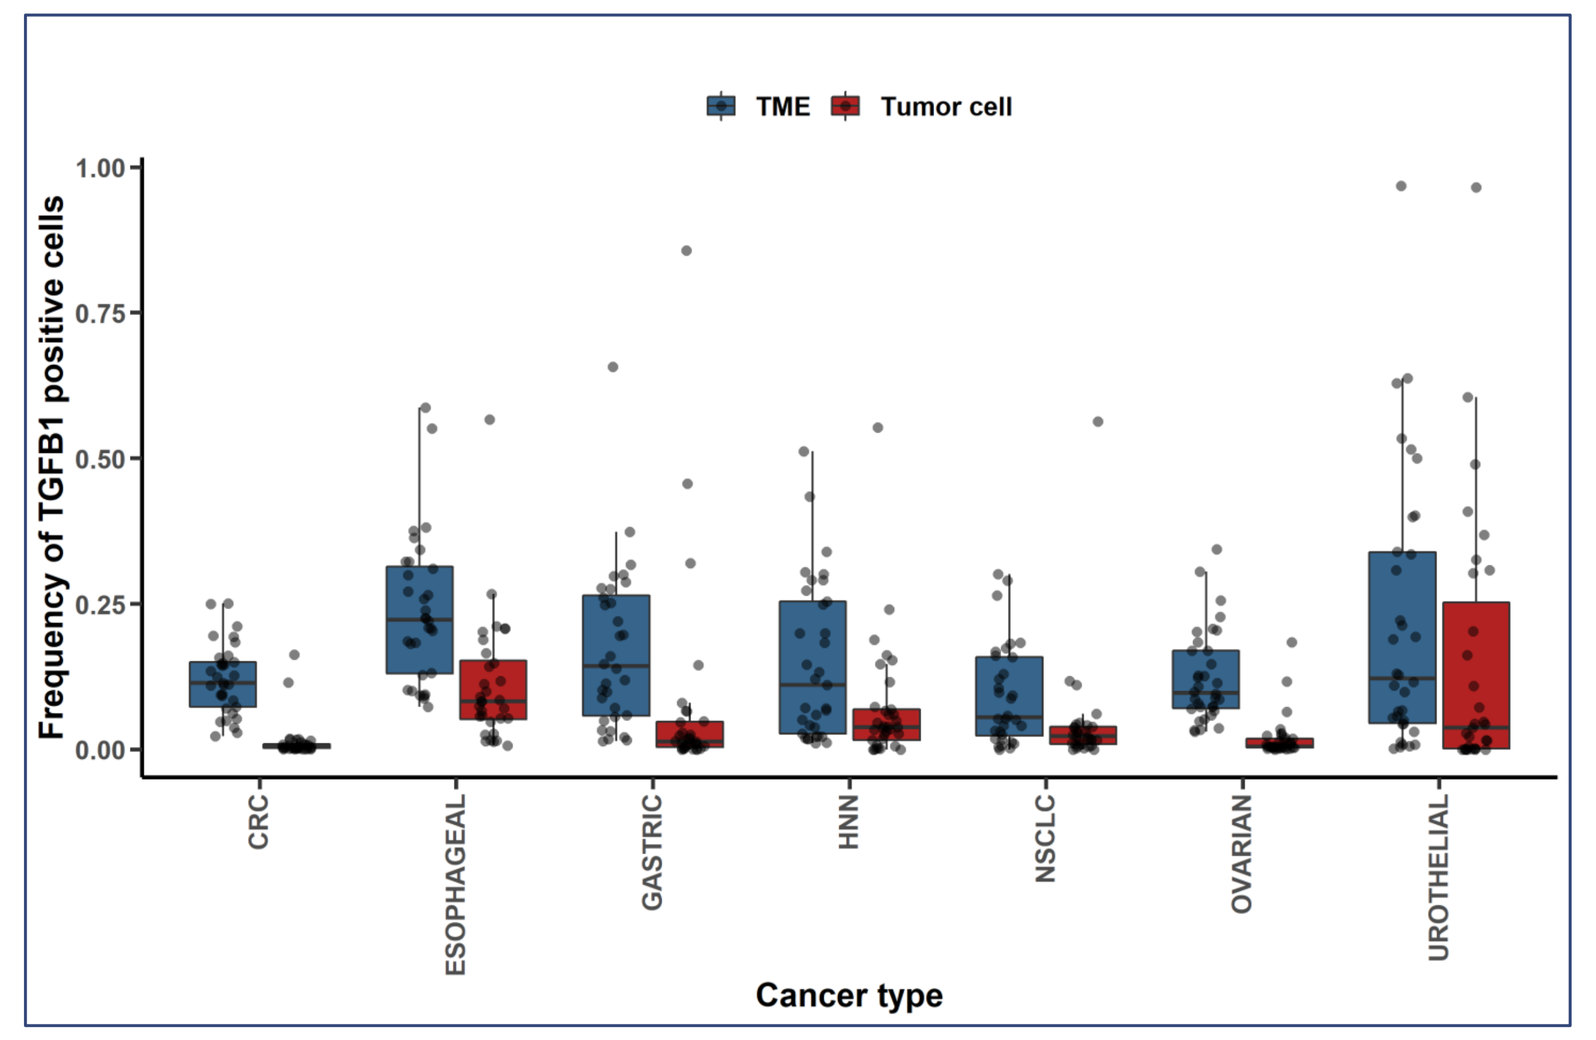

TME
Tumor cell
Frequency of TGFB1 positive cells
CRC
ESOPHAGEAL
GASTRIC
HNN
NSCLC
OVARIAN
UROTHELIAL
Cancer type

Boxplots and datapoints show the frequency of TGFB1 expression in tumor cells and the tumor microenvironment (TME, not tumor cells). The fraction of TGFB1 positive cells in each individual region of interest (ROI) is shown as a single datapoint.

TGFB1 is expressed by various cell types in the TME

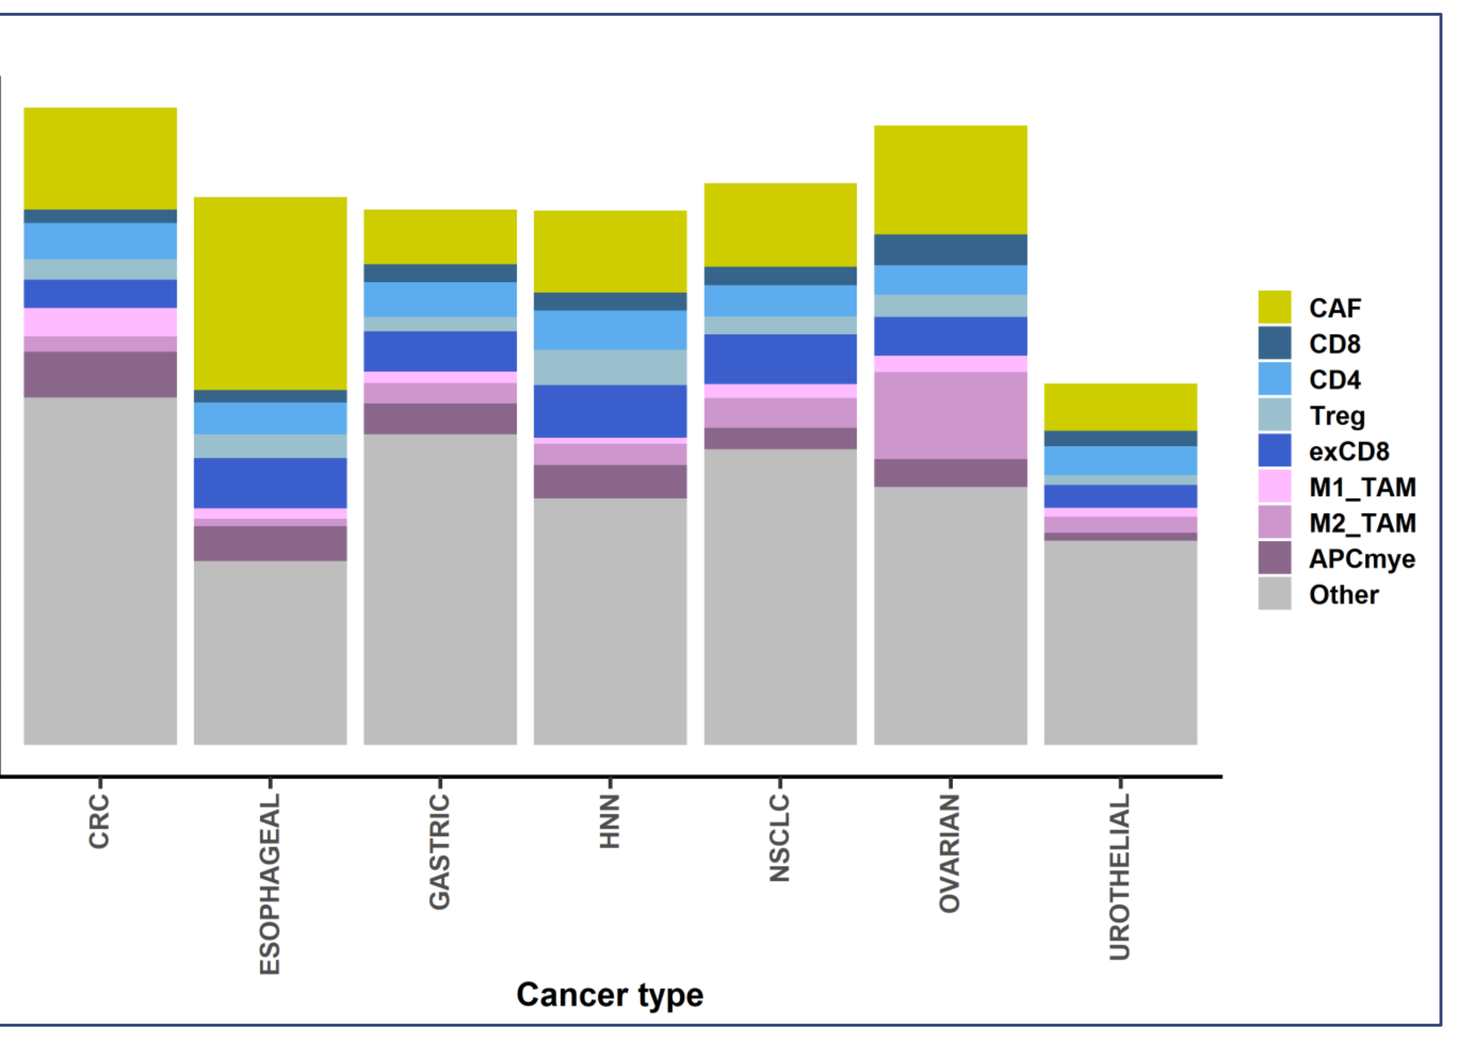

Fraction cells
CAF
CD8
CD4
Treg
exCD8
M1_TAM
M2_TAM
APCmye
Other
Cancer type

The column charts show the fraction of TGFB1 positive cells of each cell type in the TME (tumor cells excluded) of the indicated cancer types. Note that some categories are non-exclusive, so the total can exceed 1.0.

TGFB1 expressing cells are not confined to the TME population expressing other immune suppression markers

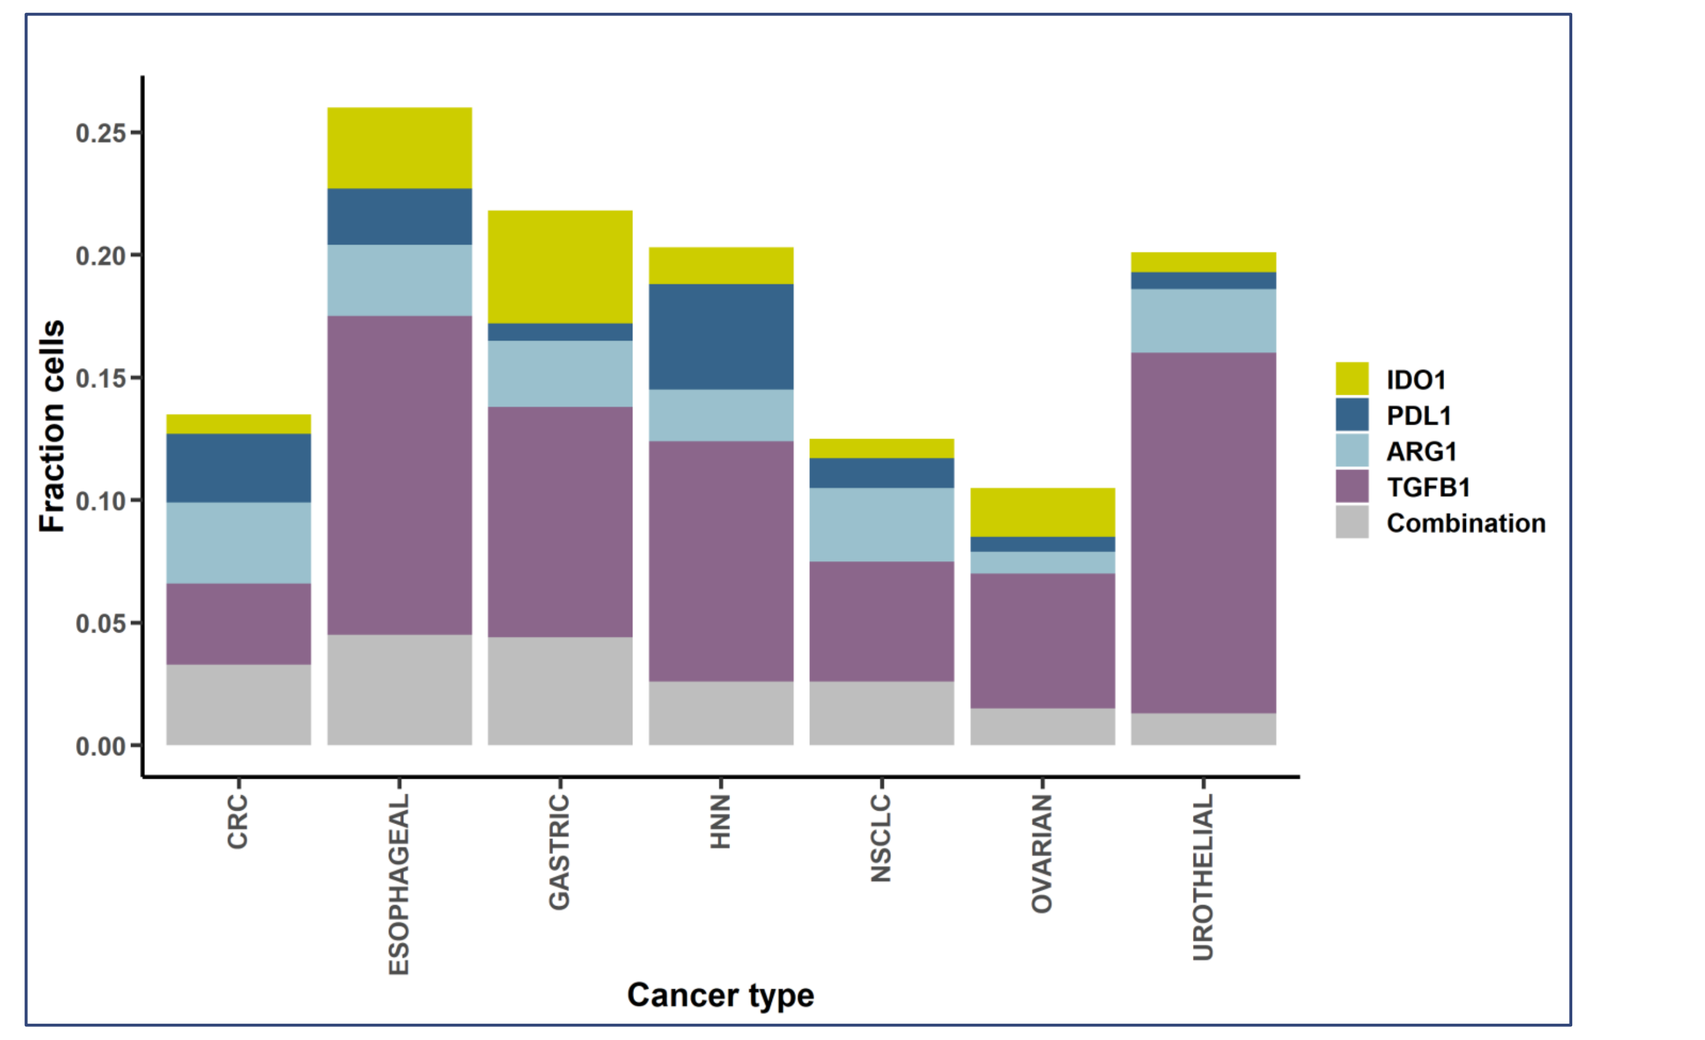

Fraction cells
IDO1
PDL1
ARG1
TGFB1
Combination
Cancer type

The column chart shows the fraction of cells expressing the indicated immunosuppressive antigens. Cells expressing any combination of two or more antigens are shown in grey.

Identification and characterization of TGFB1-specific peptide antigens

BACKGROUND Therapeutics based on small molecule inhibitors targeting TGFB receptors, and trap ligands based on soluble TGFB receptors, target all three isoforms of TGFB (TGFB1, TGFB2, and TGFB3). Targeting of additional TGFB isoforms (TGFB2, TGFB3) has been suggested to contribute to adverse clinical effects. Activating T cells against TGFB1 specifically (no cross-reactivity to TGFB2 and TGFB3) may allow targeting of pro-inflammatory immune response to TGFB1-expressing tumors while avoiding the toxicity associated with pan-TGFB inhibition. Here we sought to enhance the specificity anti-TGFB1 immune responses by identifying peptides with high TGFB1 selectivity (versus TGFB2 and TGFB3).
RESULTS We identified low homology TGFB1 peptides that elicit strong (number of IFNg ELISPOT spots) and frequent (number of donors) immune responses. Five peptides with the strongest and most frequent immune responses were tested for TGFB1-selectivity. IFNg ELISPOT immune responses to TGFB1 peptides selected for low homology failed to cross-react with the homologous TGFB2 and TGFB3 peptides. In contrast, a peptide selected for high homology (Pep12), induced immune responses that cross-reacted with TGFB2 and TGFB3 in a manner that mirrored the magnitude of the TGFB1-specific response.

Sequence alignment of TGFB1, TGFB2, and TGFB3 homologs and the peptides used to assay TGFB1-specific immune responses

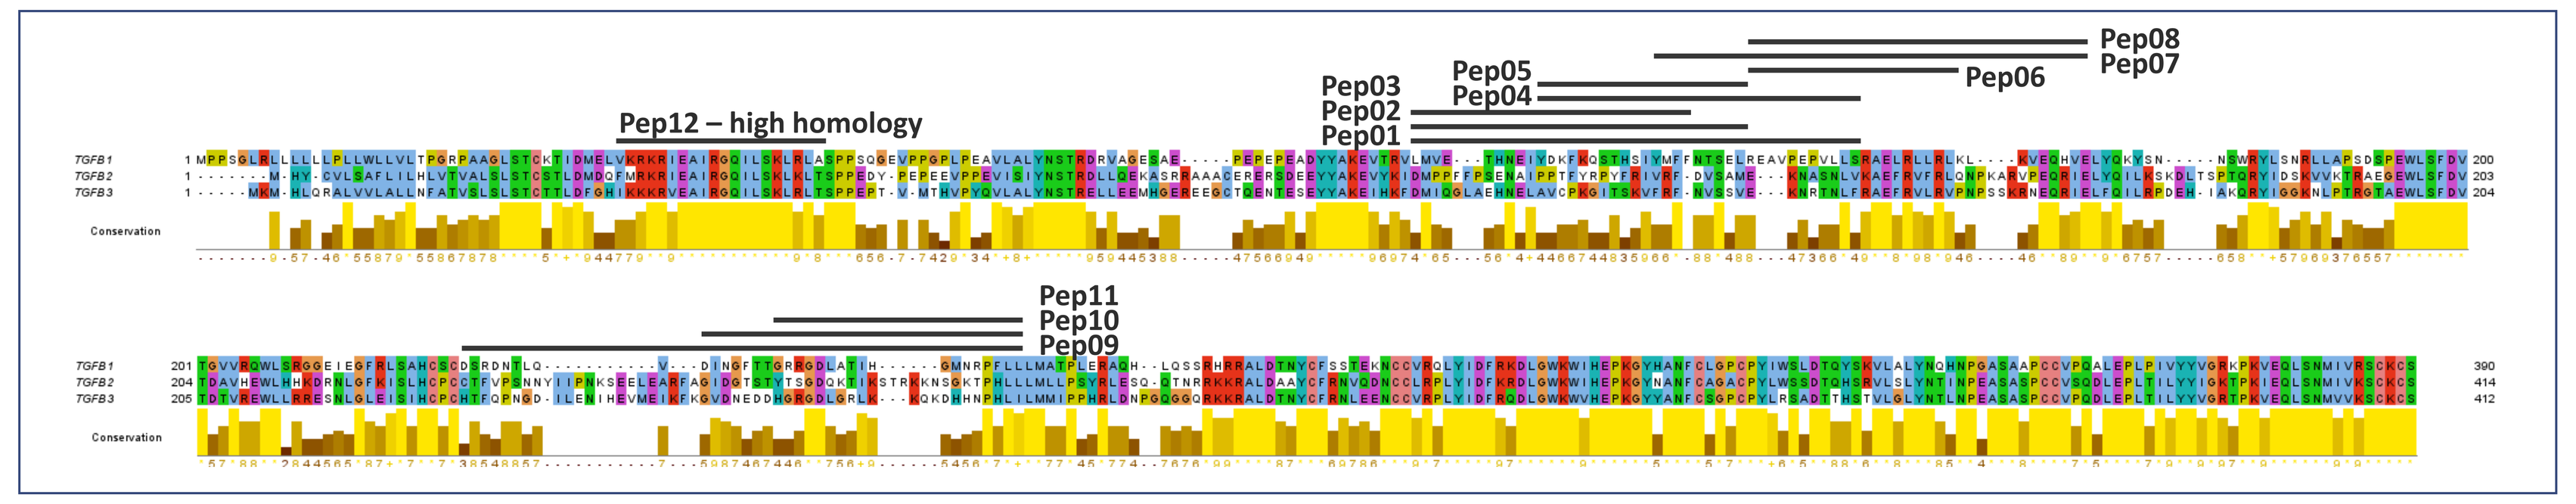

Pep12 – high homology
Pep01
Pep02
Pep03
Pep04
Pep05
Pep06
Pep07
Pep08
Pep09
Pep10
Pep11
TGFB1
TGFB2
TGFB3
Conservation

Clustal omega sequence alignment of the human TGFB1, TGFB2, and TGFB3 proteins. TGFB1 peptides assayed in this study are indicated.

Interferon gamma (IFNg) ELISPOT detects immune responses against TGFB1 peptides

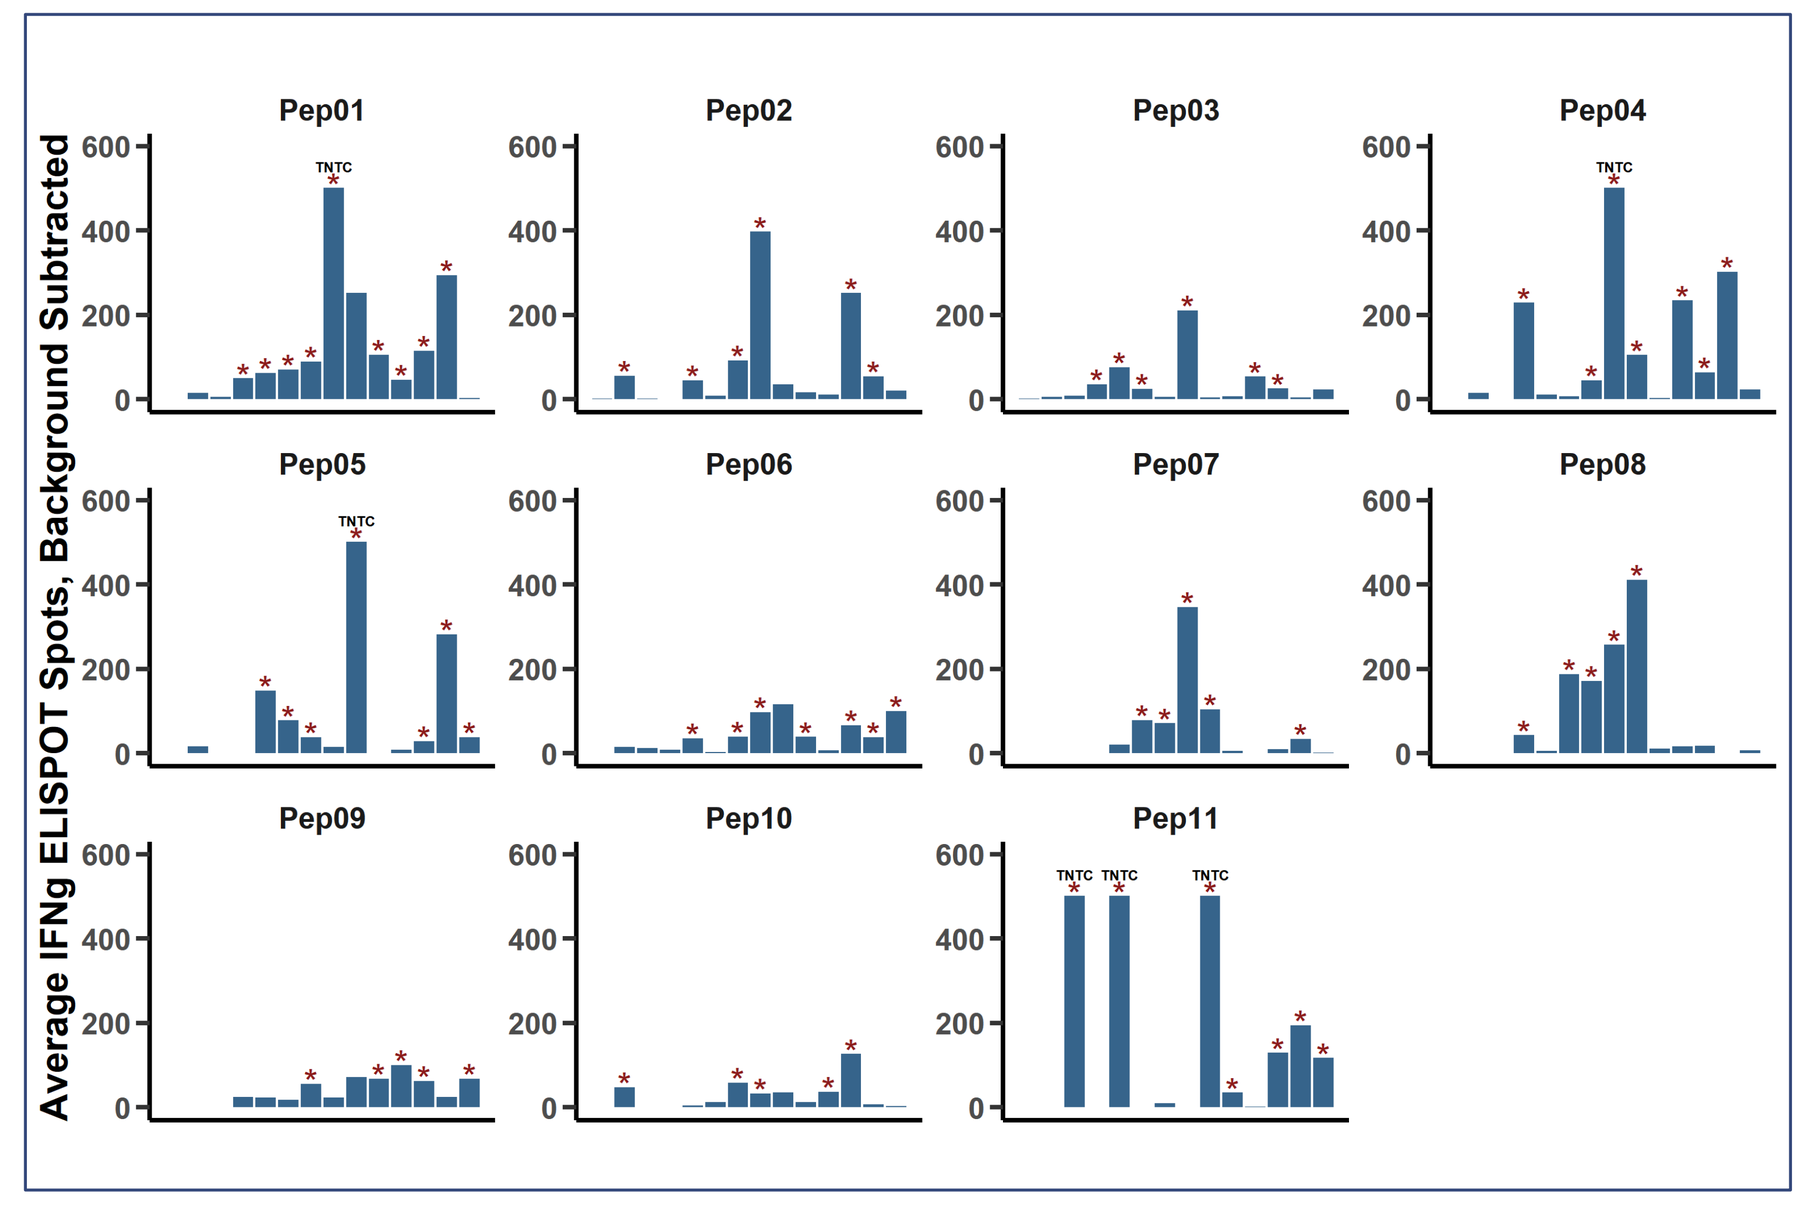

Average IFNg ELISPOT Spots, Background Subtracted
Pep01
Pep02
Pep03
Pep04
Pep05
Pep06
Pep07
Pep08
Pep09
Pep10
Pep11
TNTC

Peptide ID	Median Spots	Positive Donors
Pep01	67	9
Pep02	28	6
Pep03	16	6
Pep04	34	7
Pep05	23	7
Pep06	37	7
Pep07	8	5
Pep08	14	5
Pep09	25	5
Pep10	12	5
Pep11	22	7

Immune responses to TGFB1 peptides were identified in PBMCs from 14 healthy donors. PBMCs were stimulated with the indicated TGFB1 peptides and responses were analyzed after seven days by IFNg ELISPOT. Significant responses () were defined by a Fisher exact P value < 0.01, a ratio of peptide to control spots > 2, and background subtracted spots > 25. Peptides that elicited responses in the greatest number of donors are highlighted and were subsequently tested for the specificity of the response to TGFB1.*

Immune responses against low homology TGFB1 peptides do not cross react with TGFB2 and TGFB3 peptides

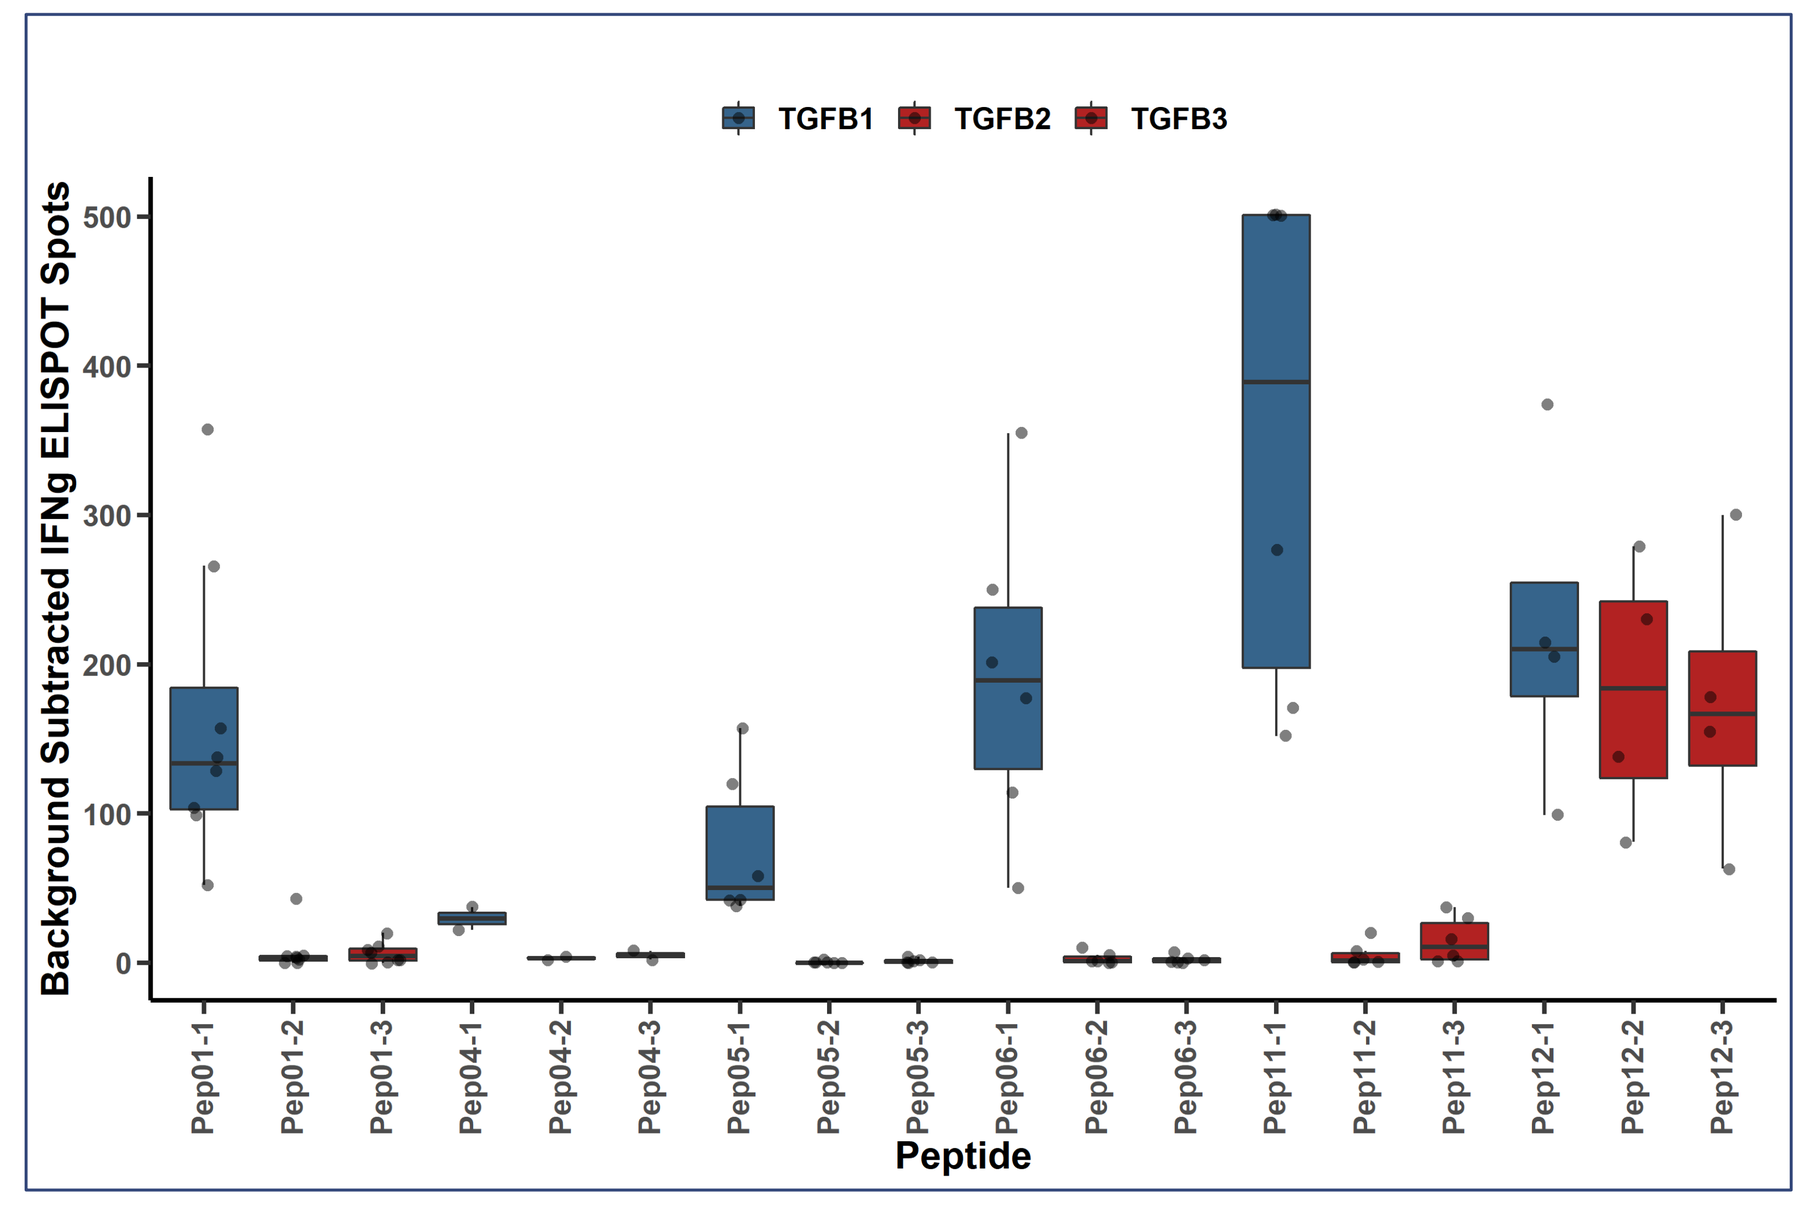

TGFB1
TGFB2
TGFB3
Background Subtracted IFNg ELISPOT Spots
Peptide

Representative ELISPOT assays (triplicate)

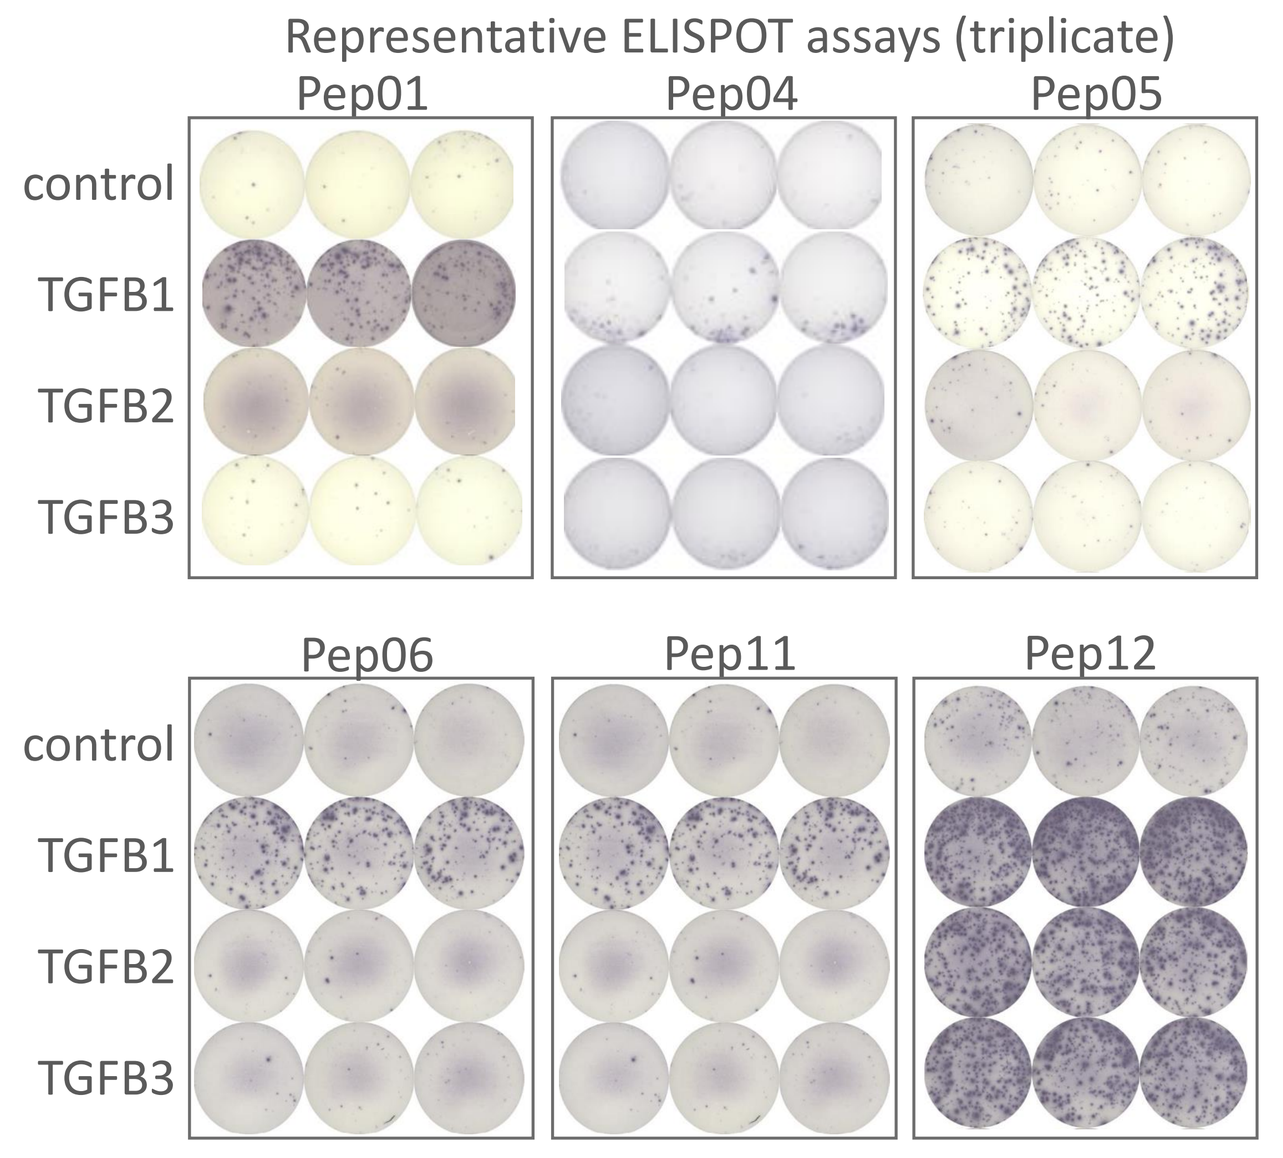

Pep01
Pep04
Pep05
Pep06
Pep11
Pep12
control
TGFB1
TGFB2
TGFB3

Testing peptide specificity for inducing TGFB1-specific immune responses. Donor PBMCs were stimulated with TGFB1 peptide and seven days later were tested for a recall response by IFNg ELISPOT assay using the same TGFB1 peptide, or homologous TGFB2 or TGFB3 peptides. The highly homologous peptide Pep12 was included as a positive control for detecting cross-reactive immune responses to TGFB2 and TGFB3.

Functionality of a TGFB1 peptide vaccine in a mouse tumor model

BACKGROUND We developed a murine TGFB1 vaccine composed of two different peptide antigens encoding predicted MHC class I and class II epitopes. Pre-clinical studies of anti-TGFB therapies often fail to impact tumor growth when administered as a monotherapy. Here we aimed to 1) develop a vaccine that elicits a potent immune response consisting of both CD4+ and CD8+ T cells 2) determine any effect on tumor growth, 3) determine any impact on the TME, and 4) develop assays to further characterize the functionality of vaccine-induced T cell responses.
RESULTS Both peptide antigens (PA1 and PA2) elicited strong immune responses by IFNg ELISPOT assay. Vaccination with PA1 resulted in recognition of minimal peptides encoding class I and class II epitopes. In contrast, PA2 mainly induced a class II response (although we cannot rule out class I epitopes that were not tested). As anticipated, TGFB1 vaccine alone had little impact on tumor growth, with a trend toward reduced tumor growth in PA2-vaccinated animals. There was a similar not significant trend toward reduced TGFB1 expression as assayed by latency associated peptide (LAP) staining. CD4+ T cell infiltration was significantly enhanced in PA2-vaccinated animals while CD8+ T cell infiltration remained unchanged. An in vivo cytotoxicity assay demonstrated that vaccination with PA1 results in cytotoxic activity toward cells loaded with the class I peptide antigen PA1_Ib. Notably, cytotoxic activity was higher in animals vaccinated with PA1_Ib, suggesting superior induction of cytotoxic T cells by the minimal epitope antigen PA1_Ib compared to PA1. Similar assays are being developed to evaluate the targeting of native TGFB1 expressing cells in vitro and in vivo.

TGFB1 vaccine induces robust immune responses

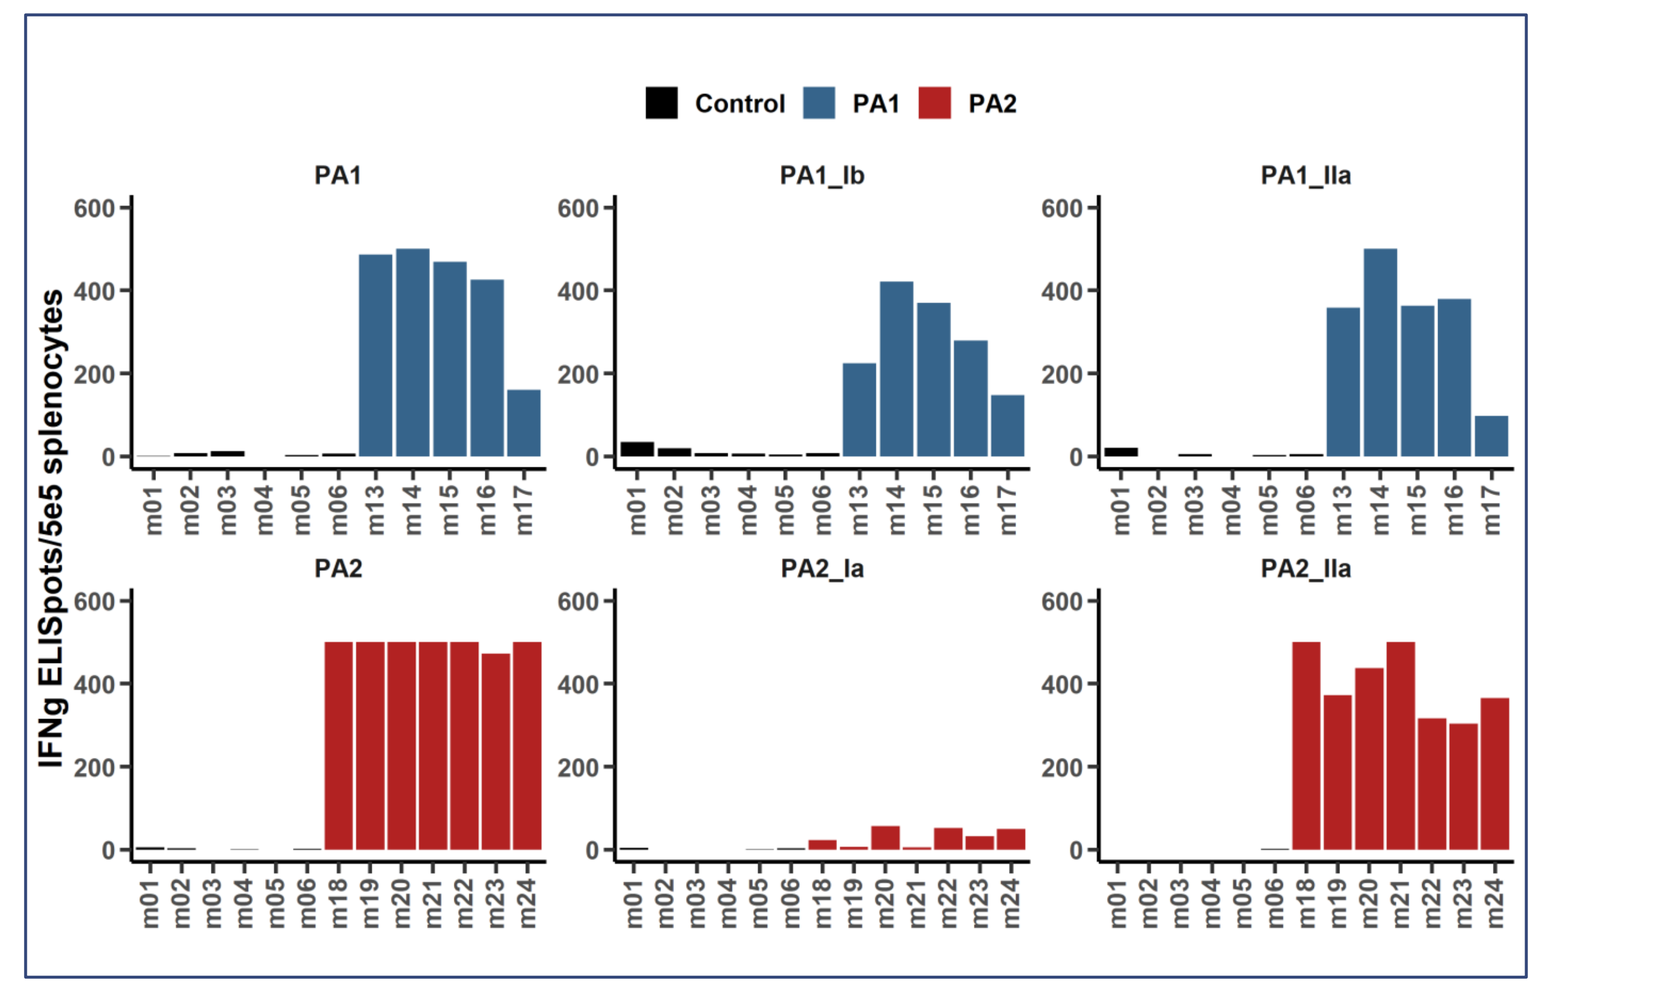

Control
PA1
PA2
PA1
PA1_Ib
PA1_IIa
PA2
PA2_Ia
PA2_IIa
IFNg ELISpots/5e5 splenocytes

Mice were vaccinated with two different SLP peptides and immune responses were analyzed by interferon gamma (IFNg) ELISPOT assay using the peptides shown below. ELISPOT responses for individual mice are shown.

PA1	KDISHSIYMFFNTSDIREAVPEPPL
PA1_Ib	SIYMFFNT
PA1_IIa	IYMFFNTSDIREAVP

PA2	SLDTQYSKVLALYNQHNPGASASP
PA2_Ia	TQYSKVLAL
PA2_IIa	LALYNQHNPGASASP

No significant effect on tumor growth in the MC38 colon cancer model

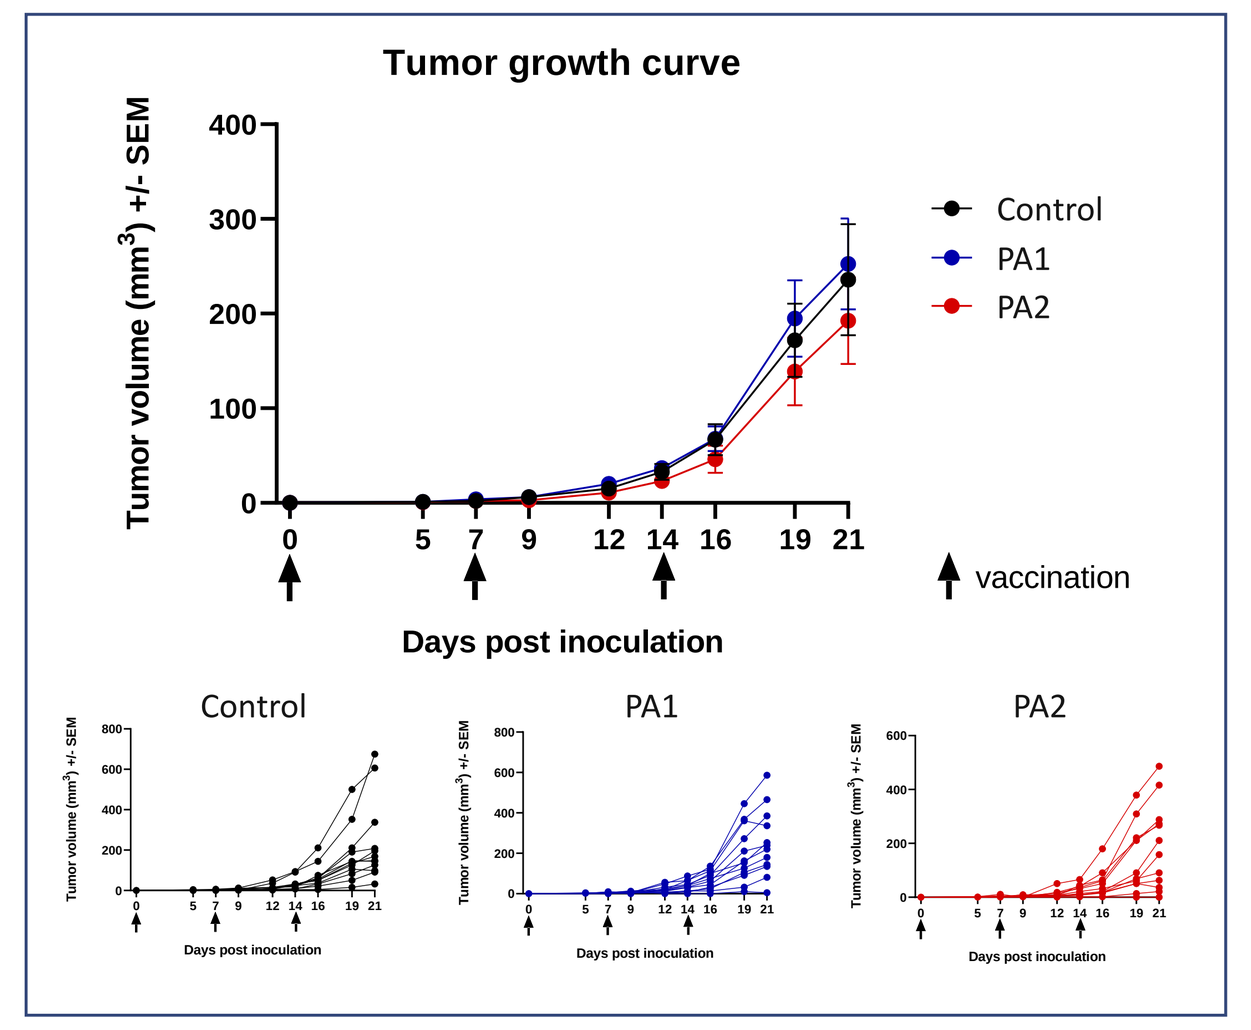

Tumor growth curve
Tumor volume (mm³) +/- SEM
Control
PA1
PA2
vaccination
Days post inoculation

The anti-tumor activity of the TGFB1 vaccine was tested in the MC38 colon cancer model. Vaccine consisting of 100ug peptide in Montanide adjuvant was administered on days 0, 7 and 14, as indicated.

TGFB1 vaccine drives desired immune permissive changes in the TME

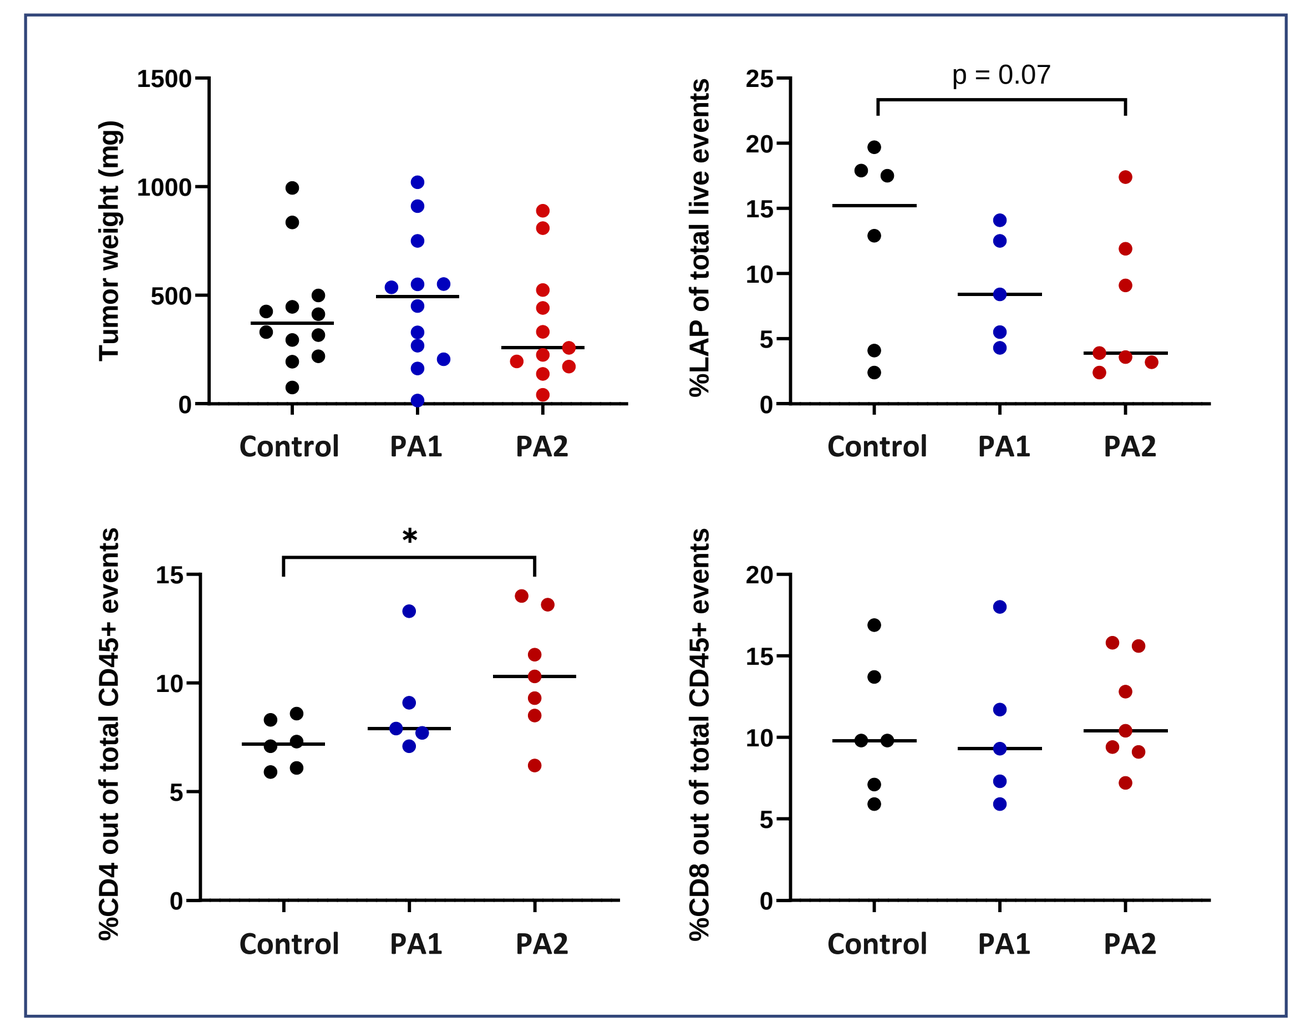

Tumor weight (mg)
%LAP of total live events
p = 0.07
%CD4 out of total CD45+ events
%CD8 out of total CD45+ events
Control
PA1
PA2

Tumor weight is compared between groups at experiment termination (day 21). TGFB1 expression is assayed by latency associated peptide (LAP) staining. CD4+ T cell infiltration was significantly enhanced in PA2 vaccinated animals while CD8+ T cell infiltration remained unchanged.

TGFB1 vaccine promotes targeted *in vivo* cell killing

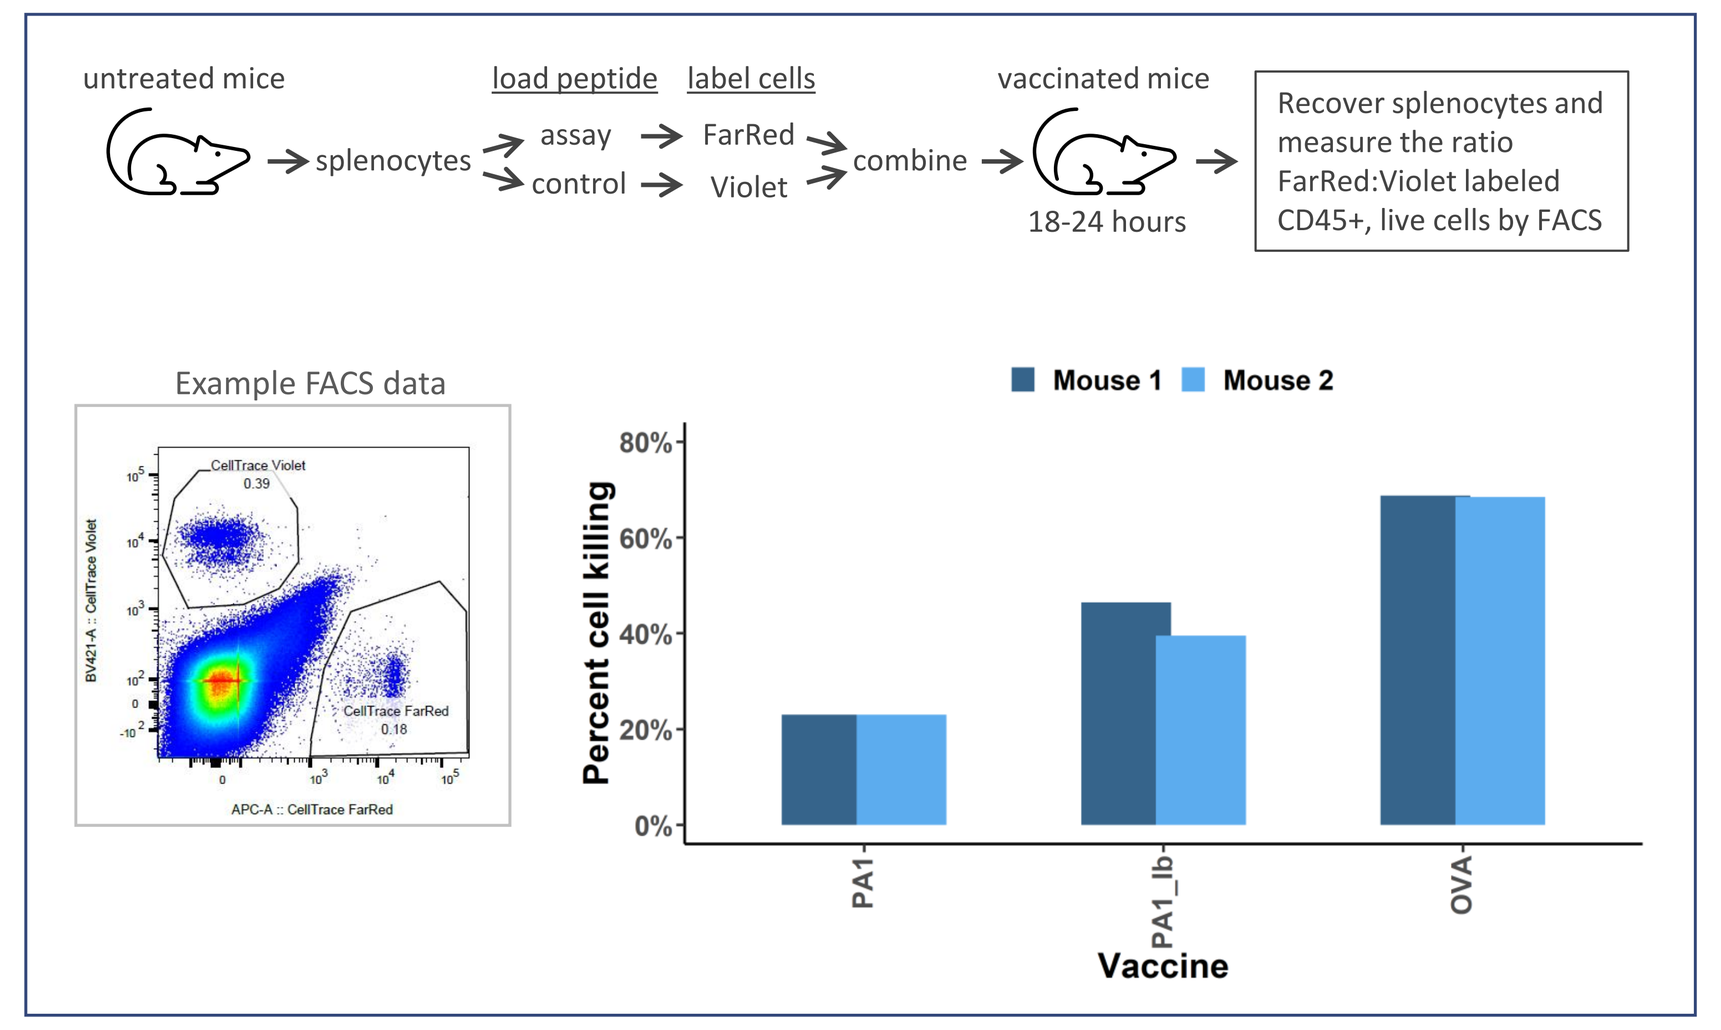

untreated mice
splenocytes
load peptide
assay
control
label cells
FarRed
Violet
combine
vaccinated mice
18-24 hours
Recover splenocytes and measure the ratio FarRed:Violet labeled CD45+, live cells by FACS
Example FACS data
Mouse 1
Mouse 2
Percent cell killing
PA1
PA1_Ib
OVA
Vaccine

An in vivo cytotoxicity assay was used to compare cell killing in mice vaccinated with PA1 versus the class I epitope PA1_Ib (SIYMFFNT). Vaccinated mice (duplicates) were injected with differentially labeled splenocytes loaded with the assay peptide (PA1_Ib) or a control peptide. Cell killing was determined by comparing the recovery of the splenocytes using control-peptide-loaded splenocytes as an internal control for cell recovery. OVA peptide was included as a positive control.

Conclusions

- TGFB1 expression marks a diverse set of immune suppressive cells in the tumor microenvironment independent of other TME antigens and known to contribute to immune therapy resistance
- Targeting TGFB1 expressing cells in the TME via a vaccine approach presents a novel and attractive way to modulate the pathway and drive therapeutic benefit in cancer setting
- We developed and characterized TGFB1-selective peptide vaccines capable of inducing strong immune responses
- Using the murine MC38 tumor model we show preliminary evidence that treatment with a TGFB1 vaccine drives CD4+ T cell infiltration in the TME and promotes *in vivo* targeted cell killing
- Experimental work to elucidate the cellular and molecular mechanisms of a TGFB1 vaccine is ongoing
- The combined data supports further development of a TGFB1 vaccine to modulate the TME for therapeutic benefit in a wide range of cancer indications

References

1. Kjeldsen, J.W., et al., A phase 1/2 trial of an immune-modulatory vaccine against IDO/PD-L1 in combination with nivolumab in metastatic melanoma. Nat Med, 2021. 27(12): p. 2212-2223.
2. Andersen, M.H., Tumor microenvironment antigens. Semin Immunopathol, 2022.
3. Holmstrom, M.O., et al., Cytotoxic T cells isolated from healthy donors and cancer patients kill TGFbeta-expressing cancer cells in a TGFbeta-dependent manner. Cell Mol Immunol, 2021. 18(2): p. 415-426.
4. Mortensen, R.E.J., M.O. Holmstrom, and M.H. Andersen, Characterization of TGFbeta-specific CD4(+)T cells through the modulation of TGFbeta expression in malignant myeloid cells. Cell Mol Immunol, 2021. 18(11): p. 2575-2577.
5. Ma, Y., Q. Guan, A. Bai, C.R. Weiss, C.L. Hillman, A. Ma, G. Zhou, G. Qing, and Z. Peng, Targeting TGF-beta1 by employing a vaccine ameliorates fibrosis in a mouse model of chronic colitis. Inflamm Bowel Dis, 2010. 16(6): p. 1040-50.
6. Chu, X., Y. Li, W. Huang, X. Feng, P. Sun, Y. Yao, X. Yang, W. Sun, H. Bai, C. Liu, and Y. Ma, Combined immunization against TGF-beta1 enhances HPV16 E7-specific vaccine-elicited antitumour immunity in mice with grafted TC-1 tumours. Artif Cells Nanomed Biotechnol, 2018. 46(sup2): p. 1199-1209.